Poster # 1188

The Anti-Respiratory Syncytial Virus (RSV) Nucleoside Analog, AL-8112, and its Orally Bioavailable Prodrug, AL-8176, Exhibit a High Barrier to the Emergence of Viral Resistance

Jin Hong, PhD, Hong Liu, MS, Joshua Taylor, BS, Matthew Mcclure, MD, Sushmita Chanda, PhD, John Fry, BS RGN, Leo Beigelman, PhD, Lawrence Blatt, PhD, Julian Symons, D.Phil*

Alios BioPharma Inc, South San Francisco, CA, USA

*Presenting Author:
Julian Symons, D.Phil
Vice President,
Alios BioPharma Inc.,
260 E. Grand Ave,
South San Francisco,
CA 94080, USA.

ABSTRACT

Background: The triphosphate of AL-8112, a nucleoside analog, is a potent and specific inhibitor of RSV replication in vitro. AL-8176, a prodrug of AL-8112, is in development to treat RSV infection. AL-8176 has demonstrated a rapid decline and clearance of RSV RNA compared to placebo in a challenge study in healthy volunteers (HV) infected with a clinical strain of RSV. Generally, antiviral nucleoside analogs exhibit higher barriers to the emergence of viral resistance compared to other compound classes. The resistance profile of RSV after exposure to AL-8112 is unknown.

Objectives: To characterize the development of viral resistance to AL-8112 in vitro and in vivo.

Methods: In vitro, RSV A2 was passaged in HEp-2 cells with increasing concentrations of AL-8112 or a fusion inhibitor (FI). In the challenge study, nasal wash samples were collected before, during, and after dosing. Prespecified viral load criteria to identify resistance and breakthrough were established. Using viral population sequencing, amino acid (aa) sequences were compared to the subject's first available RSV-positive sample and the reference RSV sequence.

Results: After 35 passages, AL 8112 exhibited a >50-fold shift in activity compared to the control passaged virus in an antiviral assay. In contrast, resistance to RSV FI developed after 4 passages. Sequencing of the genome after passaging with AL-8112 revealed 4 aa substitutions in the RSV polymerase domain; M628L, A789V, L795I and I796V (quad mutant). Clonal sequencing of 94 clones revealed that 93.6% of the genomes contained all 4 aa substitutions. Engineering the quad mutant into the RSV mini-genome system showed a 39-fold shift of the mutant vs. wild-type polymerase.

In the challenge study, no subjects met the prespecified criteria for resistance or breakthrough; consequently we evaluated 17 AL-8176- and 12 placebo-treated subjects for evidence of viral resistance. Analysis of the RSV polymerase coding domain did not detect the aa variants associated with in vitro resistance to AL-8112 or identify any new resistance-associated variants.

Conclusion: In vitro, AL-8112 demonstrated a higher barrier to resistance compared to an RSV FI. Further, we observed no emergence of resistance to AL-8112 in an RSV human challenge model.

INTRODUCTION

- Respiratory syncytial virus (RSV), a pneumovirus and member of the family *Paramyxoviridae* is a non-segmented, single-stranded, negative sense RNA virus, which infects the ciliated bronchial epithelial cells leading to severe lower respiratory infection (LRI).[1-5]
- Palivizumab, a monoclonal antibody for RSV, is used in high-risk populations as a prophylaxis but its expense limits its use especially in developing countries.[6]
- AL-8176 is a diester prodrug of the cytidine nucleoside analog, AL-8112, and is currently under development for the treatment of RSV infection. After oral administration, AL-8176 is rapidly metabolized to AL-8112, the parent nucleoside, which subsequently converts within cells to the pharmacologically active nucleoside triphosphate (NTP).
- AL-8112 is a potent and selective inhibitor of RSV RNA replication in vitro[7] while the NTP is known to inhibit the RNA dependent RNA polymerase activity of RSV polymerase.
- Generally, antiviral nucleoside analogs exhibit higher barriers to the emergence of viral resistance compared to other compound classes. However, the resistance profile of RSV after exposure to AL-8112 was previously unknown.

OBJECTIVES

- To characterize the development of viral resistance to AL-8112 in vitro and in vivo.

MATERIALS AND METHODS

In vitro viral drug resistance generation

- RSV A2 was passaged in HEp-2 cells with increasing concentrations of AL-8112 (starting concentration: 1 µM, approximately 10 times the EC_{50} of AL-8112 against RSV A2). After passaging 6 times at the same concentration, the drug concentration was raised to 2× that of the previous concentration and passaging repeated again for 6 times. When the AL-8112 concentration reached the highest level of 16 µM, the virus was passaged more than 20 times in the same drug concentration. As a positive control, RSV A2 resistance to the RSV FI, BMS433771, (starting concentration: 30 nM; 10× EC_{50} of BMS433771 against RSV A2) was generated in parallel with AL-8112. As a negative control, RSV A2 was passaged in cells without the compound.

RSV antiviral assay

- AL-8112 was serially diluted (1:3) to form up to 9 concentrations distinct and was used to pre-treat the HEp-2 cells in duplicate in a 96-well format. After 24 hours of incubation with compounds, RSV A2 at a multiplicity of infection (MOI) of 0.5 was added to the cells (except background controls) and the plates were incubated for an additional 4 days. The RSV viral RNA was isolated from the collected supernatant of each well using a MagMAX™ Viral RNA Isolation Kit (Life Technologies) automated through MagMAX™ Express-96 Deep Well Magnetic Particle Processor (Life Technologies). The viral RNA was then quantified by real-time polymerase chain reaction (RT-PCR) using the following primer/probe set.

Table 1: Primer/probe for RT-PCR

Strain	Upstream Primer	Probe	Downstream Primer
RSV A2	5'-GCT CTT AGC AAA GTC AAG TTG AAT GA-3'	6FAM-ACA CTC AAC AAA GAT CAA CTT CTG TCA TCC AGC-MGBNFQ	5'-TGC TCC GTT GGA TGG TGT ATT-3'

Minigenome assay

- The RSV minigenome system was used as a phenotypic assay for evaluating the effect of amino acid changes in the RSV L protein. On day 1, HEp-2 cells were plated in each well of the 6-well plates and after five hours, serially diluted AL-8112 was added to the cells (except for the wells of no drug treatment control), and incubated overnight. The next day, modified virus Ankara T7, vaccinia virus expressing T7 RNA polymerase, was added to the cells at an MOI of 10, and after two hours a mixture of 6 plasmids was transfected into the cells using Fugene 6 (Promega). The plasmid mixture included RSV minigenome pGem.RSV.M5.Luc plus 4 plasmids containing RSV L, N, P, and M2-1 genes, respectively, as well as a control plasmid pRL-SV40 (Promega) expressing Renilla luciferase for transfection normalization.
- After 48 hours, a dual luciferase assay (Promega) was performed on the cells. Firefly luciferase signals were normalized against Renilla luciferase signals and then the background signal from the no L gene plasmid control well was subtracted.

