726

Safety of Bivalent rLP2086 (MenB-FHbp), a Meningococcal Serogroup B Vaccine, in Young US Adults: Results From a Phase 3 Trial

Lars Østergaard, MD, PhD,[1] Brian J. Ward, MD,[2] Johannes F. Beeslaar, MD,[3] Joseph J. Eiden, MD, PhD,[4] Kathrin U. Jansen, PhD,[4] Judith Absalon, MD, MPH,[4] Laura J. York, PhD,[5] David Radley, MS,[6] Jean-Louis Prégaldien, MS,[7] Nina Breinholt Staerke, MD,[1] John L Perez, MD, MA[5]*

[1]Aarhus Universitetshospital, Skejby, Aarhus, Denmark; [2]Research Institute of the McGill University Health Center, Montreal, QC, Canada; [3]Pfizer Vaccine Clinical Research and Development, Maidenhead, UK; [4]Pfizer Vaccine Research, Pearl River, NY, USA; [5]Pfizer Medical Development and Scientific Affairs, Collegeville, PA, USA; [6]Pfizer Vaccine Research and Development, Collegeville, PA, USA; [7]Pfizer Vaccine Research and Development, Brussels, Belgium

*Presenting Author

For more information please contact:
Johannes F. Beeslaar, MD
Pfizer Ltd
Walton Oaks Dorking Road
Tadworth, KT20 7NS
United Kingdom
Johannes.Beeslaar@pfizer.com
Phone: +44 1628 692066
Fax: +44 1628 692141

ABSTRACT

Background: Bivalent rLP2086 (MenB-FHbp) was the first vaccine licensed in the United States through an accelerated approval process to prevent invasive meningococcal serogroup B (MnB) disease in individuals aged 10–25 years. Confirmatory immunogenicity and safety studies have been completed, with a large proportion of subjects having been enrolled in the United States. We present data from the safety evaluations of US subjects enrolled in one of 3 large phase 3 studies.

Methods: Healthy subjects aged 18 to <26 years were randomized to bivalent rLP2086 or saline at 0, 2, and 6 months. Safety variables, including local, systemic reactions and antipyretic use within 7 days after vaccination, and unsolicited adverse events (AEs), including immediate AEs, medically-attended AEs (MAEs), SAEs, and newly diagnosed chronic medical conditions (NDCMCs), were assessed.

Results: 2471 and 822 subjects received bivalent rLP2086 or saline, respectively; 1644 were from the United States (bivalent rLP2086, n=1232; saline, n=412). Local and systemic reactions were more common with bivalent rLP2086, mostly mild or moderate in severity and more frequent after the first vaccination. Injection-site pain (83.7%) and fatigue (59.0%) were the most common local and systemic reactions with bivalent rLP2086, respectively.

4.1% and 1.0% of bivalent rLP2086 and control recipients reported fever ≥38°C and 17.6% and 7.8%, respectively, used antipyretics. One bivalent rLP2086 recipient reported fever >40.0°C which lasted 1 day. Worsening of severity of local and systemic events with subsequent vaccinations was not observed. Few SAEs were reported overall among US subjects. Similar rates of AEs, SAEs MAEs, and NDCMC were reported between groups.

Conclusion: Bivalent rLP2086 was safe and tolerable among young US adults. Results were consistent with the safety profile observed in the global study.

ClinicalTrials.gov: NCT01352845

INTRODUCTION

- *Neisseria meningitidis* causes invasive meningococcal disease (IMD) most commonly in children <5 years of age, adolescents, and young adults.[1]
 - The highest carriage rates of *N meningitidis* are observed in adolescents and young adults.
- *N meningitidis* serogroup B (MnB) causes approximately one third of meningitis cases in the United States.[2]
 - Recent MnB outbreaks at university campuses in the United States[3] further emphasise the importance of the availability of effective MnB vaccines.
- Whereas capsular polysaccharide vaccines can prevent disease caused by 4 of the 5 major *N meningitidis* disease-causing serogroups (A, C, W, and Y),[1] this vaccine approach is not feasible to protect against MnB as the MnB polysaccharide is poorly immunogenic.[4]
- Bivalent rLP2086 (MenB-FHbp; Trumenba®) is a recombinant MnB vaccine containing a conserved, surface-exposed lipoprotein, LP2086 (a factor H binding protein [fHBP]); since fHBP sequences segregate into 2 immunologically distinct protein subfamilies (A and B),[5] the vaccine contains equal amounts of fHBP from each subfamily.[6]
 - Bivalent rLP2086 received accelerated approval in October 2014 by the US Food and Drug Administration for use in adolescents and young adults aged 10–25 years under Breakthrough Therapy designation.[7]
 - The Advisory Committee on Immunization Practices (ACIP) recommends that a licensed MnB vaccine be considered for all adolescents and young adults aged 16–23 years to provide protection against most strains causing MnB disease; the preferred age for vaccination is 16–18 years.[3]
 - The ACIP also recommends vaccination with a MnB vaccine for high-risk persons aged 10 years and older.[3]

