1982

WCK 5222 [Cefepime-Zidebactam, FEP-ZID]: Mechanistic Basis Behind Novel β-lactam–β-lactam Enhancer Combination Against Metallo-β-lactamase (MBL)-producing *E. coli* (EC) *K. pneumoniae* (KP), *P. aeruginosa* (PA) and its Impact on Therapeutically Relevant Bactericidal Exposures Assessed Through *in vitro* Pharmacodynamic Modelling (IVPM) and Mouse Lung Eradication Studies

B. Moya[1], IM. Barcelo[1], A. Udaykar[2], S. Bhagwat[2], M. Patel[2], G. Bou[3], A. Oliver[1]

[1]H. Son Espases, Palma de Mallorca, Spain; [2]Wockhardt Research Centre, Aurangabad, India; [3]Complejo Hospitalario Universitario A Coruña, Spain

Institut d'Investigació Sanitària de Palma IdISPa

WOCKHARDT LIFE WINS DISCOVERY

www.wockhardtdiscovery.com
Download: app.wockhardtdiscovery.com/install.ipa
Connect: bd@wockhardtdiscovery.com

INTRODUCTION AND PURPOSE

- WCK 5222 [Cefepime (FEP) + WCK 5107 (Zidebactam, ZID)] is a combination of Gram-negative antibiotics (completed Phase I studies in US) - FEP - a well-established 4th generation cephalosporin and ZID - a novel β-lactam enhancer antibiotic.
- ZID is a non-β-lactam, dual acting 'β-lactam Enhancer' antibiotic which in combination with FEP overcomes the challenge posed by currently untreatable MDR pathogens including MBL and OXA carbapenemases expressing Enterobacteriaceae (ENTs), *Psuedomonas aeruginosa* (PA) and *Acinetobacter baumannii* (AB). Distinctiveness of its mechanism of action lies in the fact that owing to its pan- β-lactamase stable feature, ZID is capable of effectively engage PBP2 across Gram-negatives including PA and AB[1,2]. The minimum concentration at which ZID induces spheroplastation of cell is termed as minimum spheroplasting concentrations (**MSC**) which is invariably sub-MIC[2]. Interesting features of FEP include optimal permeation, relative stability towards several β-lactamases and high-affinity binding to PBP3 across Gram-negatives. As a result even in the presence of FEP-impacting β-lactamases, FEP is able to transiently cause elongation of cells owing to compromise in PBP3 function. The minimum concentration at which FEP induces elongation of cell is termed as minimum elongation concentrations (**MEC**)[2]. Our studies have shown that ZID-mediated PBP2 dysfunction leads to severe perturbation of periplasmic physiology leading to the formation of fragile spheroplasts, consequent leakage of β-lactamases and modulation of outer membrane linked resistance mechanisms such as efflux and uptake. As a result, even against MDR Gram-negatives expressing ZID non-inhibitable β-lactamases such as MBL and OXA-carbapenamases, ZID enhances the spectrum and cidal potency of FEP by bringing in high-affinity PBP2 binding and in conjunction with FEP's PBP3 binding causes simultaneous inactivation of multiple PBPs. Cidality studies have shown that a combination of FEP at MEC and ZID at MSC, causes 2-3 log kill of various MDR Gram-negative pathogens within 4-6h[1,2]. Among β-lactams, only carbapenems are endowed with rapid cidal action owing to their primacy to PBP2 in addition to binding to other essential PBPs. Therefore it seems that, unhindered PBP2 binding is central in conferring rapid cidal action to a β-lactam. One of the most noteworthy consequence of the rapid cidal action of carbapenems is translated into PD gain leading to a substantial reduction in T>MIC required to cause *in vivo* bactericidal effect which in turn provides favourable PK-PD culminating into superior clinical outcome in serious infections. However, carbapenems are vulnerable to carbapenemases and are unable to exert antibacterial effect against carbapenemase-expressing organisms which have emerged as a major unmet need justifiably categorized as 'urgent' threat level pathogen by CDC.
- In the current study, we evaluated the impact of PBP2-mediated rapid cidality of WCK 5222 on the *in vitro*/*in vivo* PK/PD outcome against MBL expressing ENTs, and PA strains including those expressing NDM. This is the first report of FEP-based combination demonstrating therapeutic potential against NDM-expressing PA. Previously, we have reported the efficacy of WCK 5222 against NDM-expressing ENTs and VIM expressing PA[3]. Other NDM active β-lactam-based combination, -aztreonam-avibactam is reported to be active against MBL-expressing ENT[4], however data pertaining to MBL expressing PA are limited[5].
- The range of studies described here include determination of PBP binding of FEP and ZID, time-kill studies, IVPM and lung eradication studies in neutropenic mice. Further, dose-fractionation studies aimed to establish the efficacy determining PK-PD parameter and its magnitude employed highly resistant NDM-expressing PA strain with WCK 5222 MIC of 32 µg/mL (Year 2015 surveillance study: WCK 5222 PA MIC_{90} is 4µg/mL)[6].

METHODS

In vitro studies

PBP binding assay

- The PBP binding affinities for FEP and ZID were determined using membrane isolations from wild-type strains (EC, KP and PA). Membranes containing the PBPs of wildtype EC, KP, PA were obtained according to the protocols described previously[7,8]. Briefly, PBP containing solution was incubated (30 min, 37ºC) in the presence of increasing concentrations (0.0156-2 mg/L) of ZID or reference drugs – FEPand MEC and labelled with BOCILLIN FL (25 µM). The reaction mixtures were denatured and PBPs separated by SDS PAGE. LabelledPBPs were visualised and IC50s determined (Typhoon™FLA 9500 biomolecular imager and Image Quant™ TL- GE Healthcare Bio-Sciences AB Björkgatan 30 751 84 Uppsala). All experiments were performed in triplicate.

VIM-2 purification and inhibition by ZID

- The VIM-2 β-lactamase was purified from EC BL21(DE3) pLys cells carrying pET-24a(+)-blaVIM-2-S21, as previously described[9]. The inhibition of VIM-2 by WCK ZID was analysed by determining the Ki apparent (Ki app) value using an Agilent (Santa Clara, CA) 8453 diode array spectrophotometer. All reactions were conducted in 10 mM phosphate-buffered saline (PBS) at pH 7.4 at room temperature. A direct competition assay between nitrocefin (reporter substrate) and ZID with VIM-2 was conducted using pseudo first order conditions under steady-state. The three reaction components were mixed manually, and the reaction velocity for the first 10s of the reaction was collected. The data were linearized by plotting the inverse initial steady-state velocities (1/V0) vs ZID concentration. Ki app was determined by dividing the value for the y-intercept by the slope of the line and correcting for the use of nitrocefin, as previously described.

MIC determination

- Standalone antibacterial activity of ZID precludes its use at fixed 4 or 8 µg/mL thus MICs of FEP-ZID combination were performed at 1:1 ratio. MICs for comparator agents FEP, Imipenem (IPM) and Meropenem (MEM) were determined by Broth Microdilution method as per reference CLSI M07-A10 guidelines against 2 MBL-producing ENT and PA strains employed in i*n vitro/in vivo* studies. IPM and MEM MICs were also determined in the presence of EDTA (a known MBL inhibitor).

MSC & MEC determination

- The minimum concentration of FEP and ZID required for cell elongation (MEC) and spheroplast formation (MSC), respectively were determined by microscopy. Briefly, increasing concentrations of FEP or ZID were added to actively growing cultures set at the desired inoculum (5×10^6-10^7 CFU/mL). Flasks were incubated (37°C, 120 rpm) and cultures were observed under the microscope (oil immersion-100×, 1000× magnification) at 2-3h for monitoring morphological changes induced.