Growth kinetics

- Viral isolates purified from limited dilution of AL-8112 and no drug control P35 pools were inoculated into separate flasks of HEp-2 cells in compound-free media at the same MOI (0.2 or 2). The supernatants, together with an RSV A2 live virus standard were processed to the isolate viral RNA and quantified by qPCR using the viral RNA from the RSV A2 as a standard. Viral titers were reported PFU equivalent per mL (PFUe/mL).

In vivo viral resistance (RSV Human Challenge Study)

- Nasal wash samples for RSV L gene genotypic analysis were collected from all (N=35) healthy volunteers successfully infected with Memphis 37b, and dosed for 5 days with AL-8176 or placebo enrolled in a randomized, double-blind RSV challenge study (NCT02094365). Samples were collected prior to the first dose of study medication (at baseline) and twice-daily thereafter.
- Genotypic analysis was planned to be performed on all samples meeting predefined viral resistance assessment criteria, including those who may have demonstrated viral rebound, partial viral suppression or non-response to treatment with AL-8176.
 - o Viral rebound: a sustained ≥1.0 log_{10} PFUe/mL increase of RSV RNA above nadir before study day 12 (≥2 consecutive measurements), where nadir was a ≥1.0 log_{10} PFUe/mL decrease from peak viral titer.
 - o Partial viral suppression: an initial RSV RNA decline (≥1.0 log_{10} PFUe/mL decrease from peak viral titer) followed by stabilization (≥2 consecutive measurements with RSV RNA levels similar to those at nadir) while receiving AL-8176 through the seventh day of dosing.
 - o Non-response: no evidence of a decrease in viral titer following initiation of AL-8176 through day 3 of dosing.

RSV L gene genotypic analysis

- The RSV viral RNA was isolated from nasal wash samples using a MagMAX™ Viral RNA Isolation Kit automated through MagMAX™ Express-96 Deep Well Magnetic Particle Processor (Life Technologies).
- The first strand cDNA was synthesized using SuperScript III First-Strand Synthesis System for reverse transcriptase-polymerase chain reaction (RT-PCR) (Life Technologies).
- RSV RNA was evaluated for potential treatment-emergent resistance mutations by amplifying and sequencing the RSV L gene region encoding the polymerase domain (amino acids 550–1100).
- The nucleotide and amino acid sequences of all samples were compared with the RSV Memphis 37b strain reference sequence and the treatment samples were compared with baseline sequences from the same individual.
- Changes in nucleotide and amino acid sequences were recorded.

Next generation sequencing

- Crude PCR products were submitted to Monogram Biosciences (LabCorp Speciality Testing Group, South San Francisco, CA) for further processing and next generation sequence analysis.