OBJECTIVE

- Safety and tolerability data from US subjects aged 18 to <26 years are presented to confirm and extend the data that were the basis for accelerated approval of bivalent rLP2086 in the United States.

METHODS

Study Design

- This was a phase 3, randomised, placebo-controlled, observer-blinded, multicenter study of bivalent rLP2086 in subjects aged 18 to <26 years (ClinicalTrials.gov identifier: NCT01352845).
 - Subjects were randomly assigned in a 3:1 ratio to receive 120 µg bivalent rLP2086 vaccine or saline at 0, 2, and 6 months.
- Immunogenicity and safety results for the global study have been presented previously.[8,9]
- Immunogenicity data for US subjects from another phase 3 study in adolescents are presented in a companion poster.

Safety Analysis

- Safety was evaluated for all subjects who received ≥1 dose (safety population) and who had safety data available.
- Reactogenicity events included local reactions (redness, swelling, and injection site pain) and systemic events (fever, vomiting, diarrhoea, headache, fatigue, chills, muscle pain other than at the injection site, and joint pain), as well as antipyretic use, that were collected using an electronic diary (e-diary) for 7 days after each vaccination.
- Adverse events (AEs) were assessed through 1 month after the last vaccination.
 - Immediate AEs were assessed within 30 minutes after vaccine administration.
- Serious AEs (SAEs), medically attended AEs (MAEs), and newly diagnosed chronic medical conditions (NDCMCs) were assessed through the final telephone contact 6 months after the last vaccination.
 - MAEs were defined as nonserious AEs that resulted in evaluation at a medical facility.
 - NDCMCs were defined as previously unidentified diseases or medical conditions expected to have persistent or long-lasting effects.

RESULTS

Subjects

- Subject disposition was comparable between groups **(Table 1)**.
 - 1644 subjects from the United States (50% of the total study population; N=3293) aged 18 to <26 years received ≥1 dose and were included in the US safety population (bivalent rLP2086, n=1232; saline, n=412).

Table 1. Subject Disposition

n (%)	Bivalent rLP2086	Saline	Total
Randomised, n	1235 (100)	413 (100)	1648 (100)
Withdrew before vaccination	3 (0.2)	1 (0.2)	4 (0.2)
Vaccinated			
Dose 1	1232 (99.8)	412 (99.8)	1644 (99.8)
Dose 2	975 (79.0)	340 (82.3)	1315 (79.8)
Dose 3	742 (60.1)	269 (65.1)	1011 (61.4)
Completed vaccination phase	702 (56.8)	256 (62.0)	958 (58.1)
Withdrew during vaccination phase	530 (43.0)	156 (37.8)	686 (41.6)
Completed study	670 (54.3)	241 (58.4)	911 (55.3)

Local Reactions

- Pain at the injection site, redness, and swelling were reported more frequently with bivalent rLP2086 than with saline **(Figure 1)**.
 - Most local reactions were mild to moderate in severity.
 - Worsening of severity of local reactions with subsequent vaccinations was not observed

Figure 1. Local Reactions at the Injection Site Occurring Within 7 Days After Any Vaccination in US Subjects

