1862

Meropenem-Vaborbactam (VABOMERE) vs. Best Available Therapy for CRE Infections: TANGO II Randomized, Controlled Phase 3 Study Results

The Medicines Company
3013 Science Park Rd.
San Diego, CA 92121 USA
8 Sylvan Way
Parsippany, NJ 07054 USA

Keith S. Kaye[1], Jose Vazquez[2], Amy Mathers[3], George Daikos[4], Elizabeth Alexander[5], Jeff Loutit[6], Shu Zhang[5], Michael Dudley[6], Oliver A. Cornely[7]

[1]University of Michigan Medical School, Ann Arbor, MI, USA; [2] Medical College of Georgia/Augusta University, Augusta, GA, USA; [3]University of Virginia Health System, Charlottesville, VA, USA; [4]National & Kapodistrian University of Athens, Athens, Greece; [5]The Medicines Company, Parsippany, NJ, USA; [6]The Medicines Company, San Diego, CA, USA; [7]University Hospital of Cologne, Cologne, Germany

Abstract

Background

Meropenem-vaborbactam (M-V) is a new beta-lactam-beta-lactamase inhibitor combination that restores the potency of meropenem in *Klebsiella pneumoniae* carbapenemase (KPC)-producing carbapenem-resistant Enterobacteriaceae (CRE). TANGO II is a randomized, open-label trial in patients with complicated urinary tract infection (cUTI), acute pyelonephritis (AP), hospital-acquired or ventilator-associated bacterial pneumonia (HABP/VABP), bacteremia, or complicated intra-abdominal infection (cIAI), due to known or suspected CRE. Clinical outcomes across indications in patients with documented CRE at baseline are presented.

Methods

Subjects were randomized 2:1 to monotherapy with M-V (2g/2g every 8h as a 3h infusion) or Best Available Therapy (BAT) for 7-14 days. BAT included the following alone or in combination: carbapenems, aminoglycosides, polymyxin B, colistin, tigecycline or ceftazidime-avibactam (monotherapy only). Clinical cure was defined as a complete resolution of signs or symptoms such that no further antimicrobial therapy was required.

Results

72 patients were enrolled; 50 (69.4%) had a gram-negative baseline organism (m-MITT), and 43 (59.7%) had a baseline CRE (mCRE-MITT). Within mCRE-MITT, 20 patients had bacteremia, 15 had cUTI/AP, 5 had HABP/VABP, and 3 had cIAI. The most common baseline CRE pathogens were *K. pneumoniae* (86%) and *Escherichia coli* (7%). There was no consensus BAT regimen; however, combination therapy was used in 66.7% of cases. Treatment duration was similar across arms (mean 8.5 days for M-V and 8.1 days for BAT). Cure rates at end of treatment (EOT) and test of cure (TOC; 7 days after EOT) in mCRE-MITT and m-MITT populations are shown:

mCRE-MITT				
Time Point	M-V (n=28) N, %	BAT (n=15) N, %	% Difference (M-V – BAT)	95% CI
EOT	18 (64.3%)	6 (40.0%)	31.0%	(1.2% t 60.7%)
TOC	16 (57.1%)	4 (26.7%)	30.5%	(1.5% to 59.4%)
m-MITT				
Time Point	M-V (n=31) N, %	BAT (n=19) N, %	% Difference (M-V – BAT)	95% CI
EOT	21 (67.7%)	8 (42.1%)	30.9%	(3.6% to 58.1%)
TOC	18 (58.0%)	6 (31.6%)	26.5%	(-0.7% to 53.7%)

Conclusion

M-V was associated with a higher rate of clinical cure at EOT and TOC than BAT across all indications in both m-MITT and mCRE-MITT populations. M-V is a promising new option for the treatment of CRE infections.

Background

- The growing prevalence of drug-resistant gram-negative pathogens has resulted in a critical need for new antimicrobial agents.[1,2]
 - In the US, the production of *Klebsiella pneumoniae* carbapenemase (KPC) enzyme is the major mechanism of carbapenemase-mediated resistance in carbapenem-resistant Enterobacteriaceae (CRE).[3]
 - Meropenem-vaborbactam (M-V; VABOMERE) is a new beta-lactam-beta-lactamase inhibitor combination developed for treatment of serious gram-negative infections, including CRE.
- TANGO II is a Phase 3, multi-center, randomized, open-label comparative trial designed to evaluate efficacy and safety of M-V vs. best available therapy (BAT) in patients with confirmed CRE infections.
 - Given the challenges of conducting clinical trials in those patients with drug-resistant infections, few new agents targeting CRE have been systematically studied in patients with these infections.[4,5]
 - Here we report clinical outcomes including clinical cure across infection types and Day 28 all-cause mortality, in patients with documented baseline CRE infection in an interim analysis.
 - The study was discontinued on July 21, 2017, on the recommendation of the Data Safety Monitoring Board following their review of these data.