RESULTS

In vitro viral resistance

Figure 1: Activity of AL-8112 versus RSV A2 at passage 35

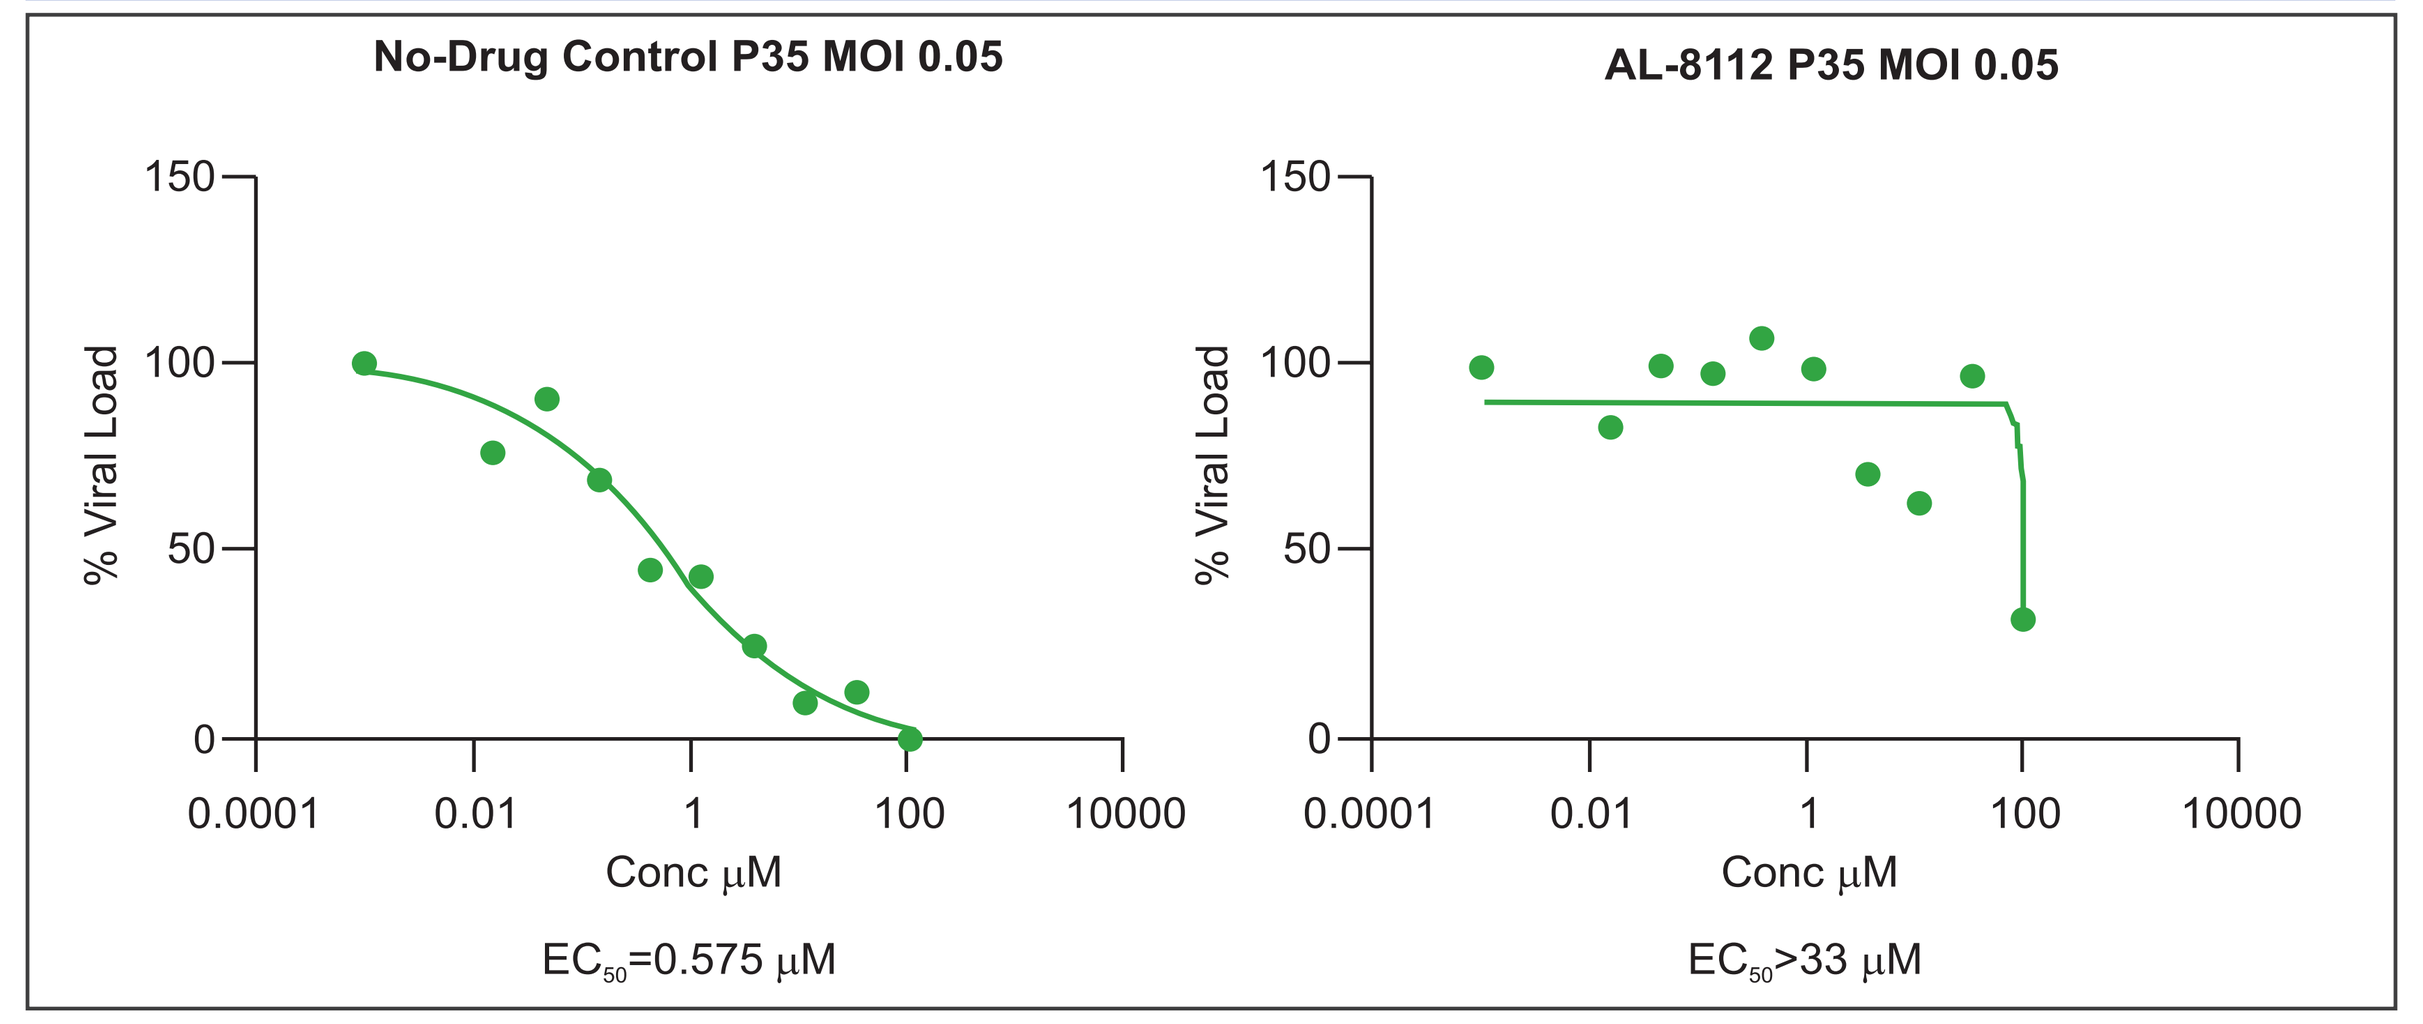

No-Drug Control P35 MOI 0.05
AL-8112 P35 MOI 0.05
% Viral Load
Conc µM
EC50=0.575 µM
EC50>33 µM

- AL-8112 demonstrated activity versus the no-drug control passaged virus. After 35 total passages AL-8112 exhibited a >50-fold EC_{50} shift in an antiviral assay when compared with the control virus pool.
- Full genome sequencing of the AL-8112 P35 virus showed 4 amino acid substitutions by population sequencing; methionine 628 to leucine (M628L), alanine 789 to valine (A789V), leucine 795 to isoleucine (L795I), isoleucine 796 to valine (I796V) in the polymerase coding region of the RSV L gene.