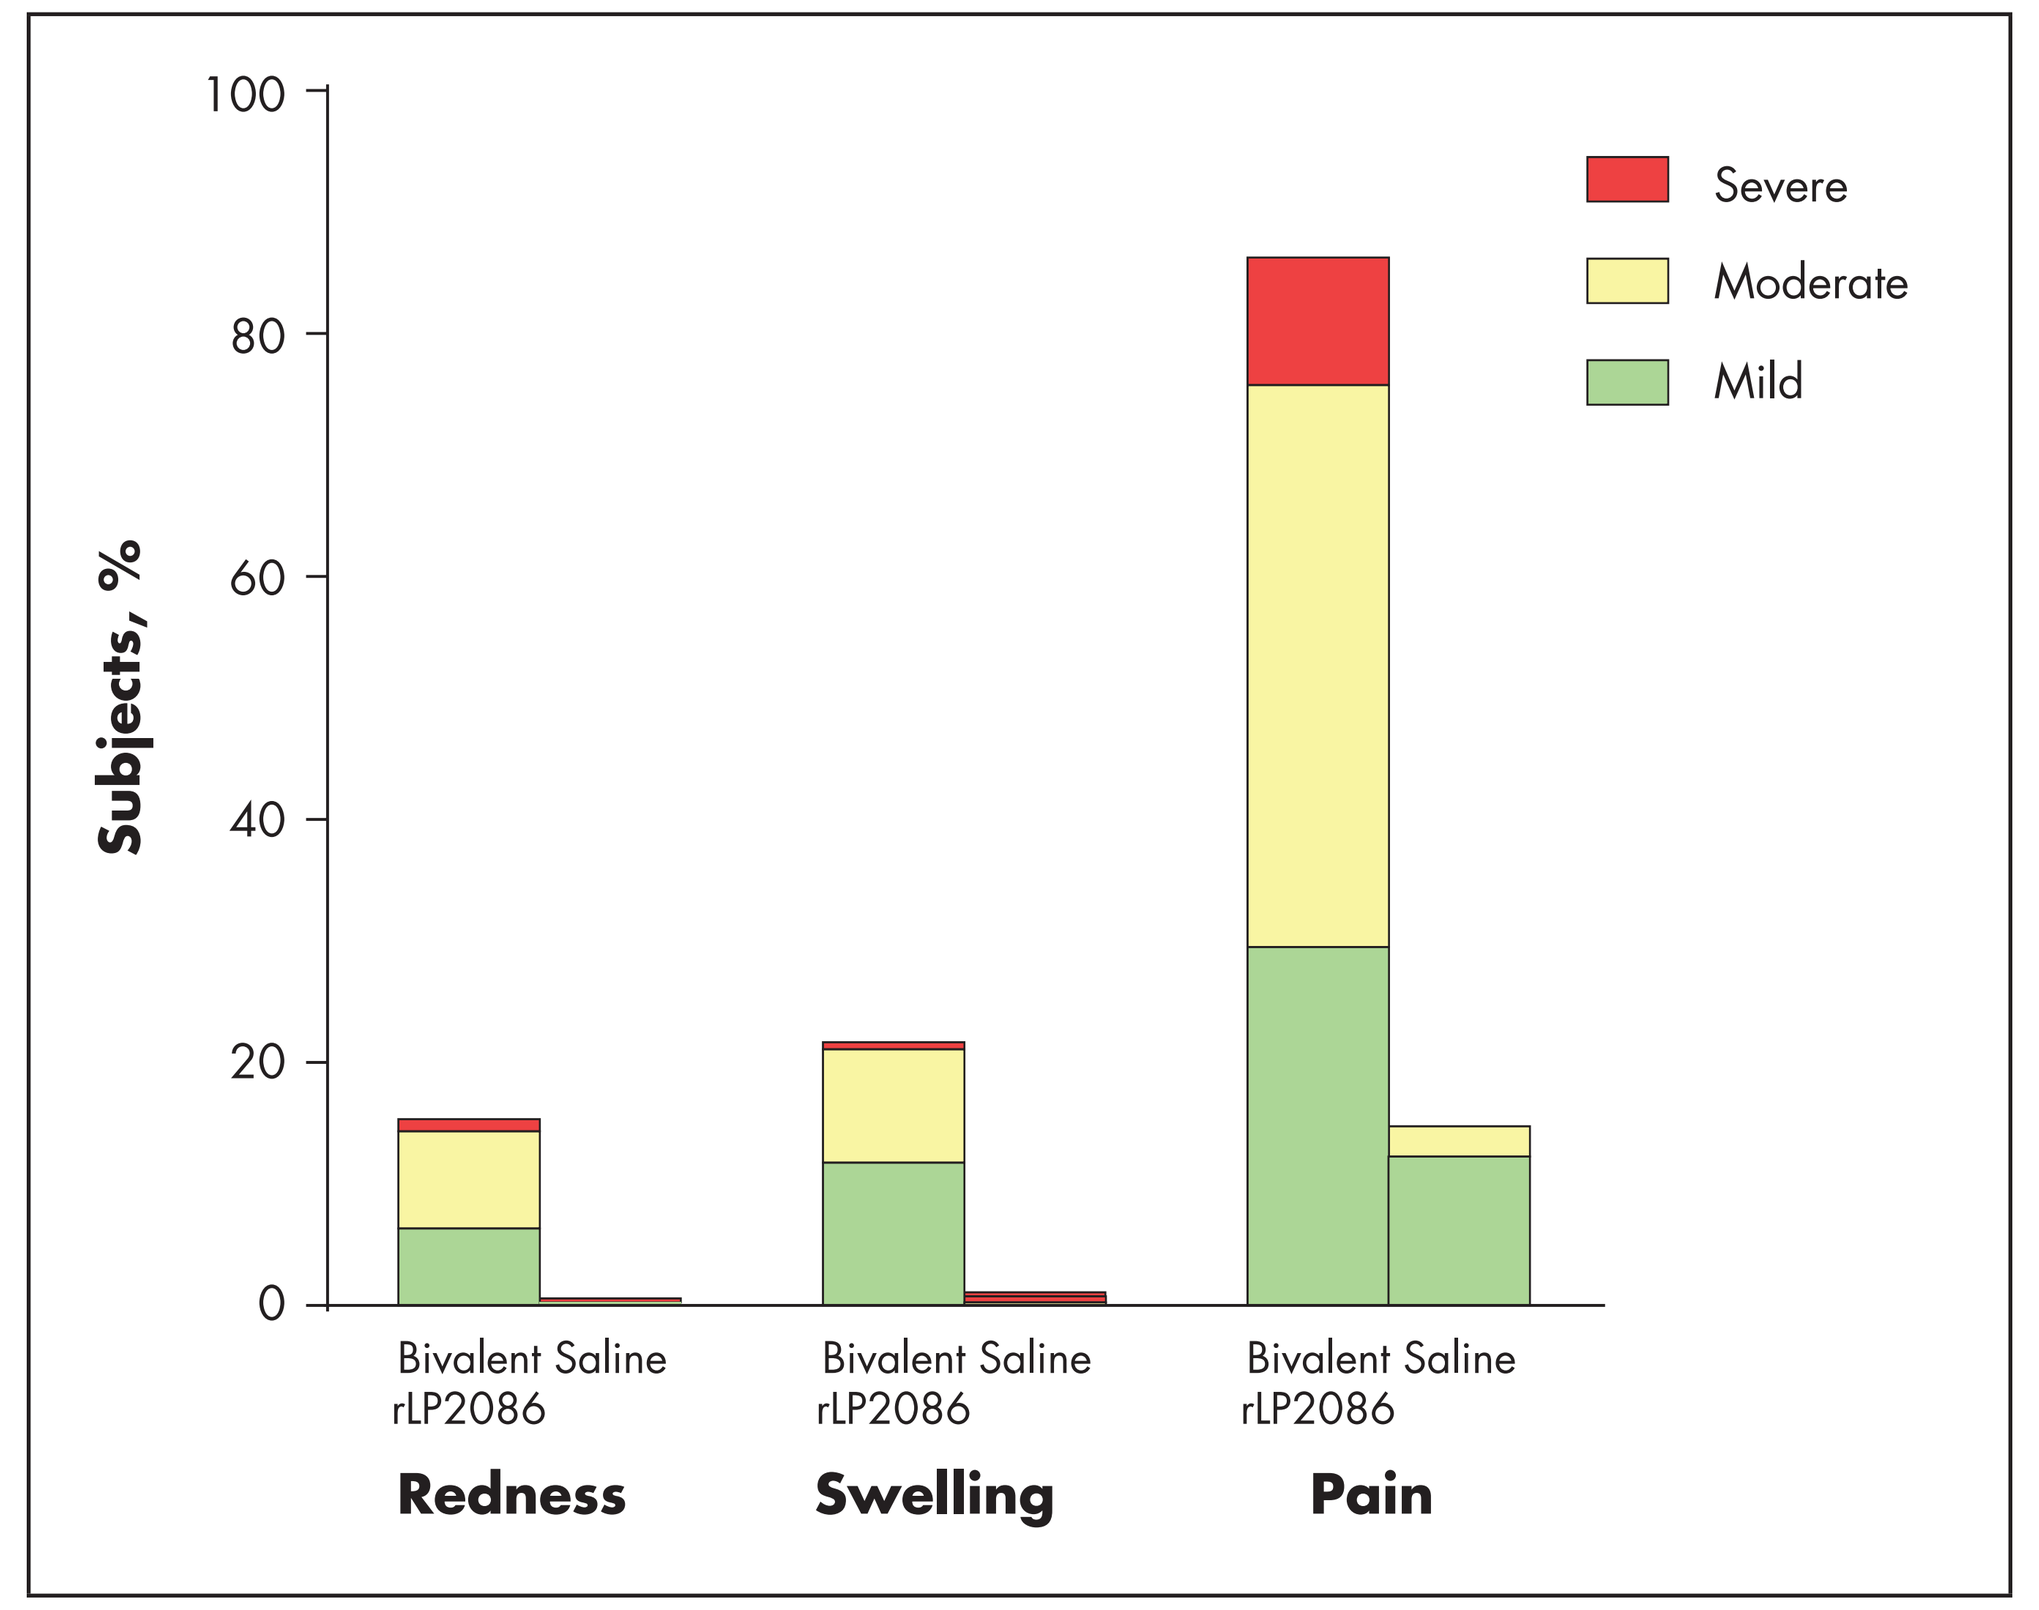
Subjects, %
0
20
40
60
80
100
Severe
Moderate
Mild
Bivalent rLP2086
Saline
Redness
Swelling
Pain