Time-kill kinetics

- Time-kill studies were initiated by adding drugs to CA-MHB (Cation-adjusted Mueller Hinton Broth) containing exponentially growing cultures at a starting count of 6.19-6.69 log_{10}CFU/mL and incubated at 37°C under shaking conditions (120 rpm). Viability counts at various time points were determined by plating serially diluted (1:10) culture on Tryptone Soya Agar medium (Hi-Media, India). Two NDM-producing EC M44, KP NCTC 13443, and one VIM-producing PA NCTC 13437 were employed in the time-kill studies.Time-kill studies involving carbapenems were also undertaken in the presence of EDTA (MBL inhibitor) to manifest the cidal effect of carbapenems against MBL-expressing pathogens.

One compartment IVPM studies

- IVPM studies were conducted in EC M 44 (NDM, TEM), KP NCTC 13443 (NDM-1, CMY, SHV, TEM, DHA1/2) and PA NCTC 13437 (VIM-10, VEB-1, OXA-10, CTX-M). The IVPM model employed in the current study consisted of a central compartment wherein the drug PK simulations were generated by introducing the drugs at their clinical C_{max} in CA-MHB which was pre-inoculated with test organisms (starting inoculums of 7-9×10^5, 1×10^6 and 2.5×10^6 CFU/mL for PA NCTC 13437, EC M44 and KP NCTC 13443, respectively). Peristaltic pump was utilized to deliver the growth medium at a flow rate that generated clinical half-life of drug in central compartment. All the drugs were infused with the help of infusion pumps in central compartment over the period of one hour in order to have a clinical C_{max} and half-life of respective drug as shown in **Table 1**. The dosing was done every q12h (BID) or q8h (TID). CFU samples were collected every hour up to first 8h and subsequently after every 2h till the termination of experiment (limit of detection: 3 log_{10} CFU/mL). LC-MS/MS analysis of samples from initial pilot IVPM studies without the organisms confirmed the attainment of target concentration-time profile. The concentrations were within 5-10% of the target levels. The minimum detection limit of FEP and ZID was 0.625 µg/mL and 0.312 µg/mL, respectively.

Table 1. FEP and ZID simulated human PK based on Phase 1 studies

Drug	Infusion time	Human corresponding dose	C_{max}(µg/mL)	Half-life (h)
FEP	60 min	2g ,TID	140	2
ZID	60 min	1g, TID	60	2
ZID	60 min	0.5g, TID	30	2
MEM	30 min	1g, TID	56	1

FEP: cefipime; ZID: zidebactam; MEM: meropenem.

In vivo studies

Murine lung infection dose fractionation study with NDM-expressing PA S814

- Lung eradication studies were conducted in neutropenic mice (cyclophosphamide-150 and 100 mg/kg at -4 and -1 days of infection, respectively). Lung infection was initiated by intranasal instillation of 10^7 log_{10} CFU/mL for all the strains under light anaesthesia. Treatment was initiated 2h post-infection (2h count 6.0±0.15 log_{10} CFU/lung) with FEP 50 mg/kg q2h (Total Daily Dose, TDD- 600 mg/kg) and ZID (TDD - 25, 50,100, 250, 400, 550 mg/kg) was fractionated q2h and q4h for 1 day. IPM and MEM (TDD: 240 mg/kg, fractionated as q2h) were co-administered with cilastatin at 1:1 ratio. This study design allowed estimating the threshold concentration of ZID. In order to determine the %*f*T >MIC for FEP, FEP TDD in the range of 300, 450, 600 and 900 mg/kg were fractionated as q2h, q4h and q6h in combination with ZID TDD of 100 mg/kg administered as q2h. Bactericidal effect in lung was assessed 27h post-infection in all the treatment groups.

RESULTS AND DISCUSSION

PBP inhibition

- ZID showed exclusive PBP 2 binding in all the organisms studied as given in Table 2. For EC and PA, ZID exhibited high affinity to PBP2 which was comparable to Mecillinam (MEC) - a known PBP2 binding penicillin. However , ZID showed 2 times higher affinity in KP as compared to MEC. As reported, among all the agents tested FEP showed multiple PBP binding with primary targeting towards PBP3 followed by 2, 1a and 1b for EC. Ironically, although FEP has been in clinical use for ~20 years, its PBP binding profile in KP is not reported. This study shows that FEP binds to multiple PBPs in KP, similar to EC. However for PA, FEP showed primacy towards PBP3 followed by PBP 1a/1b. Taking into account the PBP binding profile of FEP and ZID, WCK 5222 is expected to concurrently interfere in the function of most essential PBPs.

Table 2: PBP binding profile of ZID, MEC and FEP in EC, KP and PA

PBP's	PBP IC_{50} (mg/L) EC MG1655			KP 52145			PA PAO1		
	ZID	MEC	FEP	ZID	MEC	FEP	ZID	MEC	FEP
1A	>2	>2	1.74 ± 0.15	>2	>2	1.2 ± 0.47	>4	>4	0.12 ± 0.01
1B	>2	>2	1.82 ± 0.08	>2	>2	>2	>4	>4	0.82 ± 0.07
2	0.06 ± 0.01	0.06 ± 0.03	0.69 ± 0.32	0.08 ± 0.02	0.18 ± 0.07	0.74 ± 0.27	0.26 ± 0.06	0.19 ± 0.02	2.71 ± 0.92
3	>2	>2	0.13 ± 0.05	>2	>2	0.19 ± 0.08	>4	>4	0.15 ± 0.07
4	>2	>2	>2	>2	>2	>2	>4	>4	2.52 ± 0.27

ZID: zidebactam; MEC: mecillinam; FEP: cefepime.

β-lactamase inhibition

- The investigations undertaken at Dr. Bonomo's lab have shown that ZID is a potent inhibitor of ESBLs, Class C and KPC β-lactamases[10]. However, ZID is not an inhibitor of VIM (Ki in µM: >300).

MICs against MBL-producing ENT and PA

- FEP-ZID (1:1) MICs for EC M44, KP NCTC 13443, PA NCTC 13437 and PA S814 were 0.12, 4, 16 and 32 µg/mL, respectively. The presence of genes encoding β-lactamases was established by PCR. Higher IPM/MEM MICs against MBL-expressing EC, KP and PA and lowering of their MICs in the presence of EDTA demonstrates the adequate expression of MBL.

Table 3: Activity of WCK 5222 against the organisms employed in *in vitro* cidality, IVPM and *in vivo* studies

Organisms	Resistotypes (enzymatic/non-enzymatic)	MIC/MEC/MSC (µg/mL) FEP MIC	FEP MEC	ZID MIC	ZID MSC	FEP-ZID (1:1)	IPM	IPM - EDTA	MEM	MEM-EDTA
EC M44	NDM, TEM	>32	4	0.25	0.25	0.25	16	0.12	32	0.12
KP NCTC 13443	NDM-1, CMY, DHA1/2, SHV, TEM,	>512	8	>512	4	4	128	1	512	2
PA NCTC 13437	VIM-10, VEB-1, OXA-10, CTX-M	256	16	16	8	16	128	16	256	16
PA S 814	NDM	>512	16	32	16	32	>256	16	>256	16

MIC: minimum inhibitory concentration; MEC: minimum elongation concentration; MSC: minimum spheroplast concentration.
For stand-alone ZID, trailing endpoints were observed against KP 13443.

Time-kill studies

- For EC M44, FEP-ZID combination of MEC and MSC triggered rapid synergistic killing to the tune of 3-log over initial count and 1.5-log over the kill observed with standalone ZID at 2h (Figure 1a). Interestingly, WCK 5222 provided a higher rate and extent of kill as compared to IPM combined with EDTA. WCK 5222 caused similar comparable extent of killing was observed in both KP NCTC 13443 and PA 13437 strains.

