696

IDweek2018
San Francisco, CA
3-7 October, 2018

Mechanism of Cefiderocol High MIC Mutants Obtained in Non-Clinical FoR Studies

A. Ito, T. Nishikawa, R. Ishii, M. Kuroiwa, Y. Ishioka, N. Kurihara, I. Sakikawa, T. Ota, M. Rokushima, M. Tsuji, T. Sato, Y. Yamano
Shionogi & Co., Ltd., Osaka, JP

Contact Information:
Akinobu Ito
Address: 3-1-1, Futaba-cho, Toyonaka, Osaka, 561-0825, JAPAN
TEL: +81-6-6331-7144 FAX: +81-6-6331-8612
E-mail address: akinobu.ito@shionogi.co.jp

ABSTRACT

Background : Cefiderocol (S-649266, CFDC) is a novel siderophore cephalosporin with activity against a wide variety of Gram-negative bacteria including carbapenem-resistant strains. We previously reported that CFDC is efficiently transported into *Pseudomonas aeruginosa* via iron transporter PiuA. In this study, we examined frequency of resistance of *P. aeruginosa* to CFDC, and investigated the resistance mechanisms of post-exposure colonies.
Method : Frequency of resistance (FoR) was determined by plating an overnight culture of *P. aeruginosa* PAO1 on Mueller-Hinton Agar containing 4× or 10×MIC of CFDC or ceftazidime (CAZ). Surviving colonies were analyzed by whole genome sequencing (WGS) to identify genomic mutations. The mRNA expression was determined by real time RT-PCR, and pyoverdine production was determined by MALDI-TOF/MS and expression of outer membrane protein was analyzed by SDS-PAGE and proteomic analysis. *In vivo* efficacy was determined by using neutropenic murine systemic infection model, and the bactericidal activity and the risk of resistance emergence were evaluated by using *in vitro* PD model study.
Results : MICs of CFDC and CAZ against PAO1 were 0.5 and 1 µg/mL, respectively. The FoR to CFDC were 2.9×10^{-8} and $<7.1 \times 10^{-9}$, which were lower than those to CAZ (3.1×10^{-7} and 3.4×10^{-7}) in the conditions of 4× and 10×MIC, respectively. MIC of CFDC against CFDC-derived mutants increased from 0.5 µg/mL (MIC against PAO1) to 2 µg/mL, while the MICs of CAZ did not increase. In the case of CAZ-derived mutants, MICs of CAZ increased from 1 µg/mL (MIC against PAO1) to 32 µg/mL, though MIC of CFDC did not increase, suggesting no cross-resistance between CFDC and CAZ. WGS identified mutations in upstream regions of *pvdS* (p*pvdS* mutant), which regulates pyoverdine synthesis, or *fecI* (p*fecI* mutant), which regulates the synthesis of iron transporter FecA contributing to the transport of iron citrate. The *pvdS* expression and pyoverdine production in the *pvdS* mutant were more than 4- and 6-fold higher than those in PAO1, respectively. The expression of *fecA* in the fecI mutant was more than 9-fold higher than that in PAO1.
Conclusion : The MIC increase of CFDC against *P. aeruginosa* occurred due to the mutation of iron transporter related genes, including the regulation of pyoverdine. The resistance acquisition risks should be low as the frequency of resistance to CFDC was lower and the MIC increase of CFDC against the mutants was smaller than that of CAZ. In addition, no cross-resistance between CFDC and CAZ was observed.

INTRODUCTION

Cefiderocol (S-649266, CFDC) is a novel siderophore cephalosporin with activity against a wide variety of Gram-negative bacteria. Here, we investigated the frequency of resistance (FoR) to CFDC as well as ceftazidime and identified potential resistance mechanisms through whole genome sequencing and proteomic analysis.

MATERIALS

Bacterial strains
P. aeruginosa PAO1 was used for FoR experiments. The *pvdS* and *fec* genes were cloned into the plasmid pMMB67HE/GM and transformed with PAO1.