Figure 2: Clonal sequencing analysis of the AL-8112 P35 RSV Pool

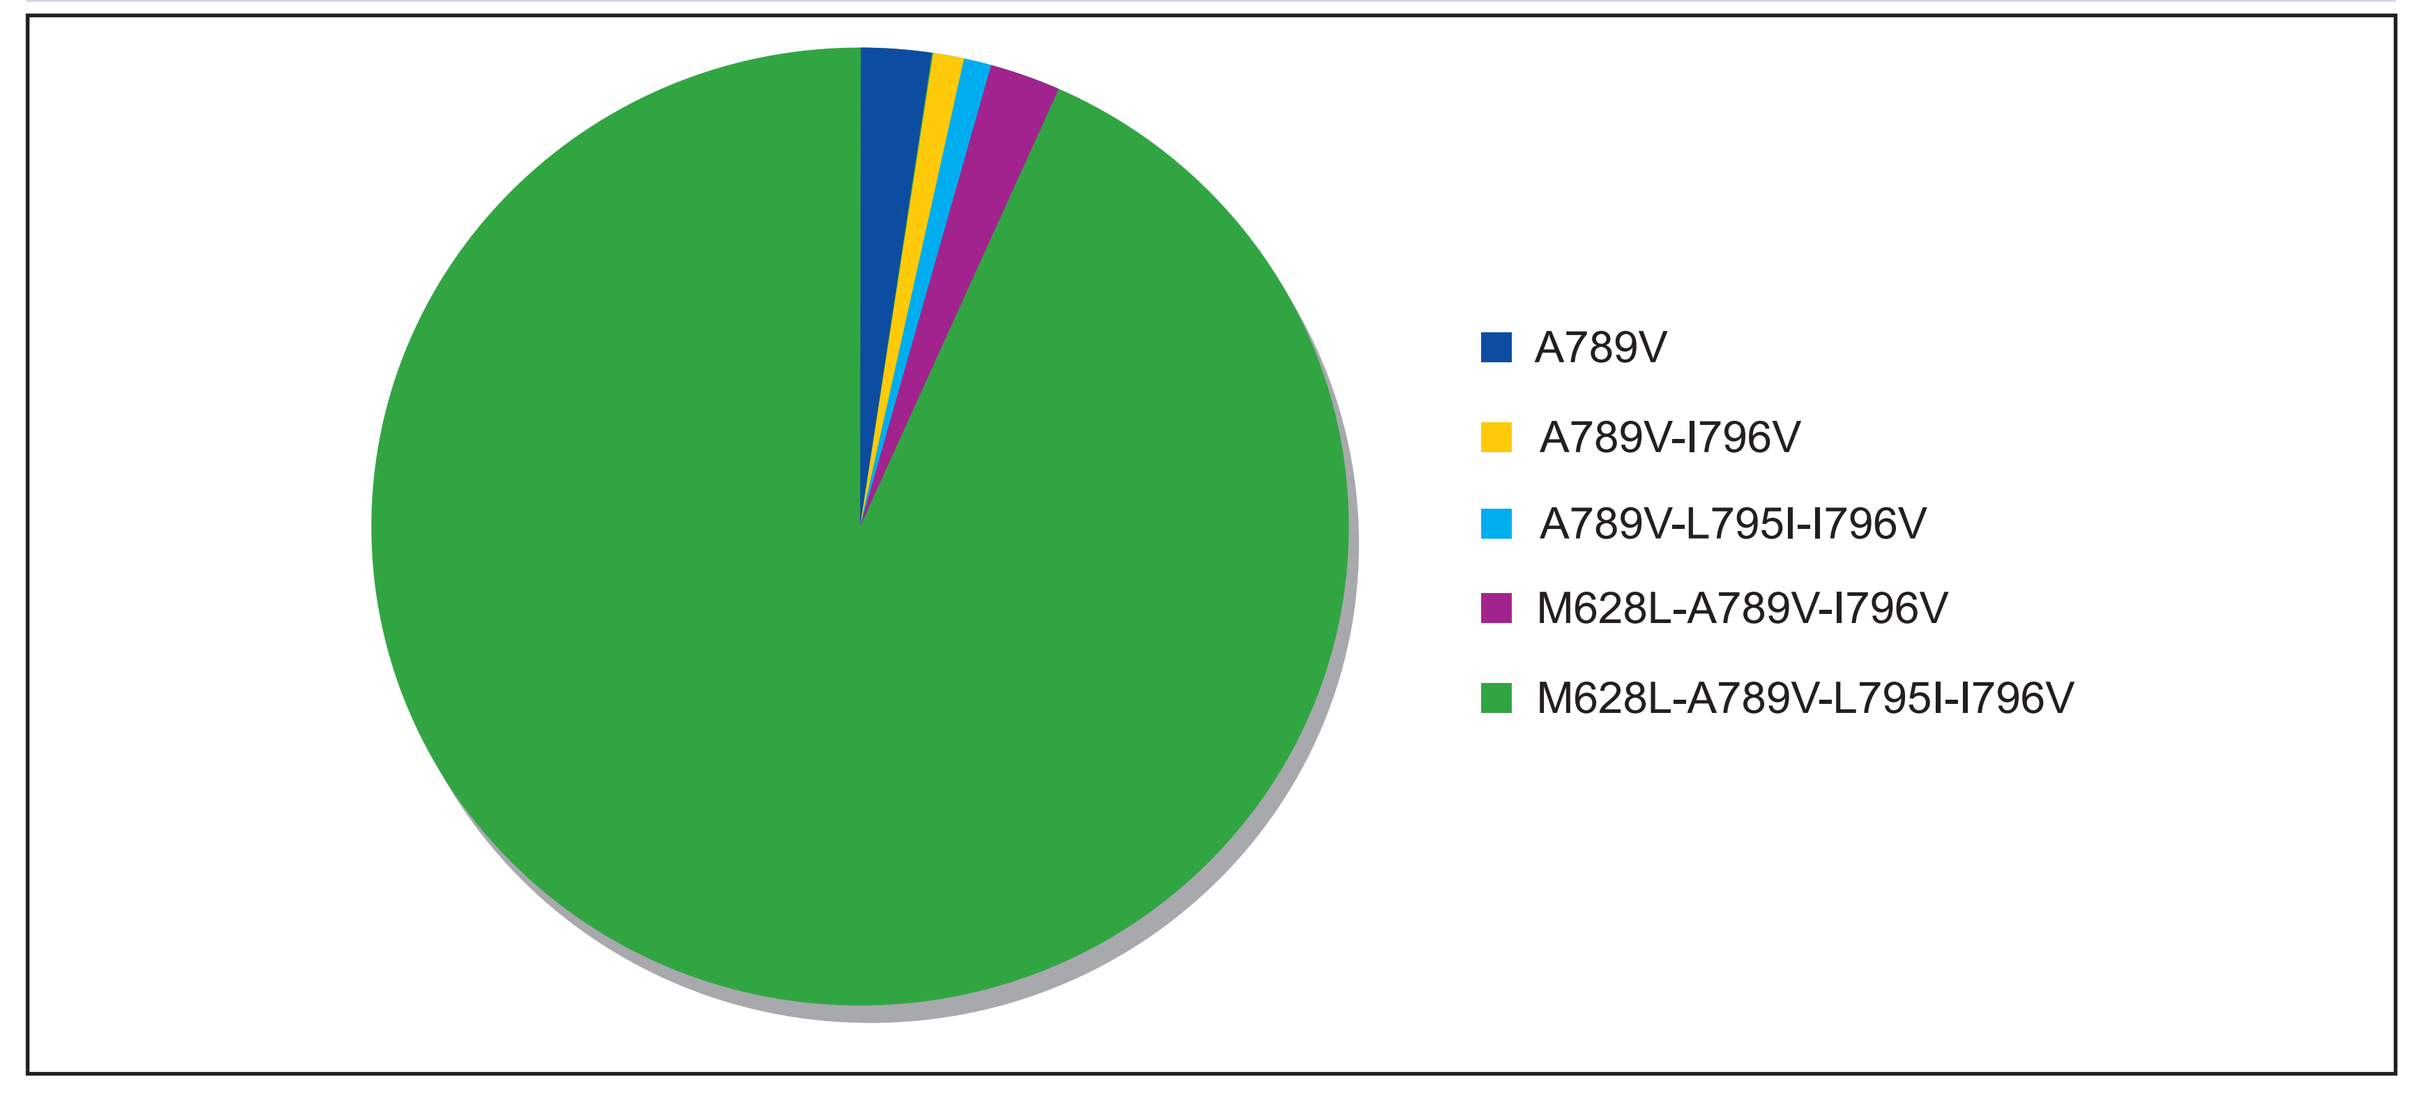

A789V
A789V-I796V
A789V-L795I-I796V
M628L-A789V-I796V
M628L-A789V-L795I-I796V

- In a parallel duplicate culture, the same amino acid changes were observed at passage 14 (38 total passages) of the 16 µM AL-8112 selection pool.
- In the AL-8112 resistant virus pool, 93.6% of the virus quasispecies contained the quadruple amino acid mutation (quad mutant) M628L/A789V/L795I/I796V. Some minor populations of single, double, and triple mutations among these 4 positions were also observed in the 98 clones.
- No mutations were observed at these 4 amino acid positions after analysis of 98 clones in the control pool.