Figure 1: Time-kill studies of FEP-ZID and comparators against EC M44 (NDM, TEM)

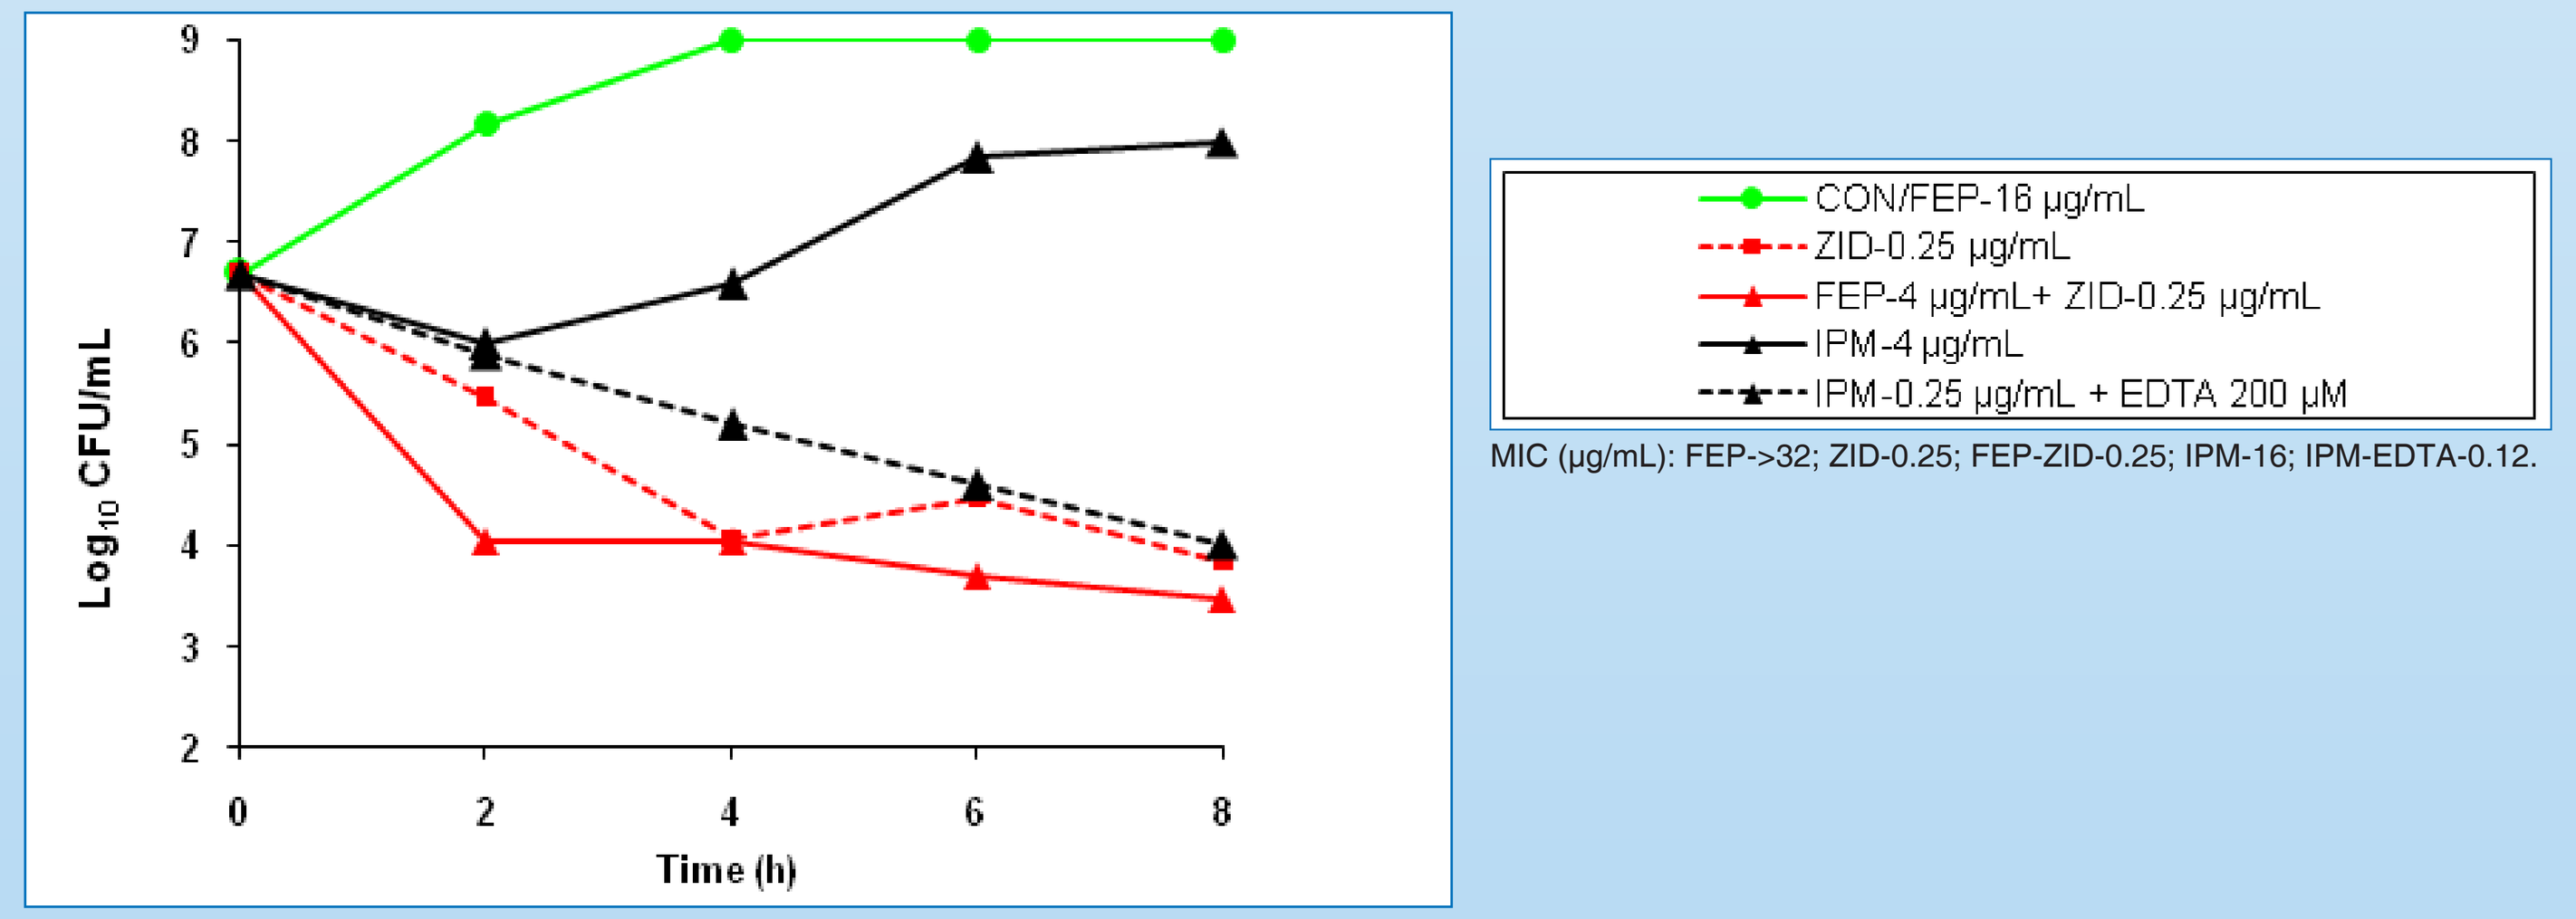

Log_{10} CFU/mL
Time (h)
CON/FEP-16 µg/mL
ZID-0.25 µg/mL
FEP-4 µg/mL+ ZID-0.25 µg/mL
IPM-4 µg/mL
IPM-0.25 µg/mL + EDTA 200 µM

MIC (µg/mL): FEP->32; ZID-0.25; FEP-ZID-0.25; IPM-16; IPM-EDTA-0.12.