METHODS

Minimum inhibitory concentration
Minimum inhibitory concentration (MIC) was determined by using broth microdilution method according to Clinical and Laboratory Standards Institute (CLSI) guidelines (1, 2). For the determination of CFDC MIC, iron-depleted cation-adjusted Mueller Hinton broth (ID-CAMHB) was prepared as previously described (3), and used according to the recommendation by CLSI. The quality control MIC ranges of CFDC using ID-CAMHB approved by CLSI were 0.06 to 0.5 µg/mL for both *Escherichia coli* ATCC 25922 and *P. aeruginosa* ATCC 27853 (4).
Evaluation of Frequency of resistance acquisition (FoR)
Overnight cultures (approximately 10^7 or 10^8 cells/100 µL) were plated on Muller-Hinton agar containing 4 and 10 times the MIC of CFDC or CAZ, and incubated at 37°C for 24 to 48 hours. The number of the resistant colonies was counted to determine the FoR. The MICs against the isolated colonies were determined.
Whole genome sequencing
Genomic DNA was extracted from overnight cultures using the illustra bacteria genomicPrep Mini Spin Kit (GE Healthcare), and unique index-tagged libraries were generated using Nextera XT DNA Sample Prep Kit (illumina) according to the manufacturer's instructions. Whole genome sequencing was performed using the MiSeq system (ilumina) with 300-base paired-end reads. The fastq reads were trimmed and assembled into contigs for each test strains using CLC Genomics workbench 9.5.3 (CLC Gx) (QIAGEN). To detect genomic mutations of individual isolates, trimmed reads were mapped against the reference genomes of and variant calls were performed using CLC Gx. Called variants were compared among the parent and FoR strains using Pipeline Pilot version 9.5 (PP) (Dassault Systèmes BIOVIA). Detected mutations were confirmed by Sanger sequencing using BigDye Terminator v3.1 (Thermo Fisher Scientific).
Determination of mRNA expression and pyoverdine production
The mRNA expressions of *pvdS* and *fecA* in the exponential growth phase were determined by real time RT-PCR, and the amount of pyoverdine in the supernatant of the overnight culture was determined by MALDI-TOF/MS.
Proteomic analysis
The cell free extract obtained at exponential growth phase was used for the proteomic analysis. Protein extracts were separated by SDS-PAGE (5 to 12.5% gradient gel, D.R.C, Japan) and visualized by SYPRO Ruby gel staining (Thermo Fisher Scientific). The protein bands were excised from stained gels. Proteins were digested in the gel with trypsin (Promega) and lysyl endopeptidase (Wako Pure Chemical Industries, Japan) (5). The resulting peptides were extracted, and samples were subjected to the analysis by mass spectrometry using the Q Exactive (Thermo Fisher Scientific).
Neutropenic murine systemic infection model
JCl:ICR male mice were rendered neutropenic by administration of cyclophosphamide (Shionogi, Japan) intraperitoneally with 150 and 100 mg/kg at 4 and 1 days before infection, respectively. Mice were infected intraperitoneally with approximately 1.0×10^6 CFU/mouse of bacterial suspension in 0.5 mL of mucin. Antibiotics were administered subcutaneously 1 and 5 hours after infection. The 50% effective doses (ED_{50}s) were calculated by the logit method using the survival numbers at each dose 7 days after infection by using Windows SAS program (SAS® Version 9.4).
In vitro PD model study
An *in vitro* computer programmed pharmacokinetics simulation system on the basis of human plasma concentration was used to evaluate the bactericidal activity and the risk of resistance emergence. ID-CAMHB was used as the test medium for CFDC and CAMHB was used for CAZ. The temperature of the model was kept at 37°C. The concentration of the antibiotic was controlled by adding the antibiotic solution and/or test medium as well as discarding the bacterial culture by computer programmed system. The PK parameters of cefiderocol (2g tid, 3-hour infusion) and ceftazidime (2g tid, 2-hour infusion) were described previously. The viable cell counts were determined on Mueller Hinton agar.

RESULTS

Table 1. Frequency of resistance (FoR) of *P. aeruginosa* PAO1.

	Frequency of Resistance [a] at:	
	4 × MIC	10 × MIC
Cefiderocol	2.9×10^{-8}	$<7.1 \times 10^{-9}$
Ceftazidime	3.1×10^{-7}	3.4×10^{-7}

[a] Number of appeared colonies per number of inoculum.

- FoR of *P. aeruginosa* PAO1 to CFDC was lower than those to CAZ.

Table 2. Mutations and MICs of appeared colonies.