Methods

- Phase 3, multi-center, randomized, open-label study of adults with infections due to known or suspected CRE, including complicated urinary tract infection (cUTI), acute pyelonephritis (AP), hospital-acquired/ventilator-associated bacterial pneumonia (HABP/VABP), bacteremia, or complicated intra-abdominal infection (cIAI).
- Eligible subjects were randomized 2:1 to monotherapy with M-V (2g/2g every 8h via 3-h infusion) or BAT for 7-14 days (**Figure 1**).
 - BAT included (alone or in combination): a carbapenem, aminoglycoside, polymyxin B, colistin, tigecycline, or ceftazidime-avibactam (monotherapy only) at doses determined by the primary investigator (PI).
- Enrollment was stratified by infection type and geographic region.
- Key inclusion criteria: known or suspected (evidence of CRE in culture or molecular testing within past 90 d) CRE pathogen, requirement of ≥7 days IV therapy, confirmed cUTI/AP, HABP/VABP, bacteremia, or cIAI.
- Key exclusion criteria: receipt of >24 hours of potentially effective antimicrobials (except in cases of clinical failure), immediate life-threatening disease, known infection due to NDM, VIM, IMI or OXA-encoded beta-lactamase.
- Clinical cure was defined as a complete resolution of signs/symptoms such that no further antimicrobial therapy was required.
- Clinical cure was assessed by the onsite blinded investigator (BI) and PI at two time points: end of treatment (EOT) and test of cure (TOC). In cases where the assessment by the BI and PI differed, clinical cure was adjudicated by the blinded independent adjudication committee.
- Safety endpoints were assessed in those who received ≥1 dose of study drug.
- The study was not powered for formal inferential testing. Results of ad hoc statistical analyses are presented where performed.