Figure 3: Evolution of resistance mutations to A) AL-8112 and B) Fusion inhibitor, BMS433771 (AL-8013)

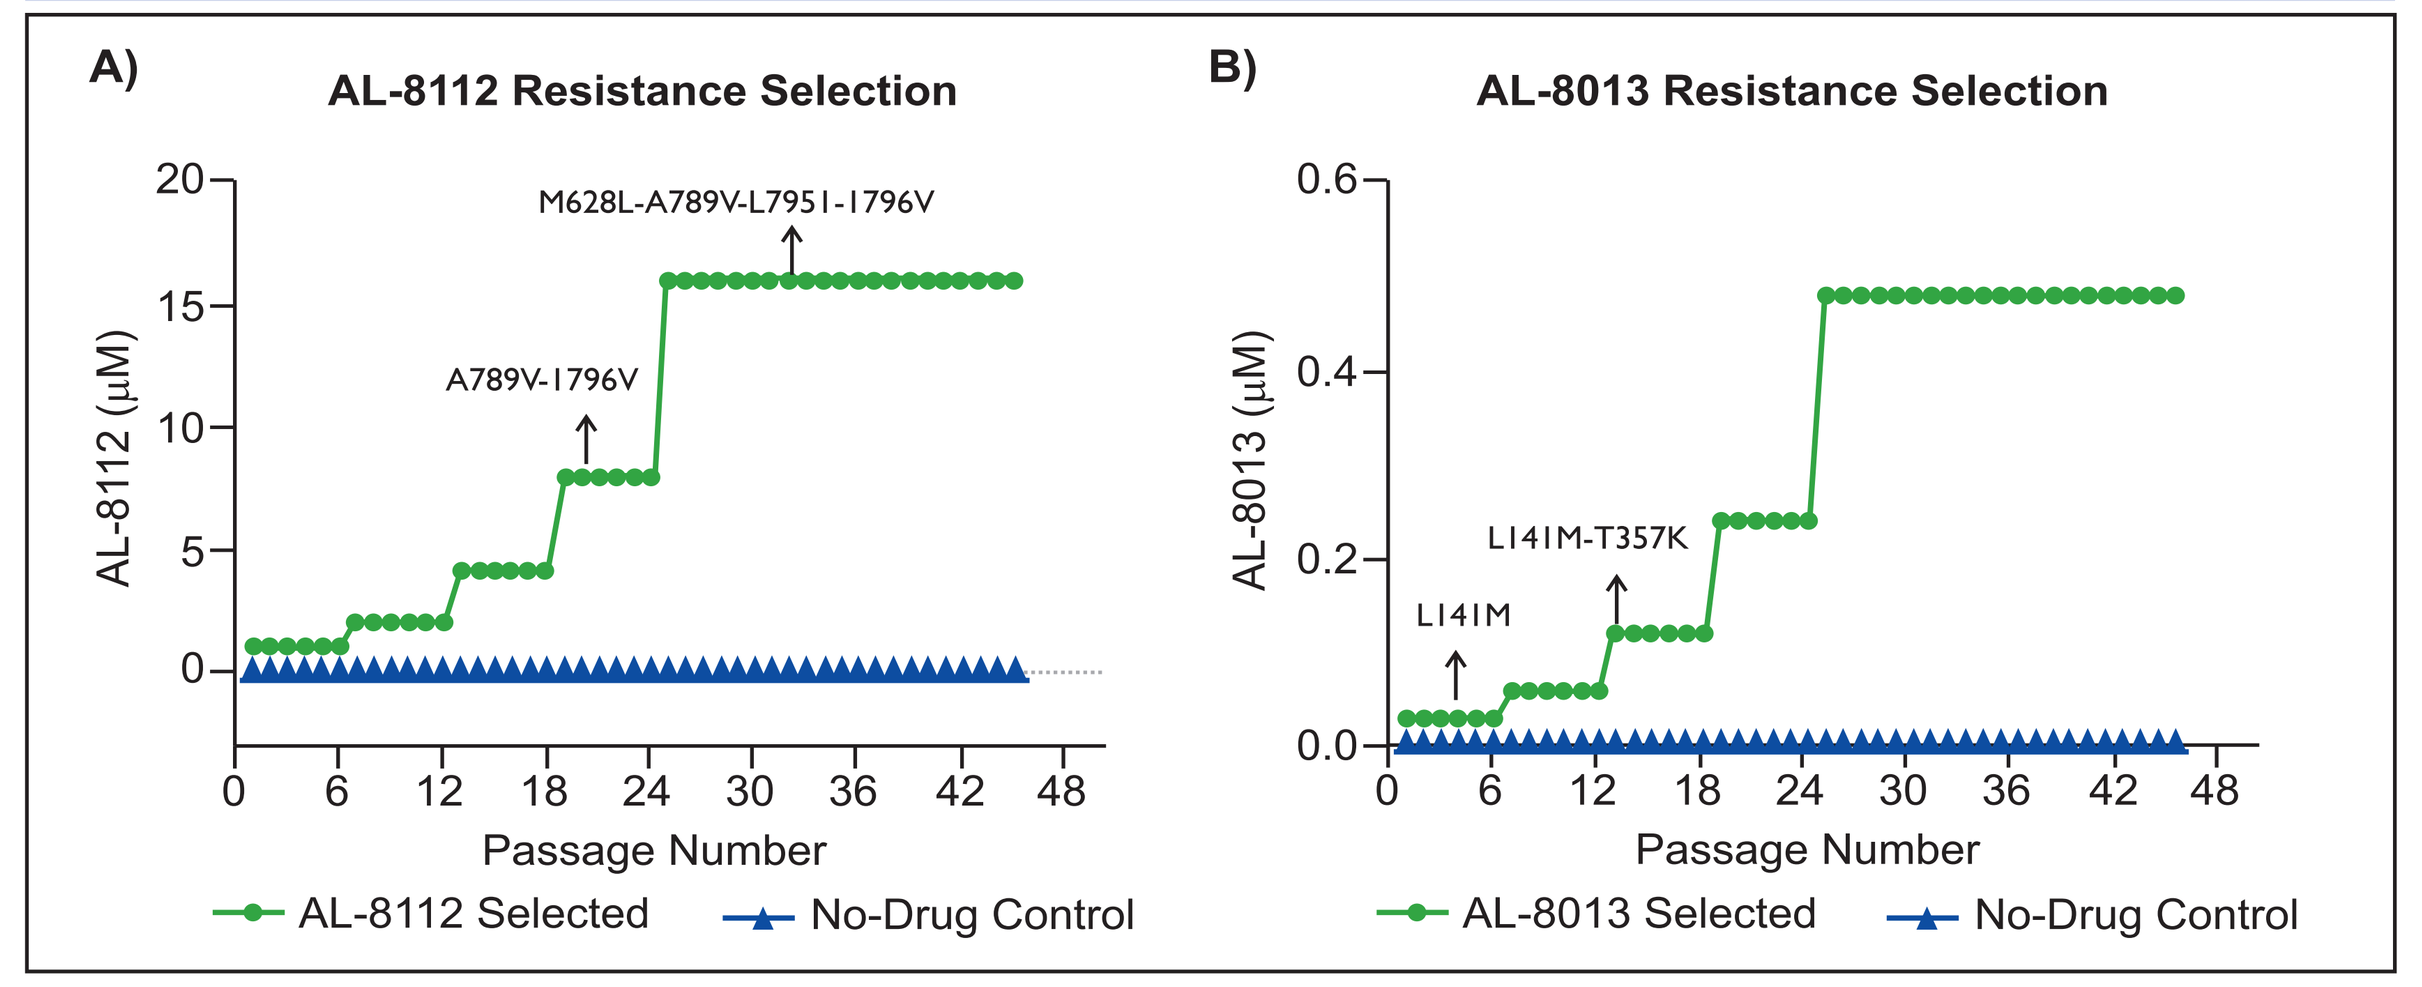

A)
AL-8112 Resistance Selection
M628L-A789V-L795I-I796V
A789V-I796V
AL-8112 (µM)
Passage Number
AL-8112 Selected
No-Drug Control
B)
AL-8013 Resistance Selection
L141M-T357K
L141M
AL-8013 (µM)
Passage Number
AL-8013 Selected
No-Drug Control

- A double mutant A789V/I796V appeared at passage 20 in the AL-8112 selection pool, at a drug concentration of 8 µM (~80×EC_{50}), which persisted in further passages until the quad mutation M628L/A789V/L795I/I796V appeared at passage 32 at a concentration of 16 µM (~160×EC_{50}).
- In contrast, the RSV FI BMS433771 (AL-8013) generated an initial resistance mutation in the RSV F protein (L141M) at passage number 4, at a drug concentration of 30 nM (~10×EC_{50}). The L141M mutation remained in the next few passages until a double mutation L141M-T357K appeared in passage 13, at a drug concentration of 120 nM (~40×EC_{50}). The L141M-T357K double mutation persisted in all of the following passages.

Table 2: Effect of RSV L gene amino acid substitutions on AL-8112 activity using the RSV minigenome system

Minigenome L Gene Construct	AL-8112 EC_{50} (µM) Mean (SEM)	Fold-shift in EC_{50}