One compartment IVPM studies

Figure 2: *In vitro* Pharmacodynamic action of WCK 5222

2a) EC M44 (NDM, TEM)

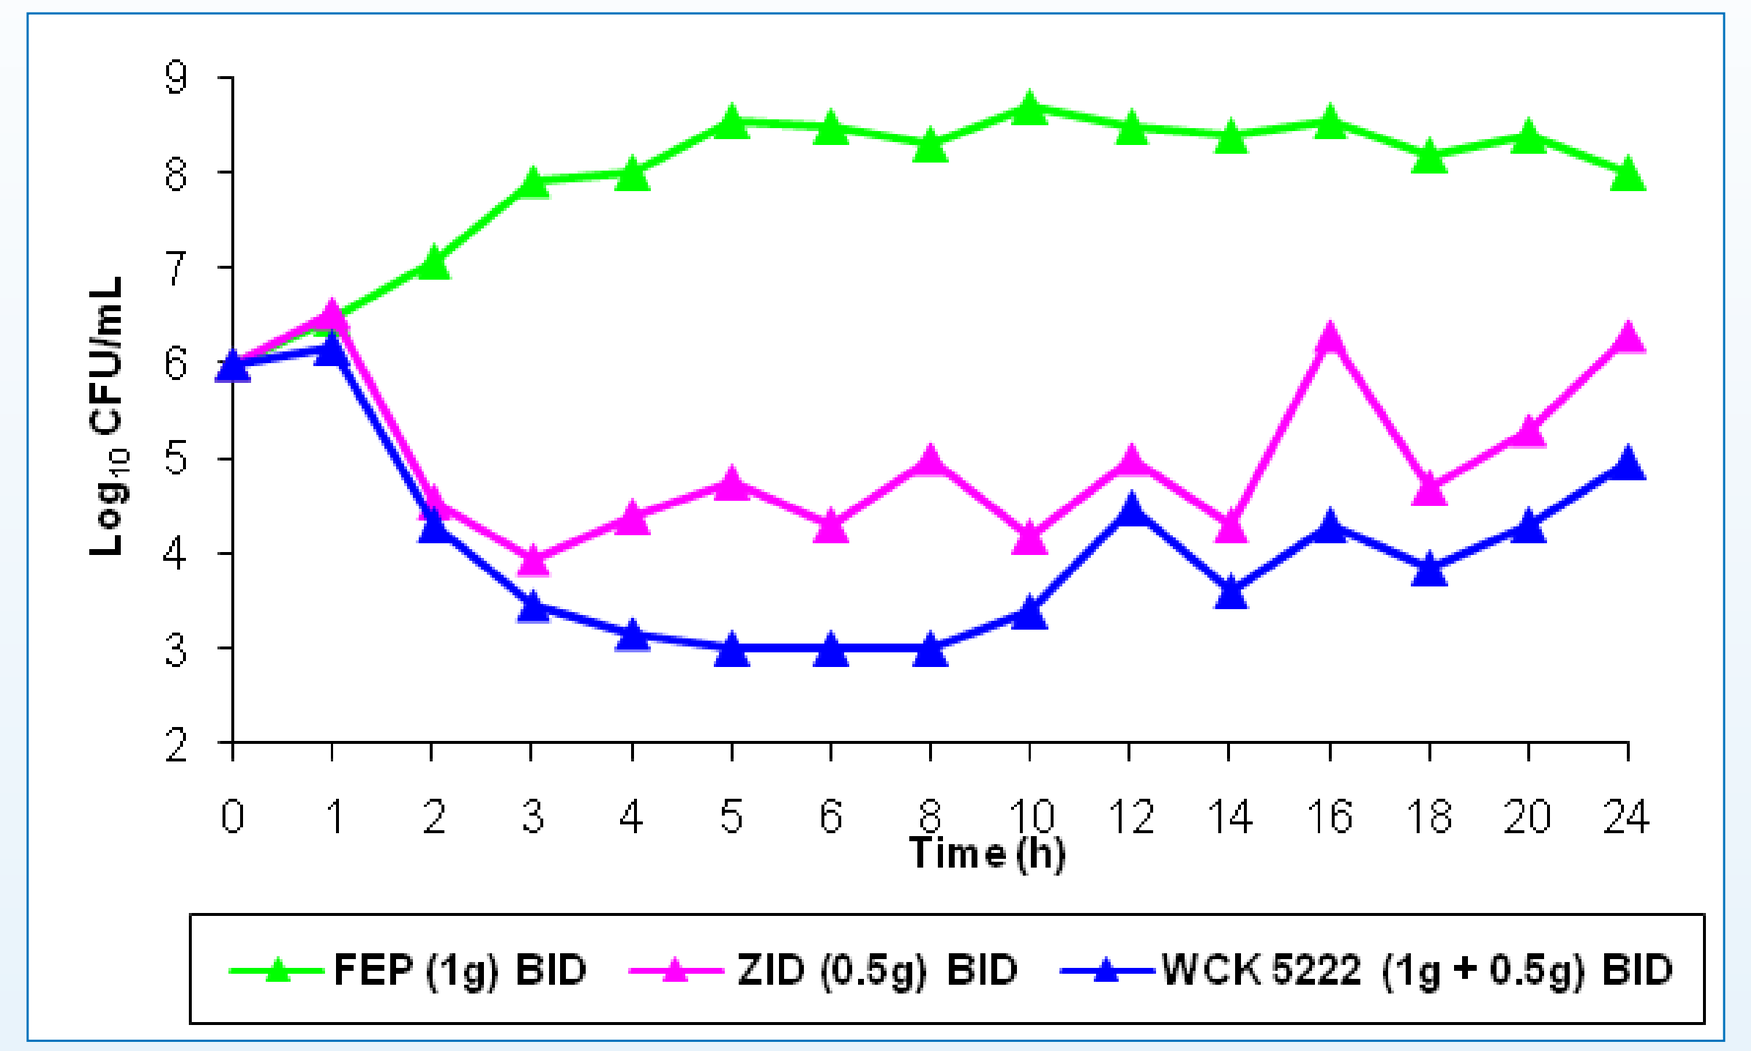

Log_{10} CFU/mL
Time (h)
FEP (1g) BID
ZID (0.5g) BID
WCK 5222 (1g + 0.5g) BID

2b) KP NCTC 13443 (NDM-1, CMY,DHA1/2, SHV, TEM)