Figure 1. Study Schema

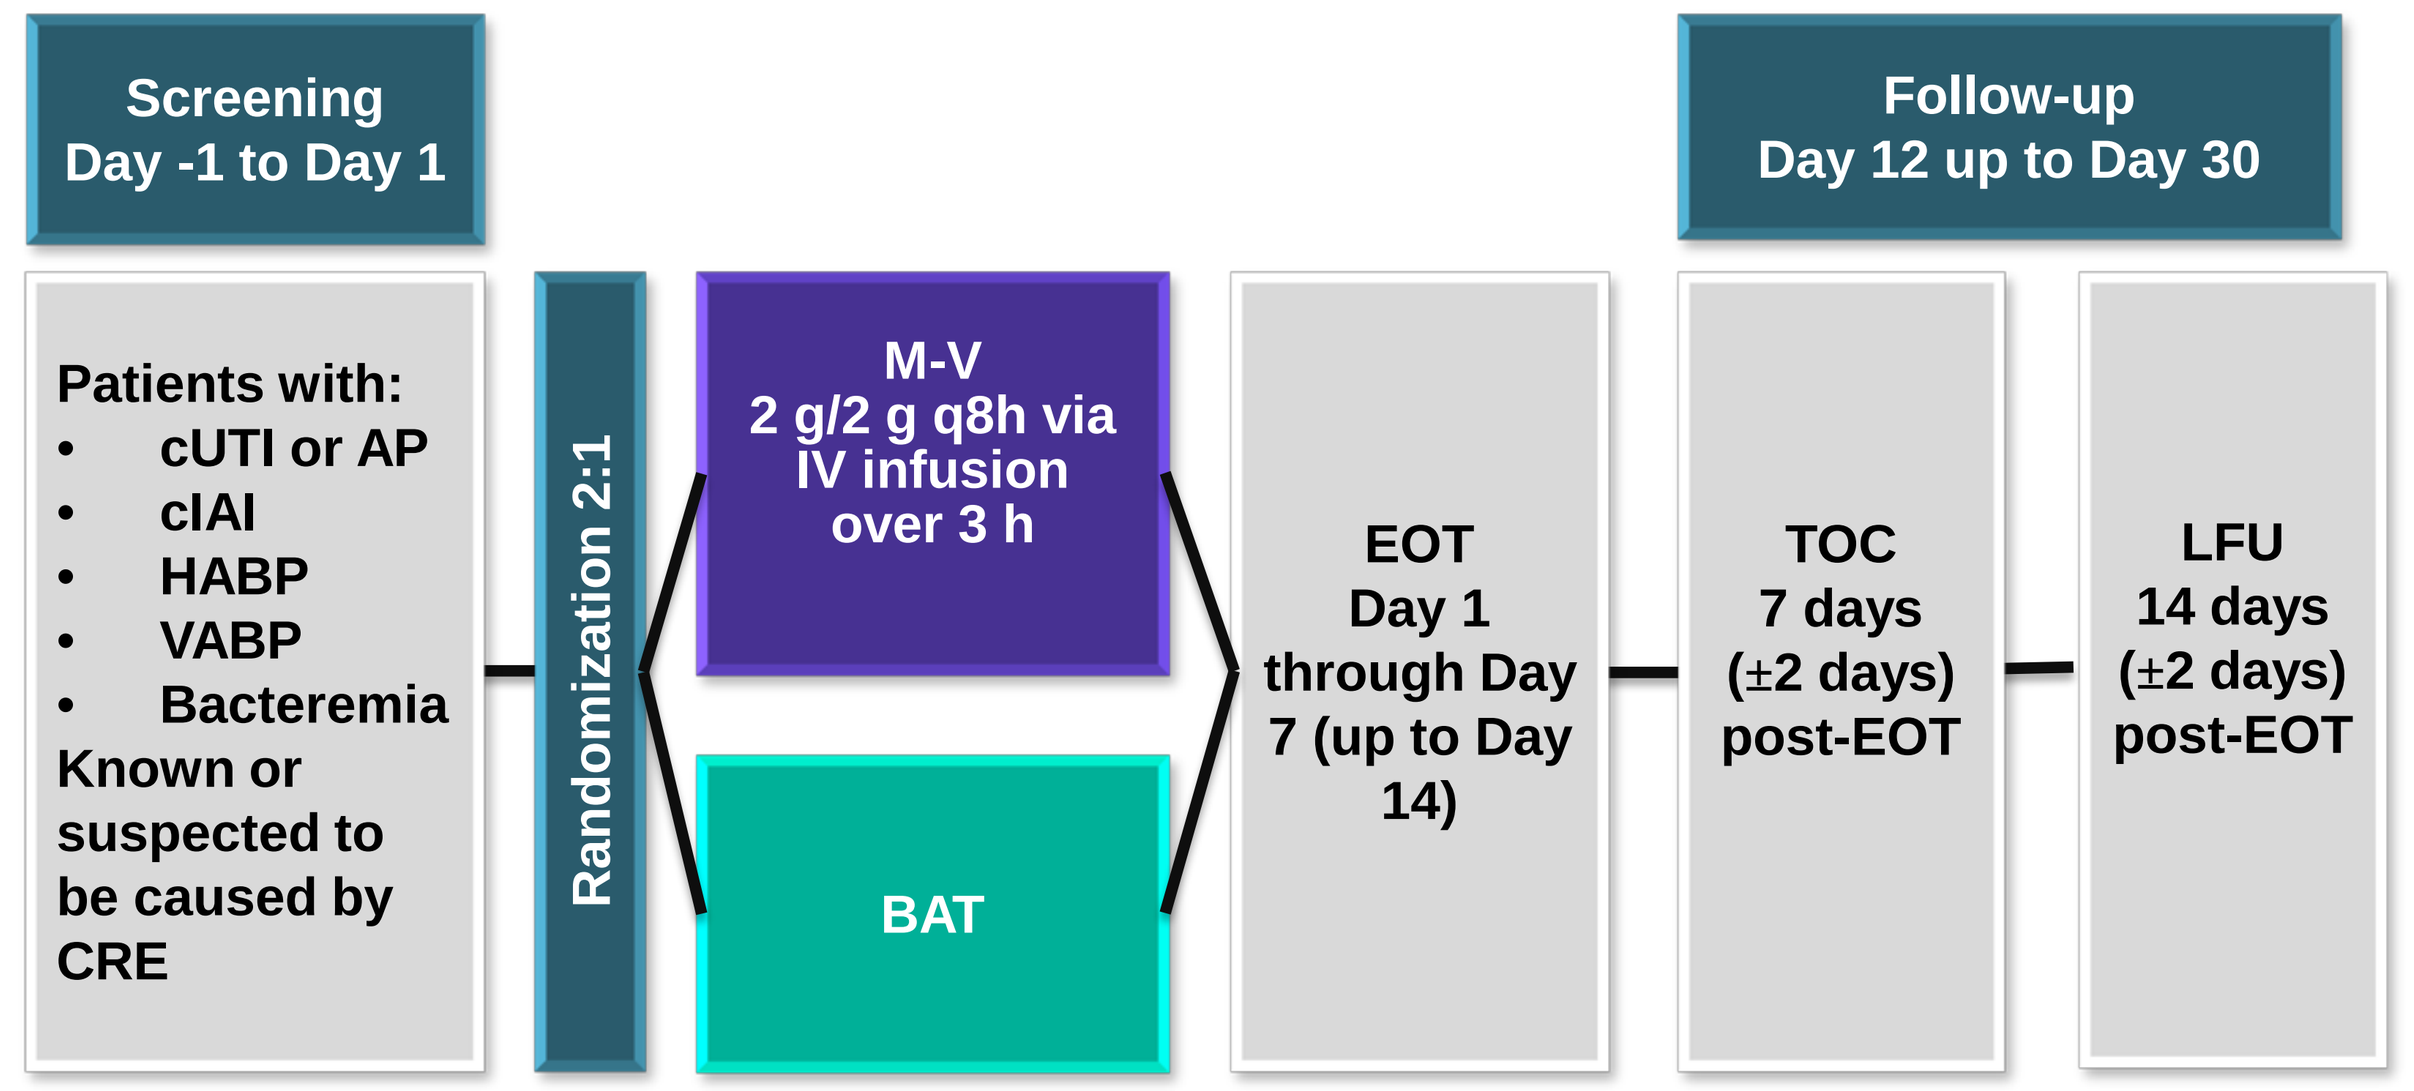

Screening
Day -1 to Day 1
Follow-up
Day 12 up to Day 30
Patients with:
• cUTI or AP
• cIAI
• HABP
• VABP
• Bacteremia
Known or suspected to be caused by CRE
Randomization 2:1
M-V
2 g/2 g q8h via IV infusion over 3 h
BAT
EOT
Day 1 through Day 7 (up to Day 14)
TOC
7 days (±2 days) post-EOT
LFU
14 days (±2 days) post-EOT

Results

Of the 72 enrolled patients, 50 (69.4%) had a gram-negative baseline organism (m-MITT), and 43 (59.7%) had a baseline CRE (mCRE-MITT). Baseline characteristics for the mCRE-MITT population are shown in **Poster#1867, Wunderink R et al, IDWeek 2017** and consistent with a highly comorbid patient population. Distribution of infection types shown below.