Systemic Events

- Systemic events were generally mild or moderate in severity and were more frequent after the first vaccination.
- Fatigue and headache were the most commonly reported systemic events among bivalent rLP2086 recipients **(Figure 2)**.
 - Other commonly reported systemic events were muscle pain, joint pain, chills, and diarrhea.

Figure 2. Systemic Events in US Subjects Occurring Within 7 Days After Any Vaccination

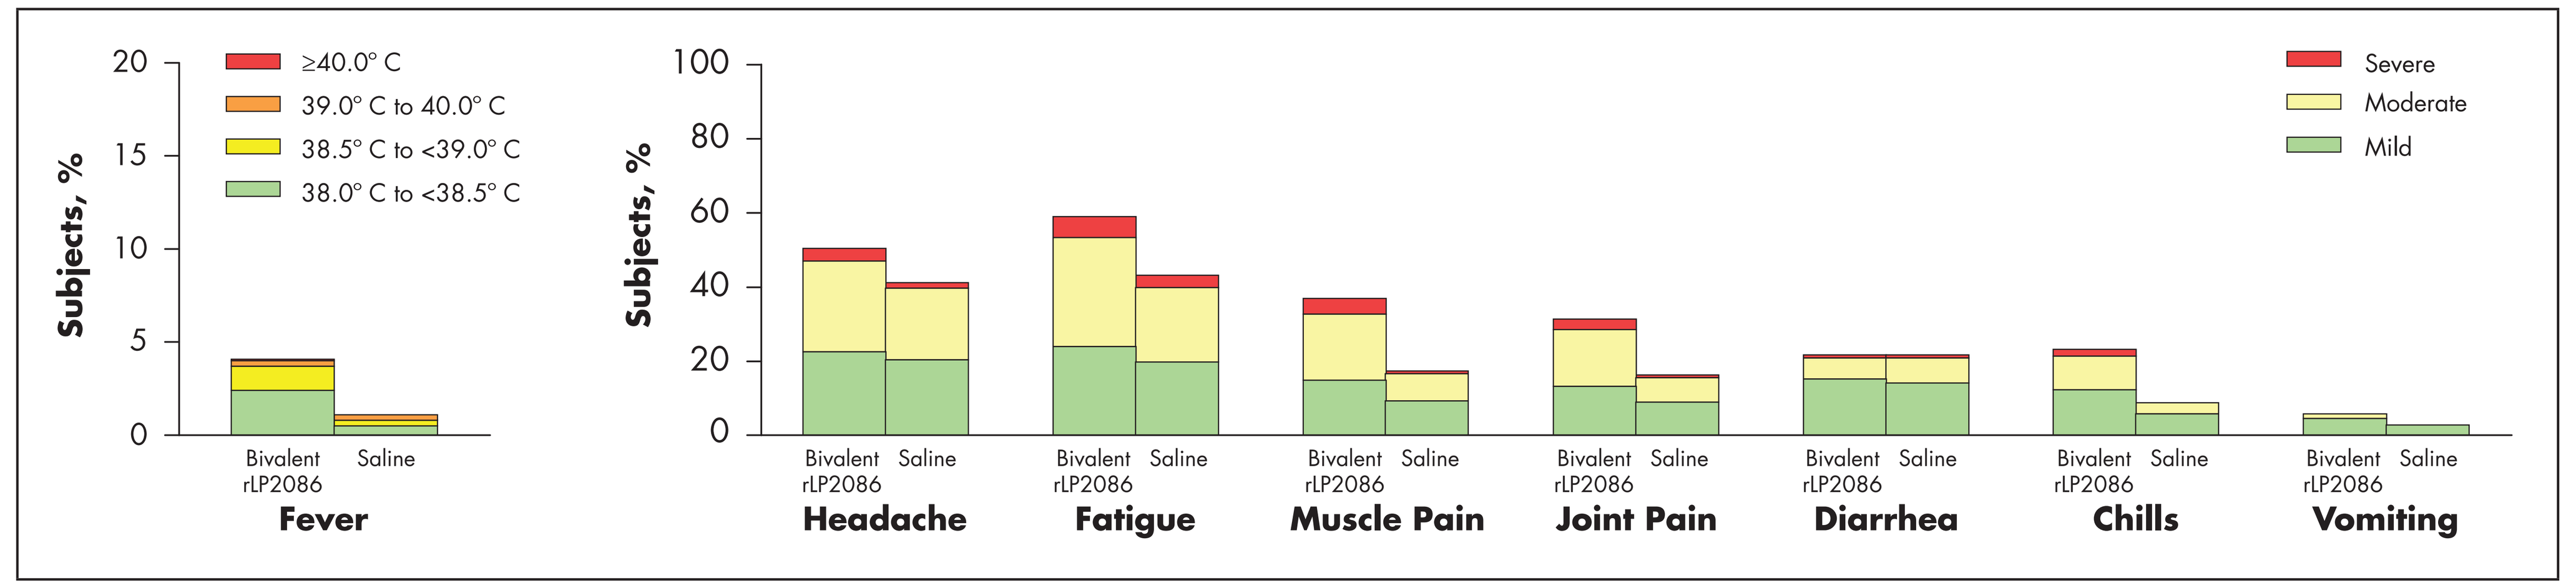
Subjects, %
0
5
10
15
20
≥40.0° C
39.0° C to 40.0° C
38.5° C to <39.0° C
38.0° C to <38.5° C
Bivalent rLP2086
Saline
Fever
0
20
40
60
80
100
Severe
Moderate
Mild
Headache
Fatigue
Muscle Pain
Joint Pain
Diarrhea
Chills
Vomiting

- 4.1% and 1.0% of bivalent rLP2086 and saline recipients, respectively, reported fever ≥38°C.
 - 1 bivalent rLP2086 recipient reported fever >40.0°C on day 1 after dose 3, which lasted 1 day. The subject did not report fever or other systemic event after dose 1 or 2.
 - Antipyretic medication was used by 17.6% of subjects in the bivalent rLP2086 group and 7.8% in the saline group.
- Worsening of severity of systemic events with subsequent vaccinations was not observed.

Adverse Events

- The proportion of subjects who reported AEs of any severity occurring during the vaccination phase was similar in the bivalent rLP2086 and saline groups **(Table 2)**.