Wild-type	0.25 (0.02*)	–
M628L/A789V/L795I/I796V	9.7 (3.12)	39
M628L	ND	ND
A789V	1.9 (0.2)	7.6
L795I	0.85 (0.06)	3.4
I796V	1.18 (0.2)	4.7
A789V/I796V	2.3 (0.4)	9.2
A789V/L795I/I796V	10.5 (3.8)	42

*n=3; ND, not determined; SEM, standard error mean.

- Individually A789V, L795I, and I796V demonstrated varying degrees of resistance to AL-8112. The quad mutant demonstrated a 39-fold shift in EC_{50} against AL-8112. The dual mutant (A789V/I796V), which evolved at passage 20, exhibited a 9.2-fold shift in EC_{50} to AL-8112. Finally, the triple mutant (A789V/L795I/I796V) demonstrated a similar level of resistance to AL-8112 as the quad mutant (42-fold shift).
- The effect of the RSV L protein amino acid substitution, M628L, on the activity of AL-8112 could not be determined as the RSV minigenome system containing this single amino acid change failed to generate a Firefly luciferase signal.
- This suggests that the M628L substitution is highly deleterious to the activity of the L protein and that one or more of the A789V/L795I/I796V amino acid changes may compensate for this loss in polymerase activity.

Fitness analysis of AL-8112

Figure 4: Fitness analysis of AL-8112 resistant variants using the RSV minigenome system