Distribution of Infection Types, mCRE-MITT (N=43)

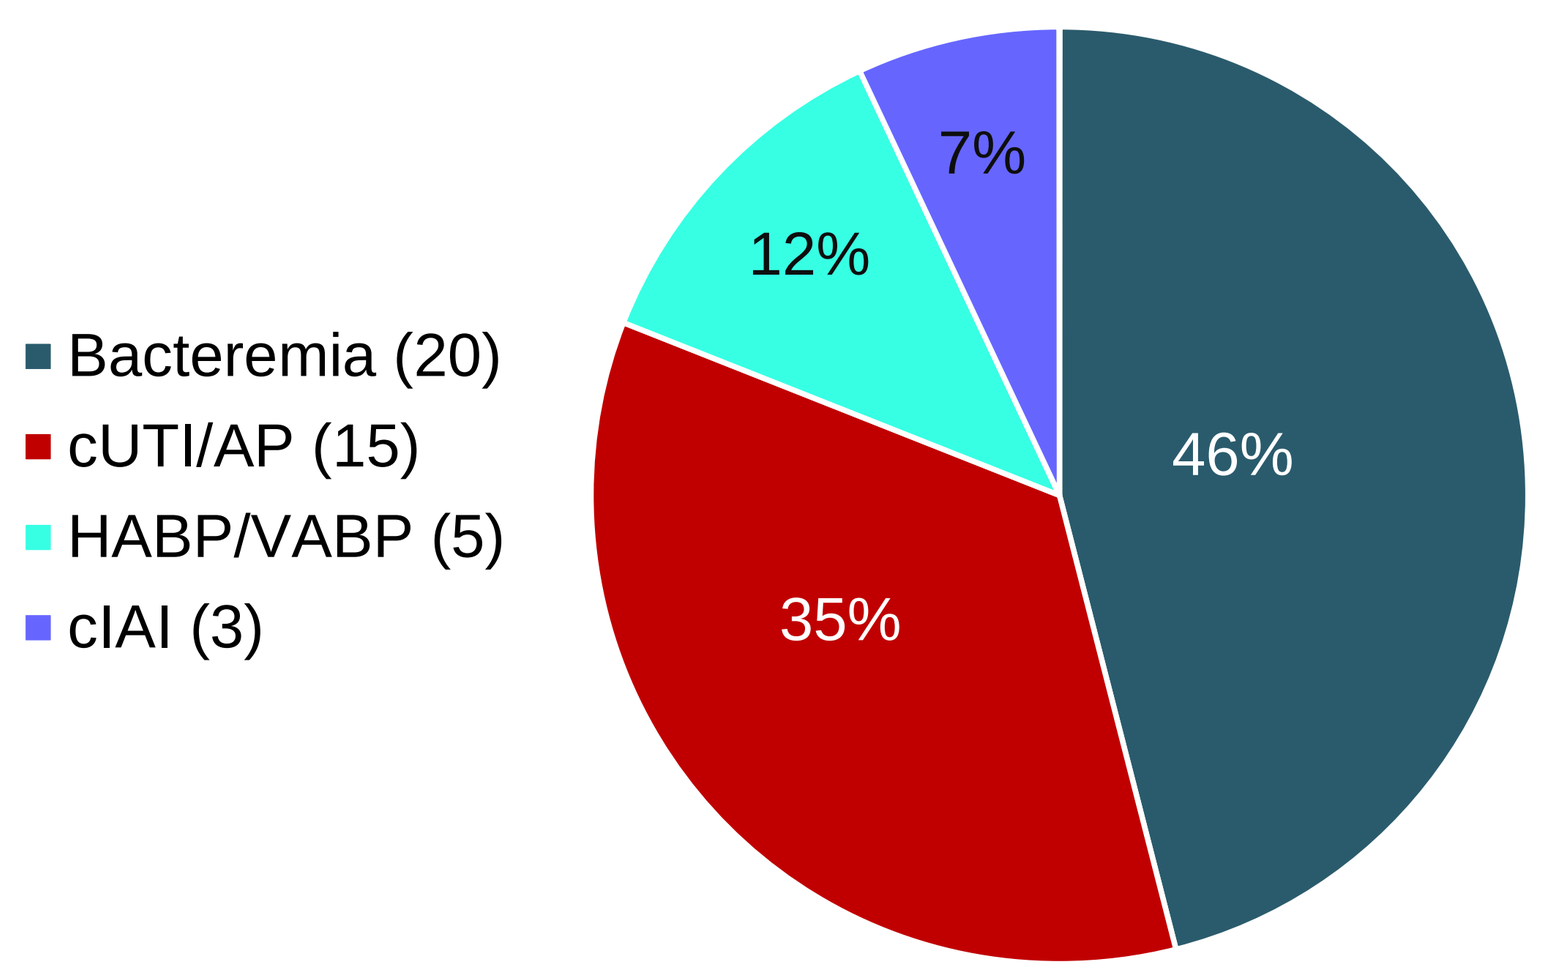

Bacteremia (20)
cUTI/AP (15)
HABP/VABP (5)
cIAI (3)
46%
35%
12%
7%

- Mean treatment duration was similar for M-V (8.5 days) and BAT (8.1 days).
- The most common baseline pathogen was *K. pneumoniae* (86%).
- Meropenem MIC50 was >32 µg/mL.
- The most common molecular mechanism of carbapenem resistance was production of KPC carbapenemase (80%).
- Meropenem MIC50 in KPC-producing *K pneumonia* was 64 µg/mL.

67% (10/15) of those who received a BAT regimen received combination therapy (mCRE-MITT), usually including a carbapenem agent (**Table 1**).

Table 1. Antibiotic Regimens in Best Available Therapy Group (mCRE-MITT)

BAT Regimen	N=15* n, (%)
Monotherapy	4 (26.7)
Aminoglycoside	1 (6.7)
Carbapenem	1 (6.7)
Ceftazidime-Avibactam	1 (6.7)
Polymyxin/Colistin	1 (6.7)
Dual Therapy	7 (46.7)
Carbapenem + Aminoglycoside	1 (6.7)
Carbapenem + Polymyxin/Colistin	1 (6.7)
Carbapenem + Tigecycline	2 (13.3)
Polymyxin/Colistin + Aminoglycoside	3 (20.0)
Triple Therapy	1 (6.7)
Carbapenem + Polymyxin/Colistin + Tigecycline	1 (6.7)
≥4 Drugs	2 (13.3)
Carbapenem + Polymyxin/Colistin + Tigecycline + Aminoglycoside	2 (13.3)

mCRE-MITT, microbiological carbapenem-resistant Enterobacteriaceae modified intent to treat.
*One subject received BAT with ciprofloxacin (not permitted in the study) and is therefore not represented.

Table 2. Efficacy Endpoints Across All Infection Types (mCRE-MITT)

Efficacy Endpoints	M-V N=28 n (%)	BAT N=15 n (%)	Absolute Difference[a] (95% CI)	*P* value	Relative Difference[b]
Patients with All Infection Types					
Clinical Cure at EOT	18 (64.3)	5 (33.3)	31.0 (1.2 to 60.7)	.04	93.1
Clinical Cure at TOC	16 (57.1)	4 (26.7)	30.5 (1.5 to 59.4)	.04	114.2
Microbiologic Cure[c] at EOT	18 (64.3)	6 (40.0)	24.3	NA	60.8
Microbiologic Cure[c] at TOC	14 (50.0)	5 (33.3)	16.7	NA	50.1
Day-28 Mortality	5 (17.9)	5 (33.3)	−15.5 (-43.2 to 12.3)	.27	−46.5