Table 2. Adverse Events

n (%)	Bivalent rLP2086 (N=1232)[a] n,[b] %	Saline (N=412)[a] n,[b] %	P-value
AE (during vaccination phase)	318 (25.8)	104 (25.2)	0.845
Related	53 (4.3)	11 (2.7)	0.184
AE (within 30 days of any vaccination)	223 (18.1)	66 (16.0)	0.370
Related	52 (4.2)	11 (2.7)	0.183
Immediate AE (within 30 minutes of any vaccination)	11 (0.4)	7 (0.9)	ND
SAE (throughout the study)	20 (1.6)	4 (1.0)	ND
Related	1 (0.1)	0	ND
NDCMC (throughout the study)	6 (0.5)	1 (0.2)	0.688
Related	0	0	ND
MAE (throughout the study)	178 (14.5)	58 (14.1)	0.935
Related	4 (0.3)	1 (0.2)	>.999

AE=adverse event; MAE=medically attended adverse event; ND=not determined; NDCMC=newly diagnosed chronic medical condition; SAE=serious adverse event.
[a]Number of subjects in the safety population
[b]Number of subjects with at least 1 event

- No notable difference in immediate AEs between bivalent rLP2086 and saline recipients was observed.
- Similar rates of SAEs, MAEs, and NDCMCs were reported between groups **(Table 2)**.
 - Overall, few SAEs were reported.
 - No NDCMCs and few SAEs or MAEs were considered vaccine-related.
 - More vaccine-related AEs (during the vaccination phase and within 30 days of vaccination) were reported in the bivalent rLP2086 group than in the saline group.
 - The difference is due to an excess of reactogenicity events in the bivalent rLP2086 group reported outside of the e-diary and mainly includes injection-site pain and headache.

CONCLUSIONS

- **Results of this large phase 3 trial demonstrated that bivalent rLP2086 is safe and tolerable for use in healthy young adults aged 18 to <26 years in the United States.**
 - Reactogenicity events were mostly mild to moderate in severity.
- **Results were consistent with the safety profile of bivalent rLP2086 observed in a large, global study[8,9] and in other clinical trials.[10-17]**
- **This study extends the phase 2 safety data that were the basis for accelerated approval of bivalent rLP2086 in the United States.**

REFERENCES

1. Cohn AC, et al. *MMWR Recomm Rep.* 2013;62:1-28.
2. Centers for Disease Control and Prevention. Active Bacterial Core Surveillance (ABCs) Report, Emerging Infections Progam Network Neisseria meningitidis, 2013-provisional. Available at: http://www.cdc.gov/abcs/reports-findings/survreports/mening13.pdf. Accessed April 13, 2015.
3. MacNeil JR, et al. *MMWR Morb Mortal Wkly Rep.* 2015;64:1171-1176.
4. Finne J, et al. *Lancet.* 1983;2:355-357.
5. Murphy E, et al. *J Infect Dis.* 2009;200:379-389.
6. TRUMENBA® (Meningococcal Group B Vaccine). Full Prescribing Information, Wyeth Pharmaceuticals Inc, a subsidiary of Pfizer Inc, Philadelphia, PA, 2014.
7. Folaranmi T, et al. *MMWR Morb Mortal Wkly Rep.* 2015;64:608-612.
8. Ostergaard L, et al. Safety of Bivalent rLP2086, A Meningococcal Serogroup B Vaccine, in Young Adults: Results from a Phase 3 Trial. Presented at: European Society for Paediatric Infectious Diseases; May 10-14, 2016; Brighten, UK.
9. Ostergaard L, et al. Phase 3 Trial of Immunogenicity of Bivalent rLP2086, A Meningococcal Serogroup B Vaccine, in Young Adults: Bactericidal Activity Against a Panel of Antigenically Diverse Strains. Presented at: European Society for Paediatric Infectious Diseases; May10-14, 2016; Brighton, UK.
10. Marshall HS, et al. *Vaccine.* 2013;31:1569-1575.
11. Nissen MD, et al. *Pediatr Infect Dis J.* 2013;32:364-371.
12. Richmond PC, et al. *Lancet Infect Dis.* 2012;12:597-607.
13. Richmond PC, et al. *Vaccine.* 2012;30:6163-6174.
14. Senders S, et al. *Pediatr Infect Dis J.* 2016;35:548-554.
15. Ostergaard L, et al. *Vaccine.* 2016;34:1465-1471.
16. Reiner DM, et al. *Vaccine.* 2016;34:809-813.
17. Vesikari T, et al. *J Pediatric Infect Dis Soc.* 2016;5:180-187.

ACKNOWLEDGMENTS

This work was sponsored by Pfizer Inc. Editorial/medical writing support was provided by Susan DeRocco, PhD, of Complete Healthcare Communications, LLC, and was funded by Pfizer Inc.

Presented at IDWeek 2016; October 26–30, 2016; New Orleans, LA