	Selection drug	Mutation	MIC (µg/mL)				
			Cefiderocol	Ceftazidime	SMC-3176	MB-1	BAL30072
PAO1	-	-	0.5	1	0.5	0.25	0.5
FoR-1	Cefiderocol	p*pvdS*	2	1	8	16	64
FoR-2	Cefiderocol	p*pvdS*	2	1	4	8	64
FoR-3	Cefiderocol	p*pvdS*	2	0.5	8	16	64
FoR-4	Cefiderocol	p*fecI*	2	0.5	8	8	8
FoR-5	Ceftazidime	*dacB*	0.25	32	NT	NT	NT
FoR-6	Ceftazidime	*dacB*	0.25	32	NT	NT	NT
FoR-7	Ceftazidime	*dacB*	0.5	32	NT	NT	NT

- p*pvdS*, promoter region of *pvdS*. *pvdS* is extracytoplasmic function (ECF) sigma-70 factor, controlling the expression of synthesis of genes for pyoverdine.
- p*fecI*, promoter region of *fecI*. *fecI* is ECF sigma-70 factor, transcriptional activator of the ferric citrate siderophore system.
- *dacB*, probable D-alanyl-D-alanine carboxypeptidase.

- CFDC selected colonies possessing mutations in the promoter region of *pvdS* or *fecI*, which were predicted binding region for ferric uptake regulator (Fur), and CAZ selected colonies possessing mutations in *dacB*.
- MIC of CFDC against p*pvdS* and p*fecI* mutants increased 4-fold, though MIC of CAZ did not increase.
- MIC of CAZ against *dacB* mutants increased 32-fold, though MIC of CFDC did not increase.