Exploratory Analysis of Risk-Benefit Profile of M-V Compared to BAT					
	M-V N=28 n (%)	BAT N=15 n (%)	Absolute Difference[a] (95% CI)	*P* value	Relative Difference[b]
Day-28 All-Cause Mortality or Nephrotoxicity[d]	7 (25.0)	6 (40.0)	-15.0 (-44.5 to 14.5)	.32	-37.5
Clinical Failure or Nephrotoxicity[e]	9 (32.1)	12 (80.0)	-47.9 (-74.5 to -21.2)	<.001	-59.9
Day-28 All-Cause Mortality or Renal AEs[f]	6 (21.4)	9 (60.0)	-38.6 (-67.7 to -9.5)	<.01	-64.3
Clinical Failure or Renal AEs[g]	9 (32.1)	12 (80.0)	-47.9 (-74.5 to -21.2)	<.001	-59.9

Table 2 (continued). Efficacy Endpoints Across All Infection Types (mCRE-MITT)

Sensitivity Analysis of Clinical Cure at TOC and All-Cause Mortality at Day 28 Across All Infection Types (mCRE-MITT) Excluding Prior Antibiotic Failure[h]	M-V N=19 n (%)	BAT N=15 n (%)	Absolute Difference[a] (95% CI)	*P* value	Relative Difference[b]
Patients with All Infection Types					
Clinical Cure at TOC	13 (68.4)	4 (26.7)	41.8 (11.1 to 72.4)	.008	156.2
Day-28 All-cause Mortality	1 (5.3)	5 (33.3)	-28.1 (-54.0 to -2.2)	.03	-84.1

[a] Data represent the difference in percentages for M-V and BAT (95% CI for that difference).
[b] Data represent (difference in percentage M-V and BAT)/BAT.
[c] Composite of either microbiologic eradication or presumed eradication at respective visit.
[d] Composite outcome of either Day-28 all-cause mortality or a post-baseline increase in serum creatinine ≥1.0 mg/dL.
[e] Composite outcome of either clinical failure at test of cure or a post-baseline increase in serum creatinine ≥1.0 mg/dL.
[f] Composite outcome of either Day-28 all-cause mortality or adverse event of renal failure, renal failure acute or renal impairment.
[g] Composite outcome of either clinical failure at test of cure or adverse event of renal failure, renal failure acute or renal impairment
[h] Patients assessed as having prior antibiotic failure at randomization (meropenem-vaborbactam, 9; BAT, 0) excluded from analysis.