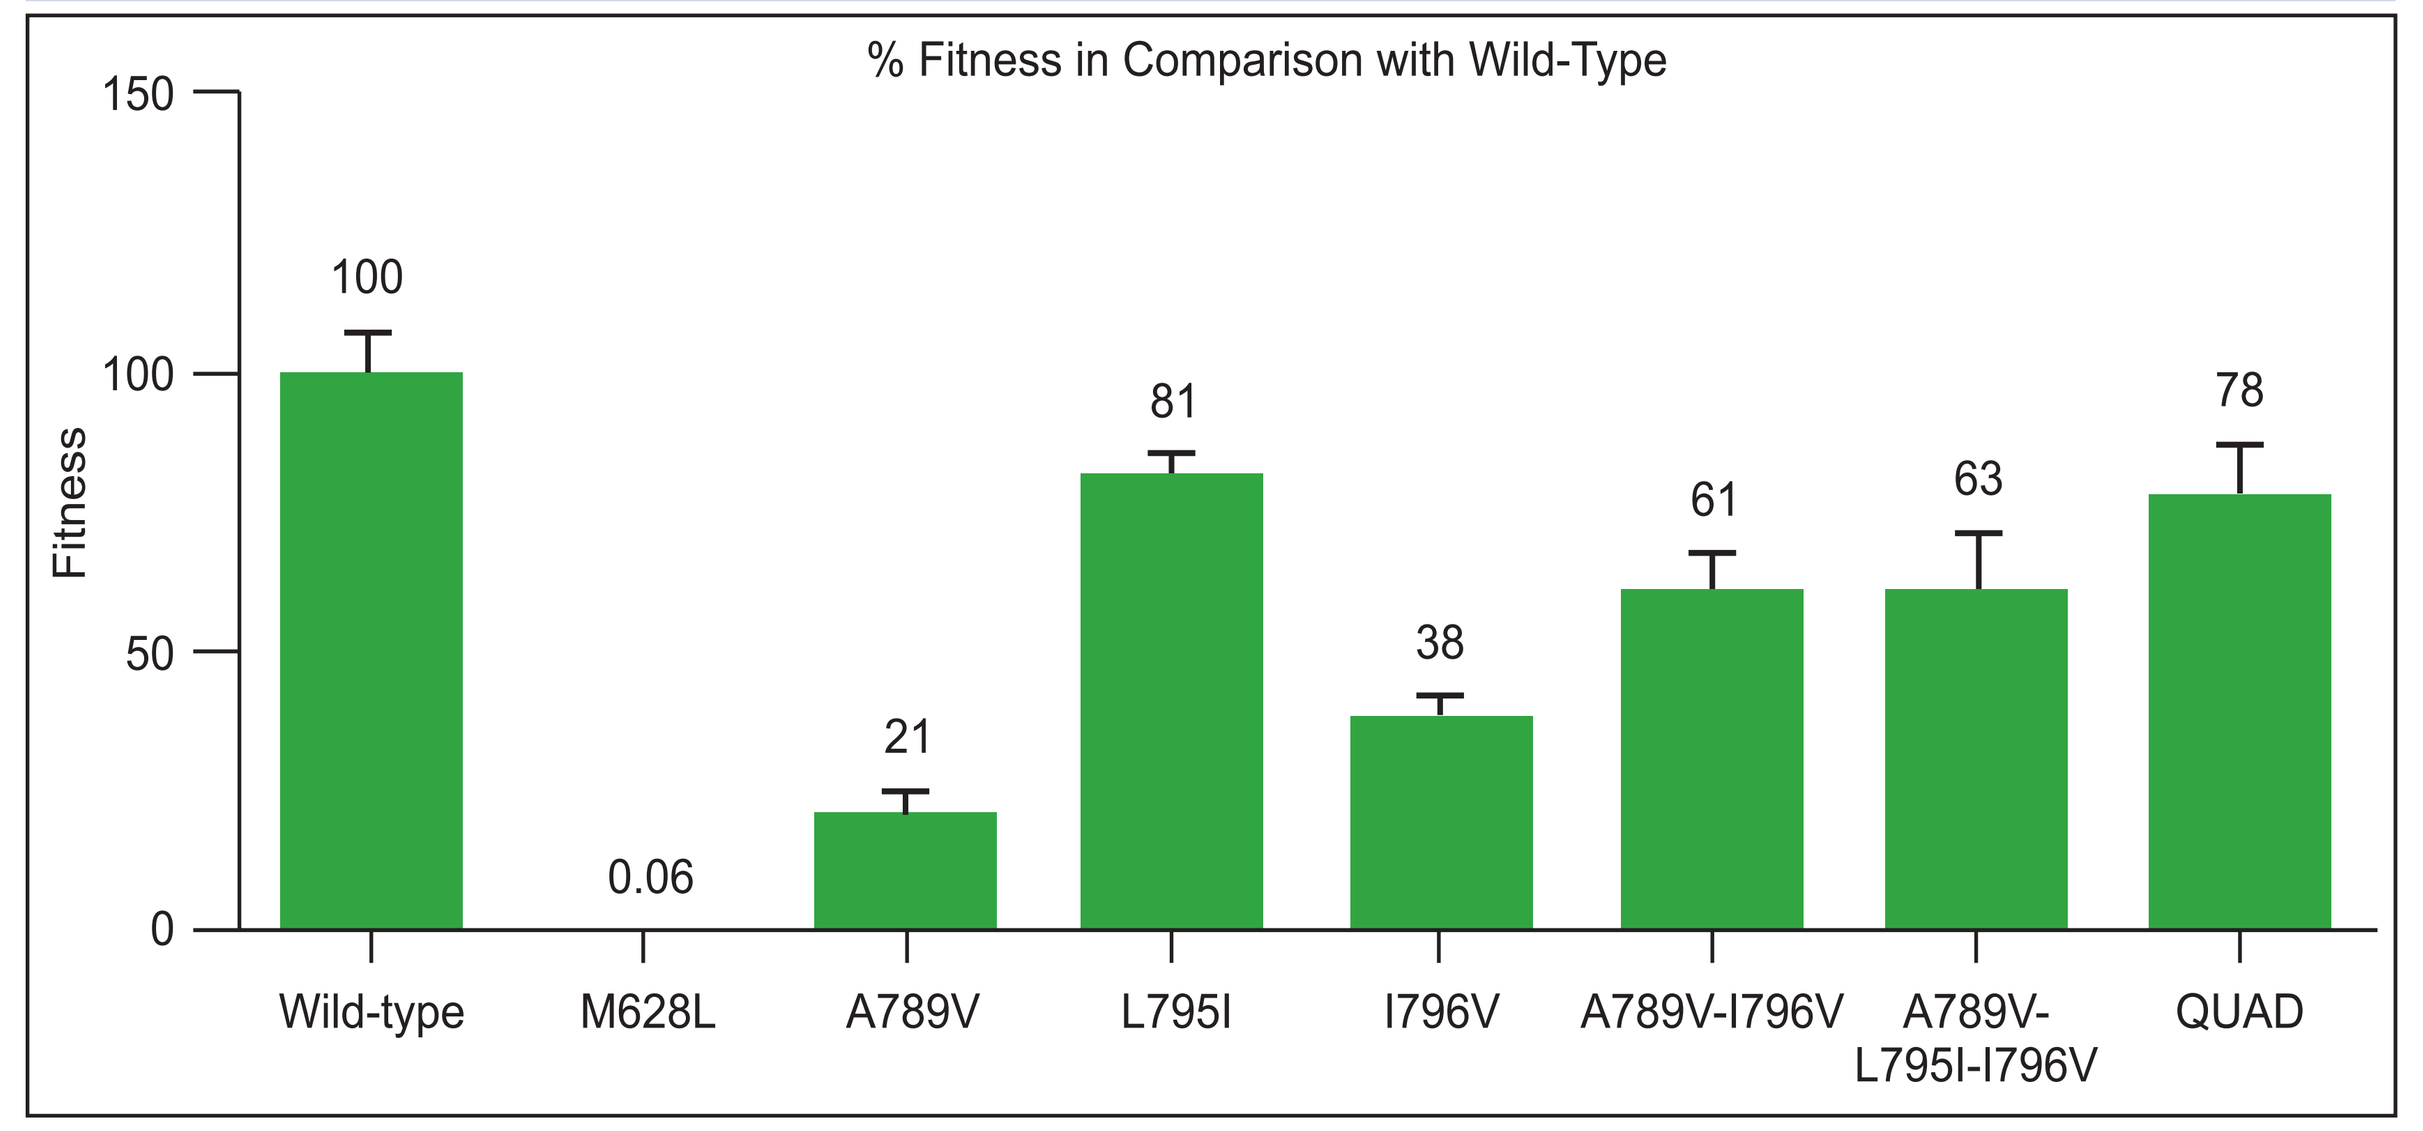

% Fitness in Comparison with Wild-Type
Fitness
100
0.06
21
81
38
61
63
78
Wild-type
M628L
A789V
L795I
I796V
A789V-I796V
A789V-L795I-I796V
QUAD

- The M628L variant demonstrated the most significant decline in fitness with only 0.06% relative fitness when compared to the wild-type (WT) construct. The single amino acid variants; A789V, L795I, and I796V showed 21%, 81%, and 38% relative fitness, respectively. Finally, the double mutant (A789V/I796V), triple mutant (A789V/L795I /I796V), and quad mutant showed 61%, 63%, and 78% relative fitness, respectively.