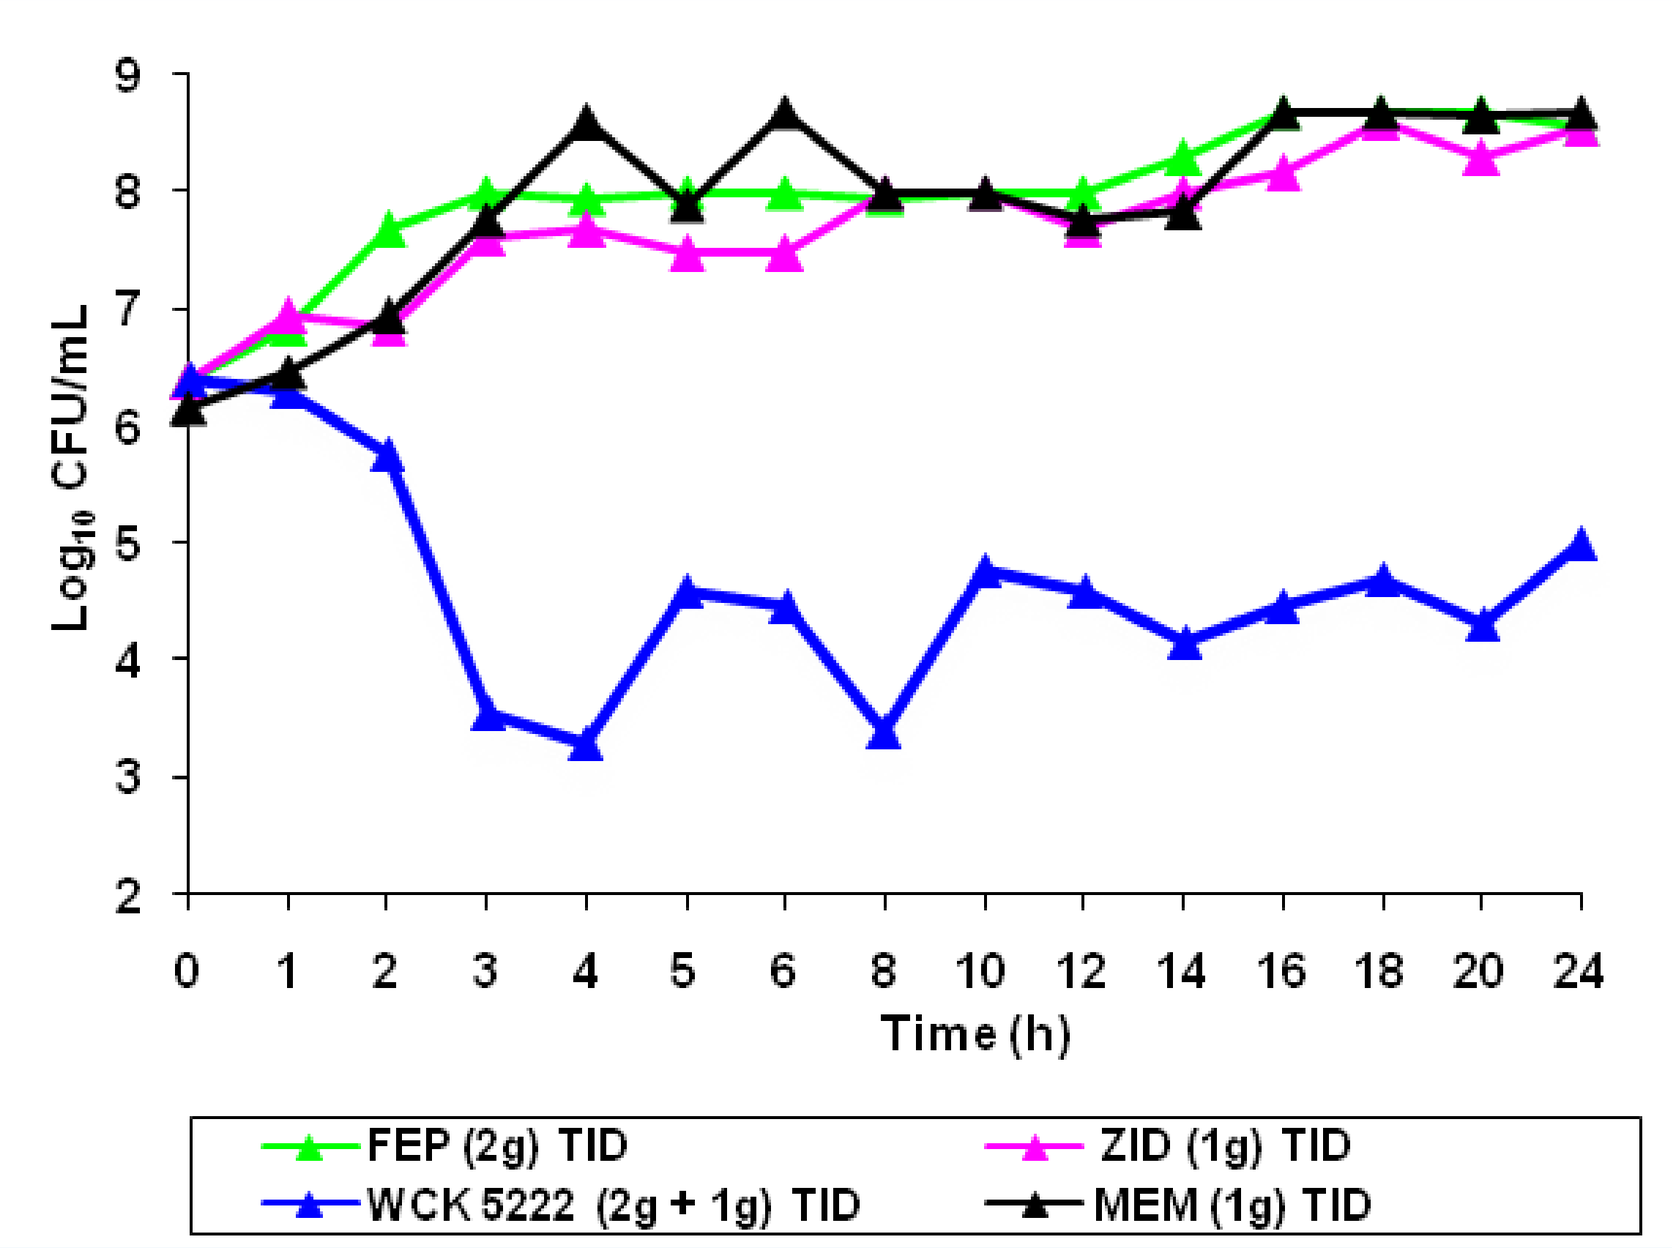

Log_{10} CFU/mL
Time (h)
FEP (2g) TID
ZID (1g) TID
WCK 5222 (2g + 1g) TID
MEM (1g) TID

2c) PA NCTC 13437 (VIM-10, VEB-1, OXA-10, CTX-M)