Table 3. Adverse Events and Safety Endpoints (Safety Population)

Adverse Events	M-V N=45 n (%)	BAT N=25 n (%)	Total N=70 n (%)
TEAEs			
Any	38 (84)	23 (92)	61 (87)
Drug-related	11 (24)	11 (44)	22 (31)
TEAEs by maximum severity			
Mild	9 (20)	4 (16)	13 (19)
Moderate	11 (24)	5 (20)	16 (23)
Severe	6 (13)	7 (28)	13 (19)
Life-threatening	2 (4)	1 (4)	3 (4)
Death	10 (22)	6 (24)	16 (23)
SAEs			
All	15 (33)	11 (44)	26 (37)
Drug-related	0 (0)	2 (8)	2 (3)
Study drug discontinuations due to TEAEs	5 (11)	3 (12)	8 (11)
Study discontinuations due to TEAEs	8 (18)	5 (20)	13 (19)

SAE, serious adverse event; TEAE, treatment-emergent adverse event; TRAE, treatment-related adverse event.

Figure 2. Post-baseline Creatinine Increases

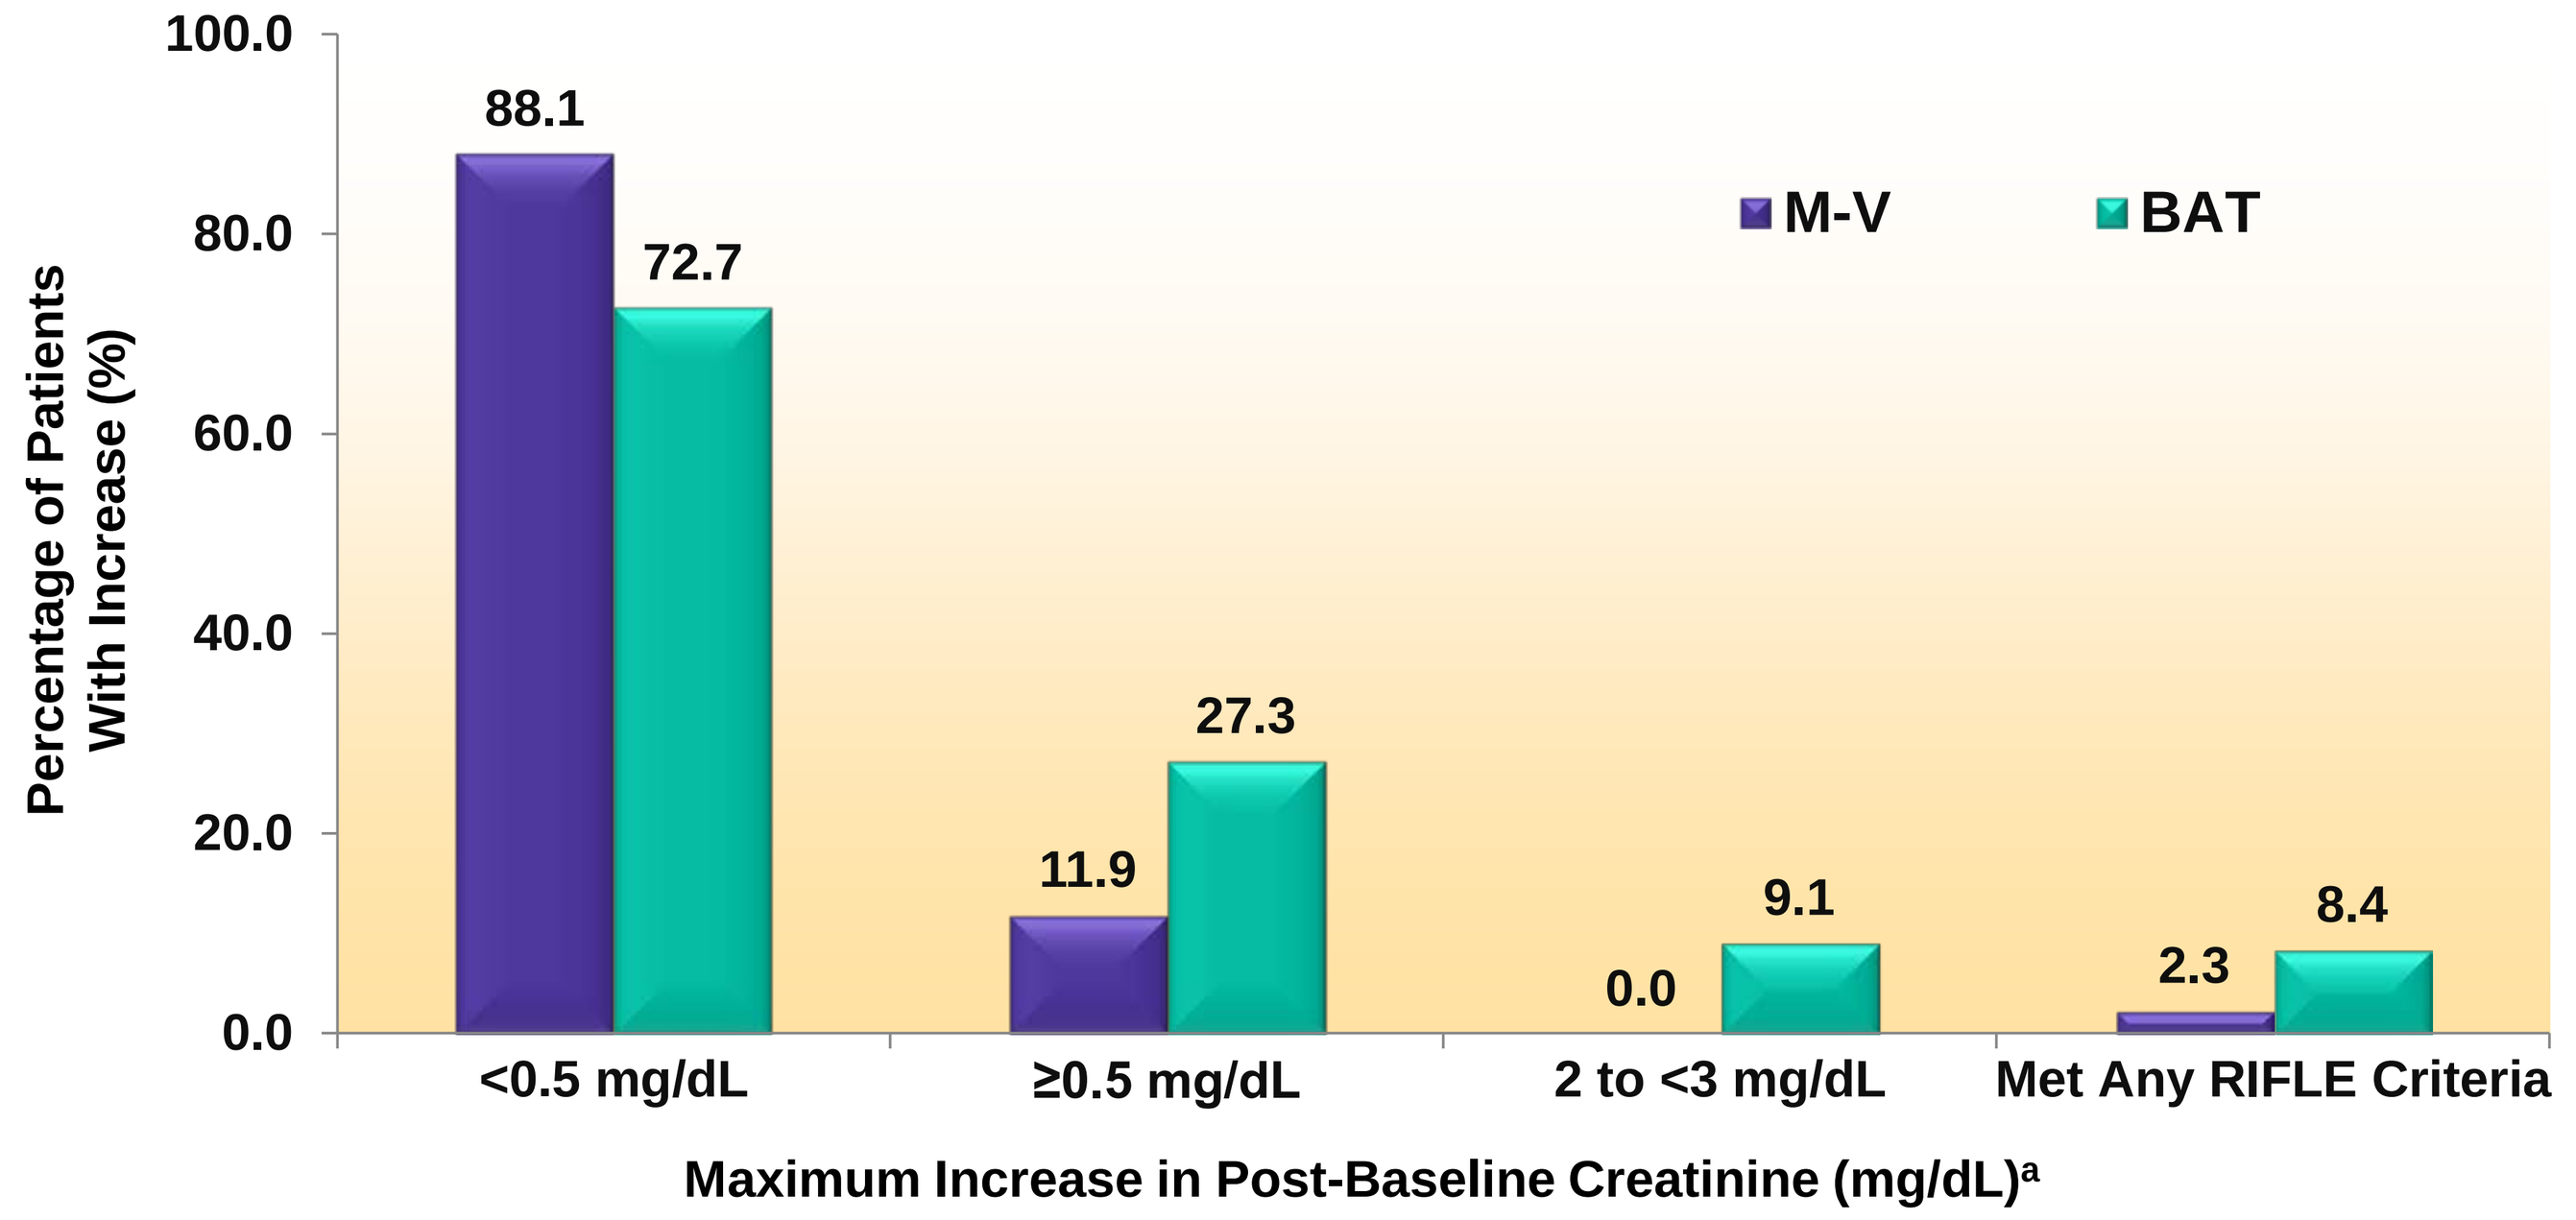

Percentage of Patients With Increase (%)
100.0
80.0
60.0
40.0
20.0
0.0
M-V
BAT
88.1
72.7
11.9
27.3
0.0
9.1
2.3
8.4
<0.5 mg/dL
≥0.5 mg/dL
2 to <3 mg/dL
Met Any RIFLE Criteria
Maximum Increase in Post-Baseline Creatinine (mg/dL)a

RIFLE, Risk, Injury, Failure, Loss, or End-stage.
[a] Values at baseline or post baseline were missing for 3 M-V patients and 3 BAT patients.

Conclusions

For the mCRE-MITT population:

- M-V treatment resulted in higher cure rates at both EOT and TOC (*P*=.04 at both time points).