Viral growth kinetics in RSV A2

- The RSV A2 virus encoding the quad mutant exhibited growth kinetics similar to the WT RSV A2 virus when inoculated either at an MOI of 0.2 or 2.0.
- A dilutionally cloned RSV A2 virus containing the quad mutation (M628L/A789V/L795I/I796V) was passaged in triplicate cultures without AL-8112. Viral RNA was isolated after each passage and the RSV polymerase domain encoding region was sequenced.

Table 3: Reversion of the quad mutation in the absence of AL-8112

	Culture #1 amino acid position				Culture #2 amino acid position				Culture #3 amino acid position			
	628	789	795	796	628	789	795	796	628	789	795	796
P0	L	V	I	V	L	V	I	V	L	V	I	V
P1	L	V	I	V	L	V	I	V	L	V	I	V
P2	M	A	L	I	L	V	I	V	L	V	I	V
P3	M	A	L	I	L	V	I	V	M/L	A/V	L/I	I/V
P4	M	A	L	I	L	V	I	V	M/L	A/V	L/I	I /V
P5	M	A	L	I	M/L	A/V	L/I	I/V	M/L	A/V	L/I	I/V
P6	M	A	L	I	M/L	A/V	L/I	I/V	M	A	L	I
P7	M	A	L	I	M/L	A/V	L/I	I/V	M	A	L	I
P8	M	A	L	I	M/L	A/V	L/I	I/V	M	A	L	I
P9	M	A	L	I	M/L	A/V	L/I	I/V	M	A	L	I
P10	M	A	L	I	M	A	L	I	M	A	L	I
P11	M	A	L	I	M	A	L	I	M	A	L	I
P12	M	A	L	I	M	A	L	I	M	A	L	I

A, alanine; I, Isoleucine; L, leucine; M, methionine; P, passage; V, valine

- After 2 passages, the quad mutation (L, V, I, V) had completely reverted back to the WT sequence (M, A, L, I) in Culture #1. In Culture #2, a mixed population was observed at passage 5 and the virus had reverted back to the WT sequence by passage 10. In Culture #3 a mixed population was observed at passage 3 and the virus had reverted back to the WT sequence by passage 6.

In vivo viral resistance (RSV Human Challenge Study)

- None of the AL-8176-treated volunteers in the study met the predefined viral resistance assessment criteria; hence, samples were selected based on posthoc sample selection criteria (some slight indication of potential viral rebound [those volunteers, with a single ≥0.5 log_{10} PFUe/mL increase of RSV RNA above nadir before study day 12, where nadir was a ≥1.0 log_{10} PFUe/mL decrease from peak viral load]).
- In total, samples from 17 AL-8176-treated volunteers and 12 placebo-treated volunteers selected as controls were evaluated.

RSV L gene genotypic analysis

- The majority of samples selected for analysis did not show any amino acid changes from either the reference or baseline sequences.
- Sequence analysis of the RSV L gene region encoding the RSV polymerase domain did not detect any of the amino acid variants associated with in vitro resistance to AL-8112 (M628L, A789V, L795I, or I796V) or identify any new variants associated with the potential emergence of resistance to AL-8112.
- Further, next generation sequence analysis was conducted to detect genetic variants in RSV containing samples present at ≥1% to allow for detection of genetic variants that may have been undetectable using conventional Sanger-based sequencing approaches. The 4 amino acid changes associated with in vitro resistance to AL-8112 (M628L, A789V, L795I, and I796V) were not present in any sample at ≥1%.

CONCLUSIONS

- In vitro AL-8112 demonstrated a higher barrier to viral resistance development compared to an RSV fusion inhibitor, where the quad mutant (M628L, A789V, L795I, or I796V) appears to mediate viral resistance to AL-8112.
- After 5 days of dosing with AL-8176 in a RSV human challenge study, sequence analysis of the RSV L gene region encoding the RSV polymerase domain did not detect any of the amino acid variants associated with in vitro resistance to AL-8112 (M628L, A789V, L795I, or I796V) or identify any new variants associated with the potential emergence of resistance to AL-8112.

REFERENCES

1. Nair H et al. *Lancet* 2010;375:1545–55; 2. Hall CB et al. *Pediatrics* 2013;132:e341–8; 3. Marcelin JR et al. *Transpl Infect Dis* 2014;16:242–50; 4. Falsey AR et al. *Drugs Aging* 2005;22:577–87; 5. Sikkel MB et al. *Pediatr Infect Dis J* 2008;27:S63–70; 6. Deval J et al. *PLoS Pathog* 2015;11:e1004995; 7. Wang G et al. *J Med Chem* 2015;58:1862–78.

ACKNOWLEDGEMENTS

The authors thank Dr. Vaibhav Deshpande (SIRO Clinpharm Pvt. Ltd.) for providing writing assistance, Dr. Bradford Challis (Janssen Research & Development, LLC.) for providing additional editorial support for this poster development and Mr. Sandeep Chavan (SIRO Clinpharm Pvt. Ltd) for poster layout. The authors also thank the study participants, without whom this study would not have been accomplished, as well as Chris Westland, Mike Yee, and Nick Khorlin from Alios BioPharma, and the clinical team at hVIVO for their assistance in conducting this study.

DISCLOSURES

All authors are employees of Alios BioPharma Inc., a Janssen Pharmaceutical Company of Johnson & Johnson, South San Francisco, CA, USA. All authors met ICMJE criteria and all those who fulfilled those criteria are listed as authors. All authors had access to the study data and contributed to the development of this poster.

REGISTRATION

ClinicalTrials.gov Identifier: NCT01906164.

Funded by Janssen Research & Development, LLC, USA and Alios Biopharma, Inc, CA, USA

Poster presented at the IDWeek 2015, October 7-11, 2015, San Diego, CA, USA