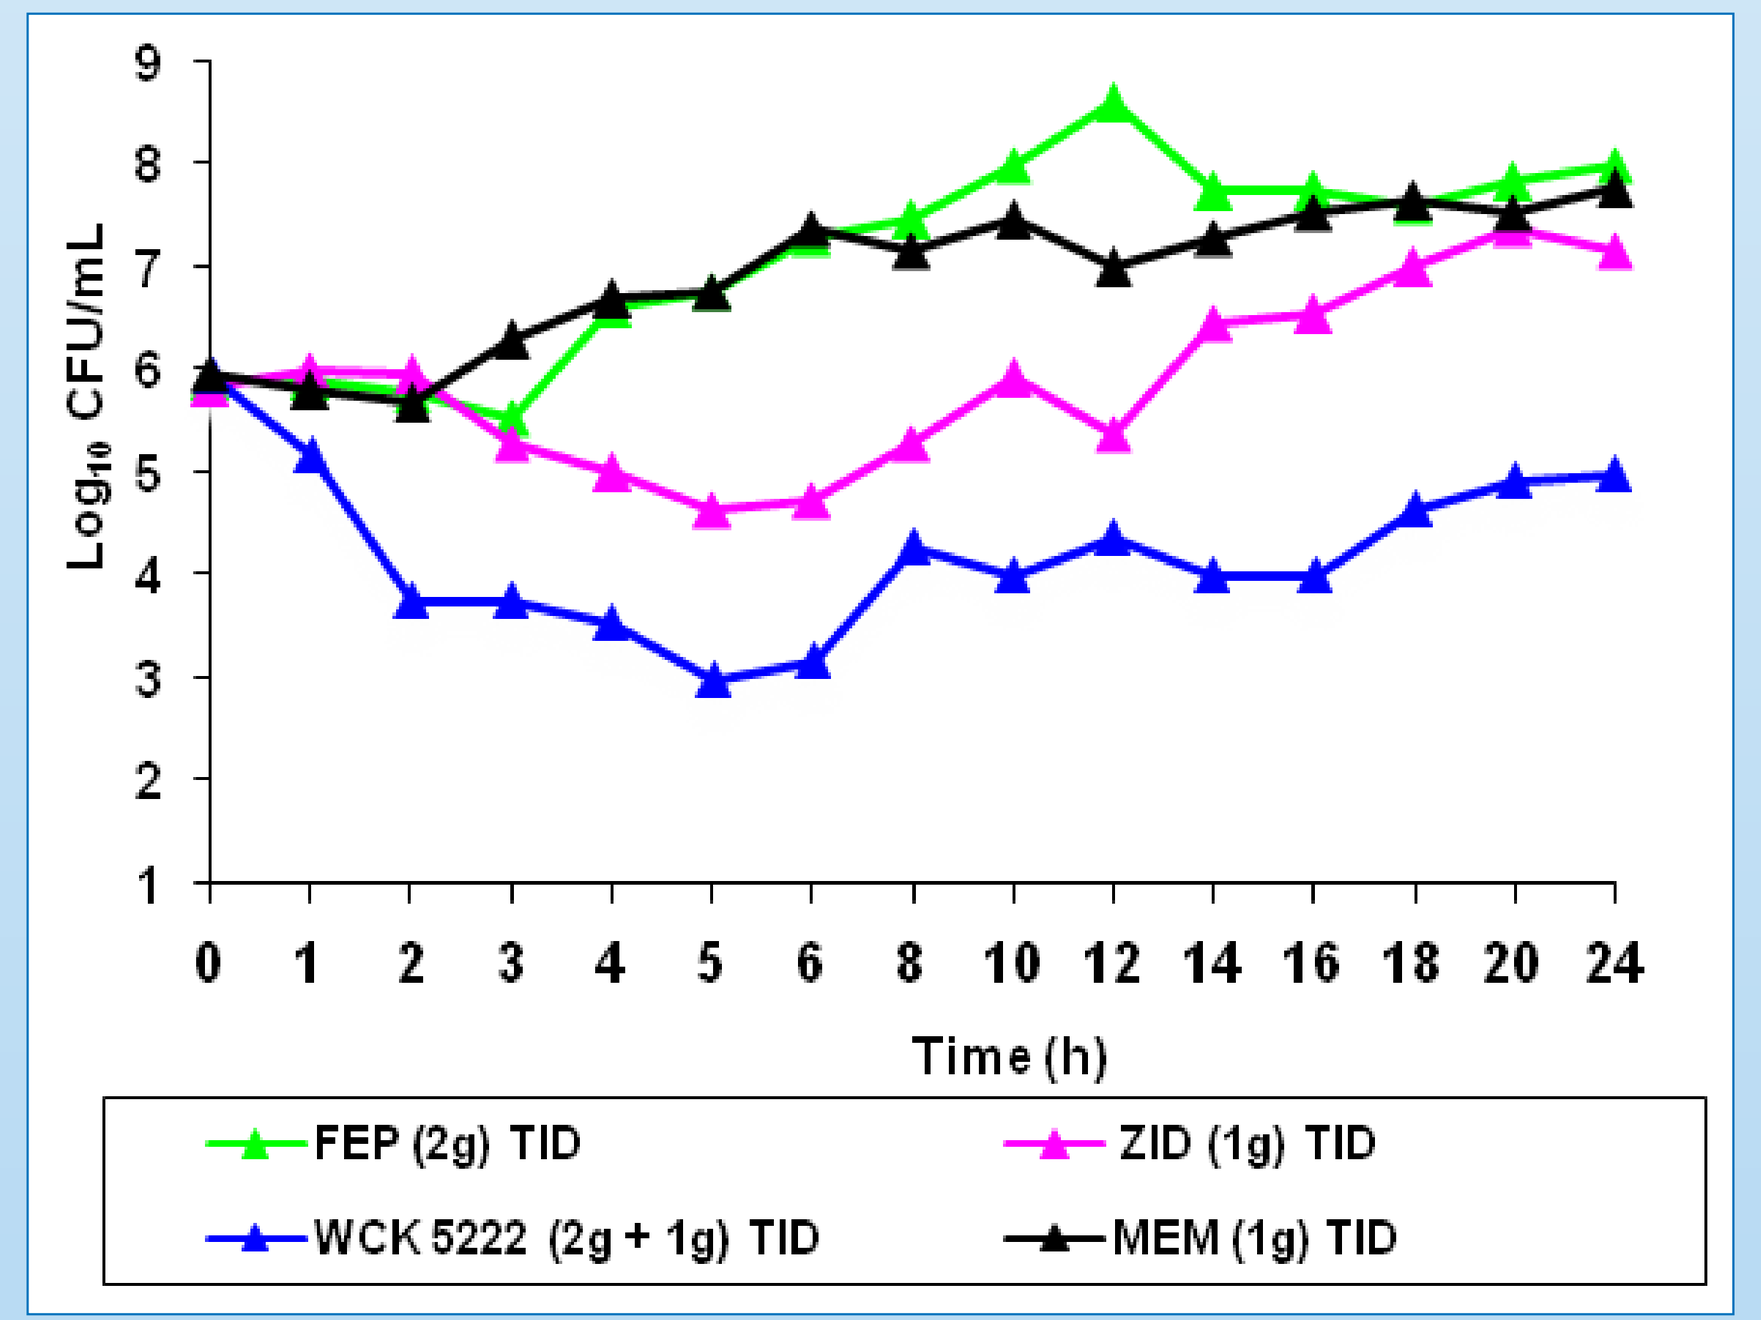

Log_{10} CFU/mL
Time (h)
FEP (2g) TID
ZID (1g) TID
WCK 5222 (2g + 1g) TID
MEM (1g) TID

- In IVPM study employing EC M44 (**Figure 2a)**, a rapid 3-log kill was observed within 3h and cidal action sustained till 24h at a dose of FEP-1g + ZID-0.5g.
- Similar to the observation with EC, even for KP NCTC 13443, WCK 5222 exerted a 3-log kill within 3h while standalone agents failed to cause bacteriostatic effect. The cidal action in terms of 1-log kill was sustained till 24h **(Figure 2b)**.
- In IVPM study employing PA NCTC 13437, standalone FEP and MEM at 2g and 1g, TID exposures, respectively, failed to inhibit the growth, whereas ZID at 1g, TID showed transient cidality of 1-log kill up to 8h whereas WCK 5222 at 2g+1g TID dose caused 2 - and 1- log kill upto 16h and 24h, respectively. Interestingly, with WCK 5222 a rapid 2 log kill of PA was attained in just 2h and increased to 3-log kill by 4-5h **(Figure 2c)**.

Murine lung infection dose fractionation study

- For PA S814, standalone FEP, ZID and IPM demonstrated regrowth ranging from 0.94-2.49 CFU/lung, whereas 100% mortality was observed in MEM-treated animals thus showing marked expression of NDM. FEP-ZID combination with a FEP dose of 50 mg/kg q2h (TDD: 600 mg/kg) along with ZID TDD of 25mg/kg (q4h and q2h regimen) resulted into regrowth of ~1-log, whereas in combination with ZID TDD of 50 mg/kg (q4h and q2h regimen) resulted into bacteriostatic effect. Further enhancing the ZID doses (TDD 100-550 mg/kg) caused progressive dose-dependent killing attaining a bactericidal effect to the tune of 3-log at its highest dose **(Figure 3)**. Exposure-response analysis undertaken to estimate the ZID threshold concentrations (C_T), showed that in the presence of FEP, ZID *f* T>C_T (4 µg/mL) was required to cause bacteriostasis was 18.4%, while to cause 1 and 2-log kill, it was 25.5% and 41%, respectively **(Figure 4)**. WCK 5222 Phase 1 clinical concentration time profile shows that at a ZID dose of 1g q8h, ZID C_T of 4 µg/mL is available for 100% of time for a 24h dosing duration, thus providing an adequate PK-PD cushion to absorb PK variability in clinical settings.
- It is important to note that, in recent WCK 5222 surveillance studies employing worldwide collection of 1291 PA isolates collected during 2015, WCK 5222 (1:1) MIC_{90} was 4 µg/mL and MIC_{99} was 8 µg/mL. Thus just 5 strains showed MIC of 16 µg/mL and only 1strain demonstrated MIC of 32 µg/mL[6]. Demonstration of therapeutically relevant efficacy against NDM-expressing PA S814 with FEP-ZID MIC of 32 µg/mLsuggest a potential for long term therapeutic effectiveness of WCK 5222 against a pathogen known for relatively rapid susceptibility shifts to all the classes of antibacterials.