- Day-28 all-cause mortality (ACM) was 17.9% for M-V and 33.3% for BAT; when prior antibiotic failures are excluded, Day-28 ACM was 5.3% for M-V and 33.3% for BAT (*P*=.03).
- M-V was associated with fewer treatment-related AEs (M-V 24.4% vs. BAT 44.0%).
- M-V was associated with decreased nephrotoxicity as evidenced by serum creatinine increase ≥0.5 mg/dL (M-V 11.1% vs. BAT 24.0%).
- An exploratory analyses using the composite endpoints of clinical failure or nephrotoxicity demonstrated a risk:benefit profile favoring M-V (M-V 32.1% vs BAT 80.0%; 95%CI, −74.5% to −21.2%; *P*<.001).
- In summary, M-V monotherapy was associated with decreased mortality, increased clinical cure, and reduced nephrotoxicity compared with BAT in the treatment of CRE infections.
- Based on the totality of these results, the independent Data Safety Monitoring Board recommended discontinuing randomization to BAT.

Disclosures

This project has been funded in whole or in part with federal funds from the Department of Health and Human Services; Office of the Assistant Secretary for Preparedness and Response; Biomedical Advanced Research and Development Authority (BARDA), under Contract No. HHSO100201400002C with Rempex Pharmaceuticals, a wholly-owned subsidiary of The Medicines Company and Agreement No. HHSO100201600026C with The Medicines Company.

References