Figure 3: Performance of ZID administered q2h to q4h in combination with FEP 50 mg/kg q2h (TDD: 600 mg/kg) against PA S814

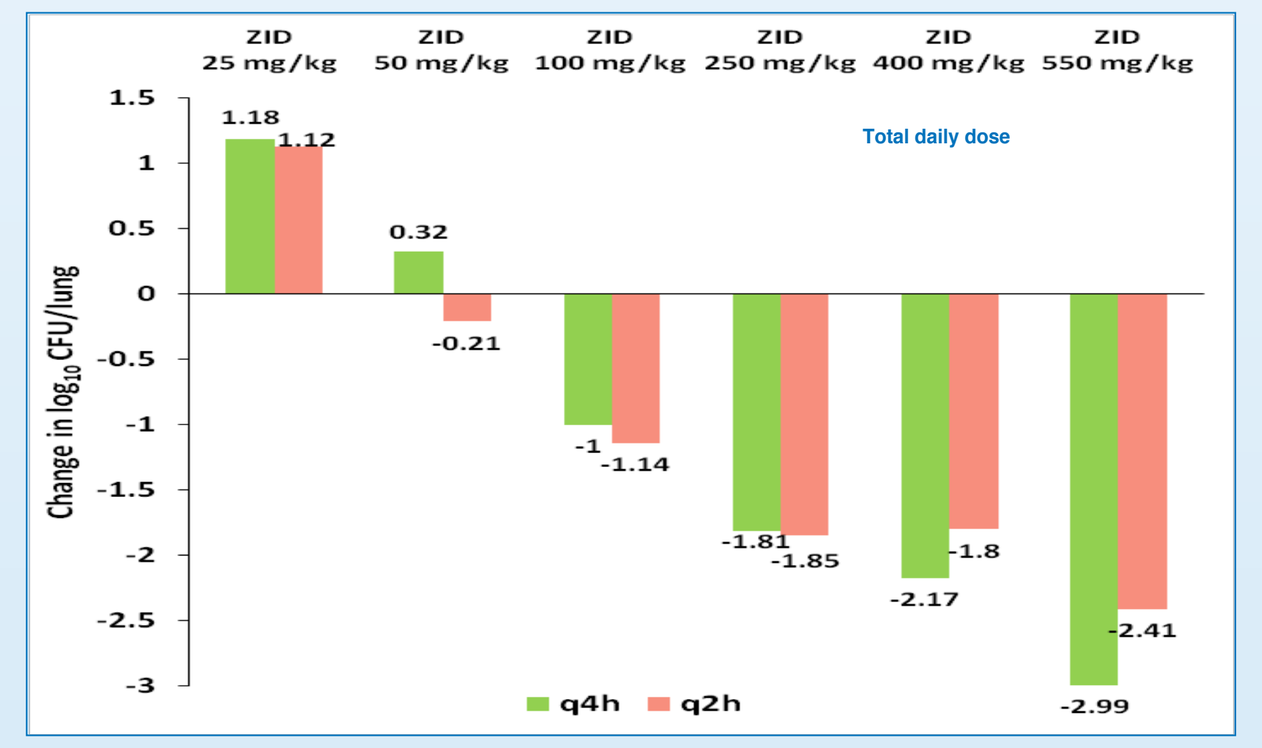

ZID 25 mg/kg
ZID 50 mg/kg
ZID 100 mg/kg
ZID 250 mg/kg
ZID 400 mg/kg
ZID 550 mg/kg
Total daily dose
Change in log_{10} CFU/lung
1.18
1.12
0.32
-0.21
-1
-1.14
-1.81
-1.85
-2.17
-1.8
-2.99
-2.41
q4h
q2h

Figure 4: Exposure–response analysis leading to identification of ZID threshold concentration (FEP-50 mg/kg q2h (TDD-600 mg/kg)

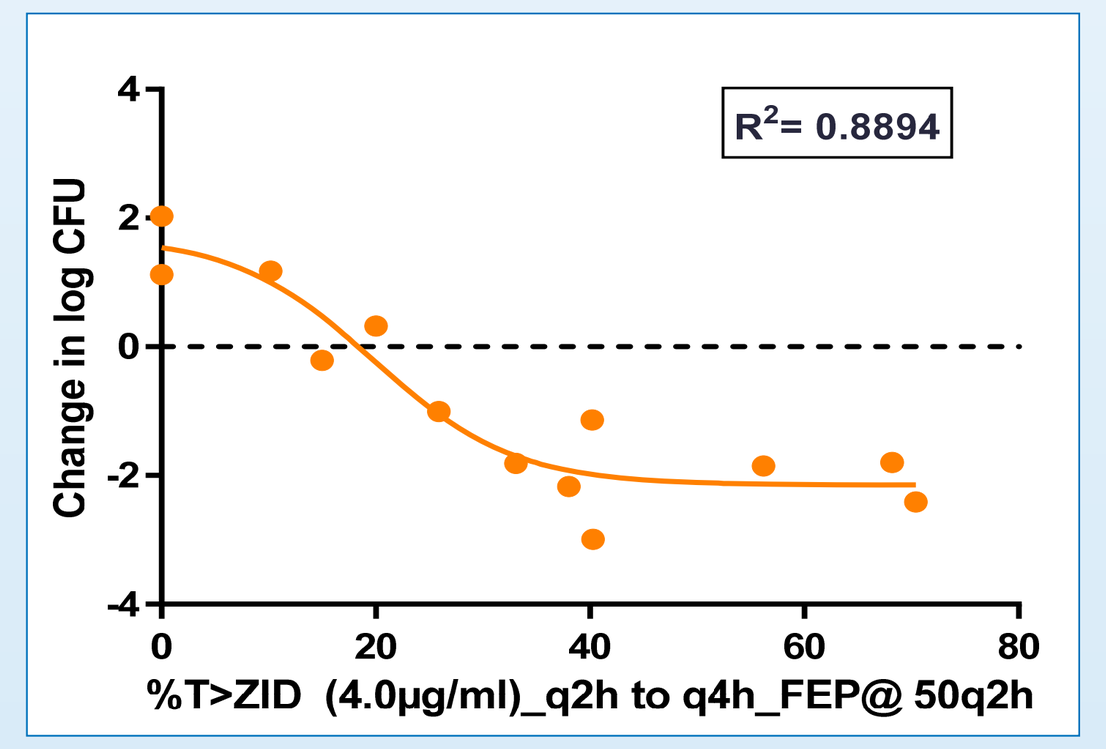

R^2= 0.8894
Change in log CFU
%T>ZID (4.0µg/ml)_q2h to q4h_FEP@ 50q2h

Determination of FEP T>MIC for PA S814

- Standalone FEP (600 mg/kg, fractionated as q2h), ZID (100 mg/kg, fractionated as q2h) and IPM (240 mg/kg, fractionated as q2h) showed bacterial regrowth thus failing to even bring about bacteriostasis. However, FEP-ZID combination at the lowest FEP TDD dose of 300 mg/kg even when given q6h in combination with ZID TDD of 100 mg/kg (q2h) demonstrated bacteriostatic effect. More fractionated regimens, q4h and q2h provided successively improved killing showing that WCK 5222 cidal action is time-dependent. Interestingly, even less fractionated q6h regimens for all the higher FEP doses provided ≥2-log kill demonstrating manifestation of enhancer effect culminating in lowered T>MIC for FEP **(Figure 5)**.
- Exposure-response analysis undertaken for identifying the %*f*T>MIC of FEP at q4h and q6h in the presence of ZID (TDD: 100 mg/kg) showed that in combination with ZID,*f*T>MIC of FEP causing1 and 2-log kill is as low as 13.55% and 21.7%, respectively as against the generally accepted requirement of 50-60% *f*T>MIC linked with 1-log kill (**Figure 6, Table 4**).