1. Boucher HW, Talbot GH, Bradley JS, Edwards JE, Gilbert D, Rice LB, et al. Bad bugs, no drugs: no ESKAPE! An update from the Infectious Diseases Society of America. Clin Infect Dis. 2009 Jan;48(1):1-12.
2. Centers for Disease Control and Prevention. Antibiotic resistance threats in the United States, 2013. Atlanta, GA (US): US Department of Health and Human Services; 2013. 114 p.
3. Castanheira M, Sader HS, Deshpande LM, Fritsche TR, Jones RN. Antimicrobial activities of tigecycline and other broad-spectrum antimicrobials tested against serine carbapenemase- and metallo-β-lactamase-producing Enterobacteriaceae: report from the SENTRY Antimicrobial Surveillance Program. Antimicrob Agents Chemother. 2008 Feb;52(2):570-3.
4. Nambiar S, Laessig K, Toerner J, Farley J, Cox E. Antibacterial drug development: challenges, recent developments, and future considerations. Clin Pharmacol Ther. 2014 Aug;96(2):147-9.
5. Boucher HW. Challenges in anti-infective development in the era of bad bugs, no drugs: a regulatory perspective using the example of bloodstream infection as an indication. Clin Infect Dis. 2010 Jan;50 Suppl 1:S4-9.
6. Uchino S, Bellomo R, Goldsmith D, Bates S, Ronco C. An assessment of the RIFLE criteria for acute renal failure in hospitalized patients. Crit Care Med. 2006 Jul;34(7):1913-7.

Presented at IDWeek 2017, October 4-8, 2017, San Diego, CA