Figure 5: Performance of FEP (TDD: 300,450, 600 and 900 mg/kg) administered q2h to q6h in combination with ZID 8.33 mg/kg q2h (TDD:100 mg/kg) against PA S814

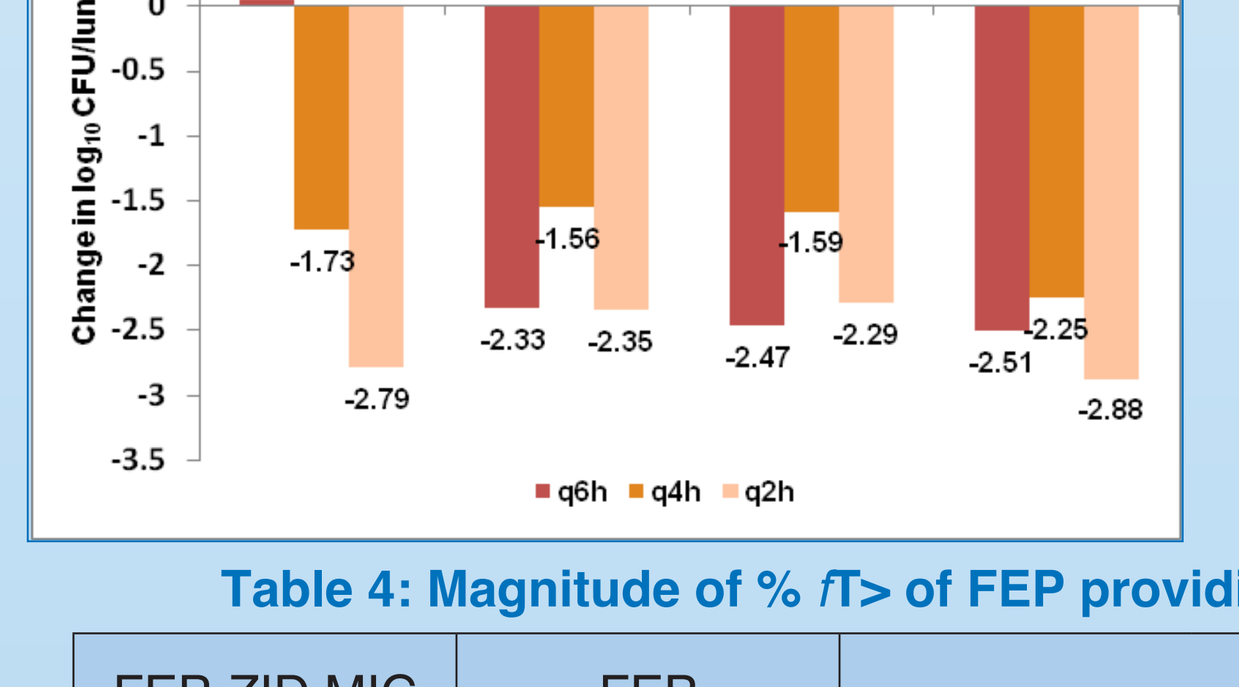

FEP 300 mg/kg
FEP 450 mg/kg
FEP 600 mg/kg
FEP 900 mg/kg
Change in log_{10} CFU/lung
0.05
-1.73
-2.79
-2.33
-1.56
-2.35
-2.47
-1.59
-2.29
-2.51
-2.25
-2.88
q6h
q4h
q2h

Figure 6: Exposure–response analysis leading to identification of %*f* T>MIC of FEP at q4h and q6h doses

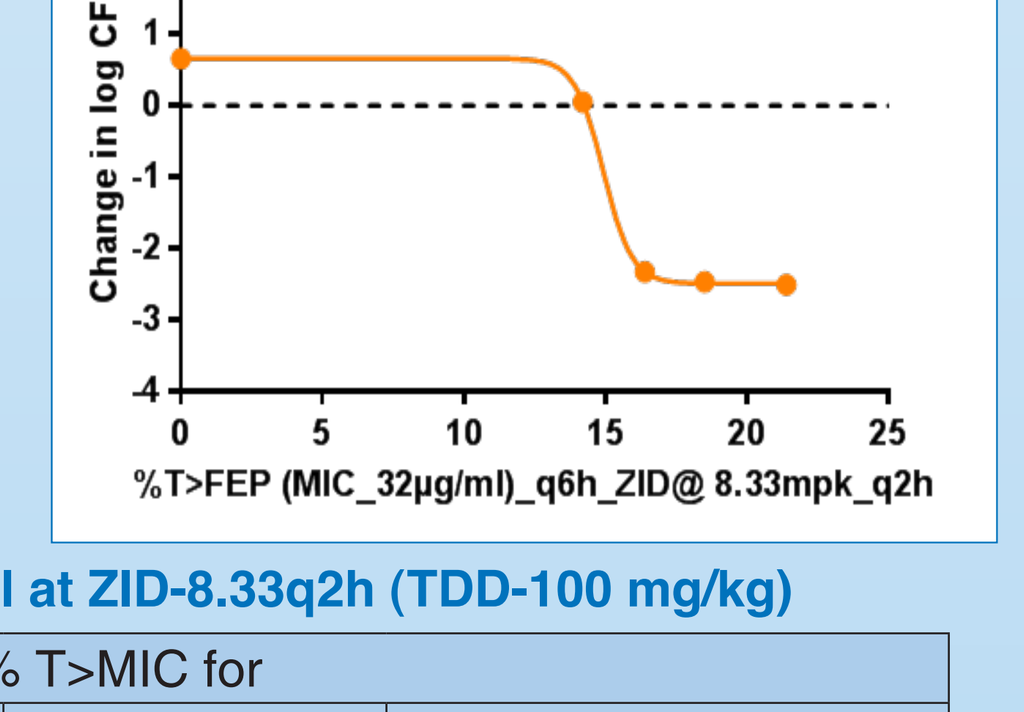

R^2= 0.9999
Change in log CFU
%T>FEP (MIC_32µg/ml)_q6h_ZID@ 8.33mpk_q2h

Table 4: Magnitude of % *f*T> of FEP providing 1 and 2-log kill at ZID-8.33q2h (TDD-100 mg/kg)

FEP-ZID MIC	FEP dosing regimen	FEP % T>MIC for 1.0-log kill	Mean for 1.0-log kill	2.0-log kill	Mean for 2.0-log kill
32 µg/mL	q4h	12.1	13.55	27.6	21.7
	q6h	15.0		15.8	

CONCLUSION

- In the presence of ZID, *f*T>MIC requirement for FEP to cause 1 and 2-log kill is dramatically lower thereby enhancing WCK 5222 therapeutic scope for the coverage of high MIC PA strains.
- ZID % *f*T>C_T linked with 1 and 2-log kill are readily attainable at ZID 1g TID clinical doses and would provide sufficient cushion for absorbing PK variability.
- Studies described here demonstrate that ZID transforms FEP PD into a 'carbapenem like' PD owing to WCK 5222's rapid cidal action emanating from multiple PBP binding.
- ZID driven 'enhancer' mechanism of action helps WCK 5222 overcome MBL-mediated resistance mechanism through complementary PBP action and thereby circumventing the need of β-lactamase inhibition.
- WCK 5222 represents a novel therapeutically pertinent mechanistic approach for tackling MDR pathogens including MBL-expressing strains.

References

1. SR Palwe *et al.,* Sunday 438. ASM Microbe 2016.
2. SR Palwe *et al.,* Sunday 448. ASM Microbe 2016.
3. SS Takalkar et al., Sunday 444, ASM Microbe 2016.
4. M Castanheira *et al.,* P1615, ECCMID 2016.
5. M Castanheira *et al.,* P1339, ECCMID 2016.
6. HS Sader *et al.,* Friday-478, ASM Microbe 2016.
7. B Moya *et al.,* Antimicrob Agents Chemother. 2010; 54:3933-7.
8. G Zhao *et al.,* Antimicrob. Agents Chemother. 43:1124-1128.
9. MF Mojica *et al.,* Biochemistry. 2015; 54:3183-96.
10. CR. Bethel *et al.,* oral presentation ASM Microbe 2016.

Disclosures

This study was funded by Wockhardt, Bio AG.

A. Udaykar, SS. Bhagwat and M. Patel are employees of Wockhardt Research Centre, Aurangabad, India.

Poster presented at ID Week, 26th – 30th October, 2016, New Orleans, LA, USA.