Figure 1. mRNA expression of *pvdS* and *fecA.*

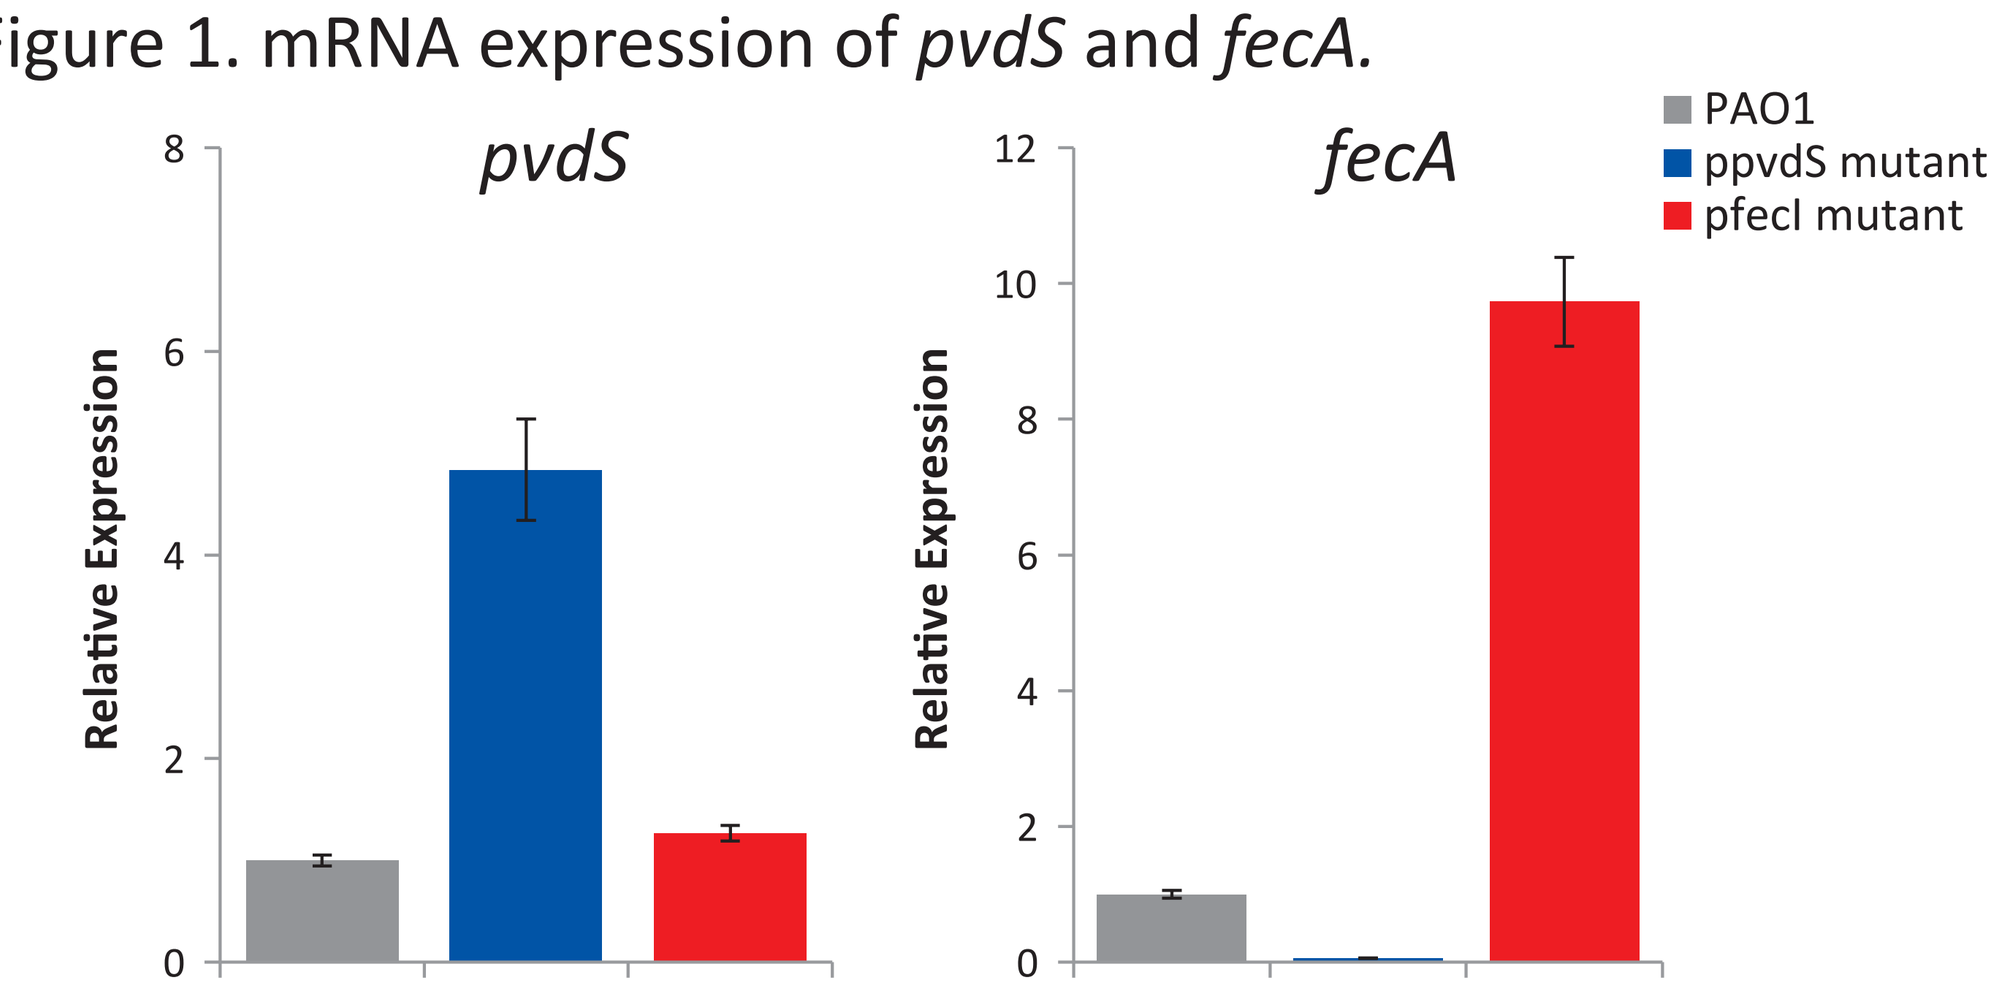
pvdS
fecA
Relative Expression
Relative Expression
PAO1
ppvdS mutant
pfecI mutant

- *pvdS* expression increased in the p*pvdS* mutants.
- *fecA* expression increased in the p*fecI* mutant, and decreased in the p*pvdS* mutants.

Table 3. Effect of overexpression of *pvdS* and *fec* genes on MIC.

	MIC (µg/mL)	
	Cefiderocol	Ceftazidime
PAO1/pMM67HE/GM	0.125	2
PAO1/pMM67HE/GM::*pvdS*	4	2
PAO1/pMM67HE/GM::*fecI*	4	2
PAO1/pMM67HE/GM::*fecR*	0.25	2
PAO1/pMM67HE/GM::*fecA*	4	2

- MIC of CFDC increased by overexpression of *pvdS*, *fecI*, and *fecA*.

Figure 2. Pyoverdine production.

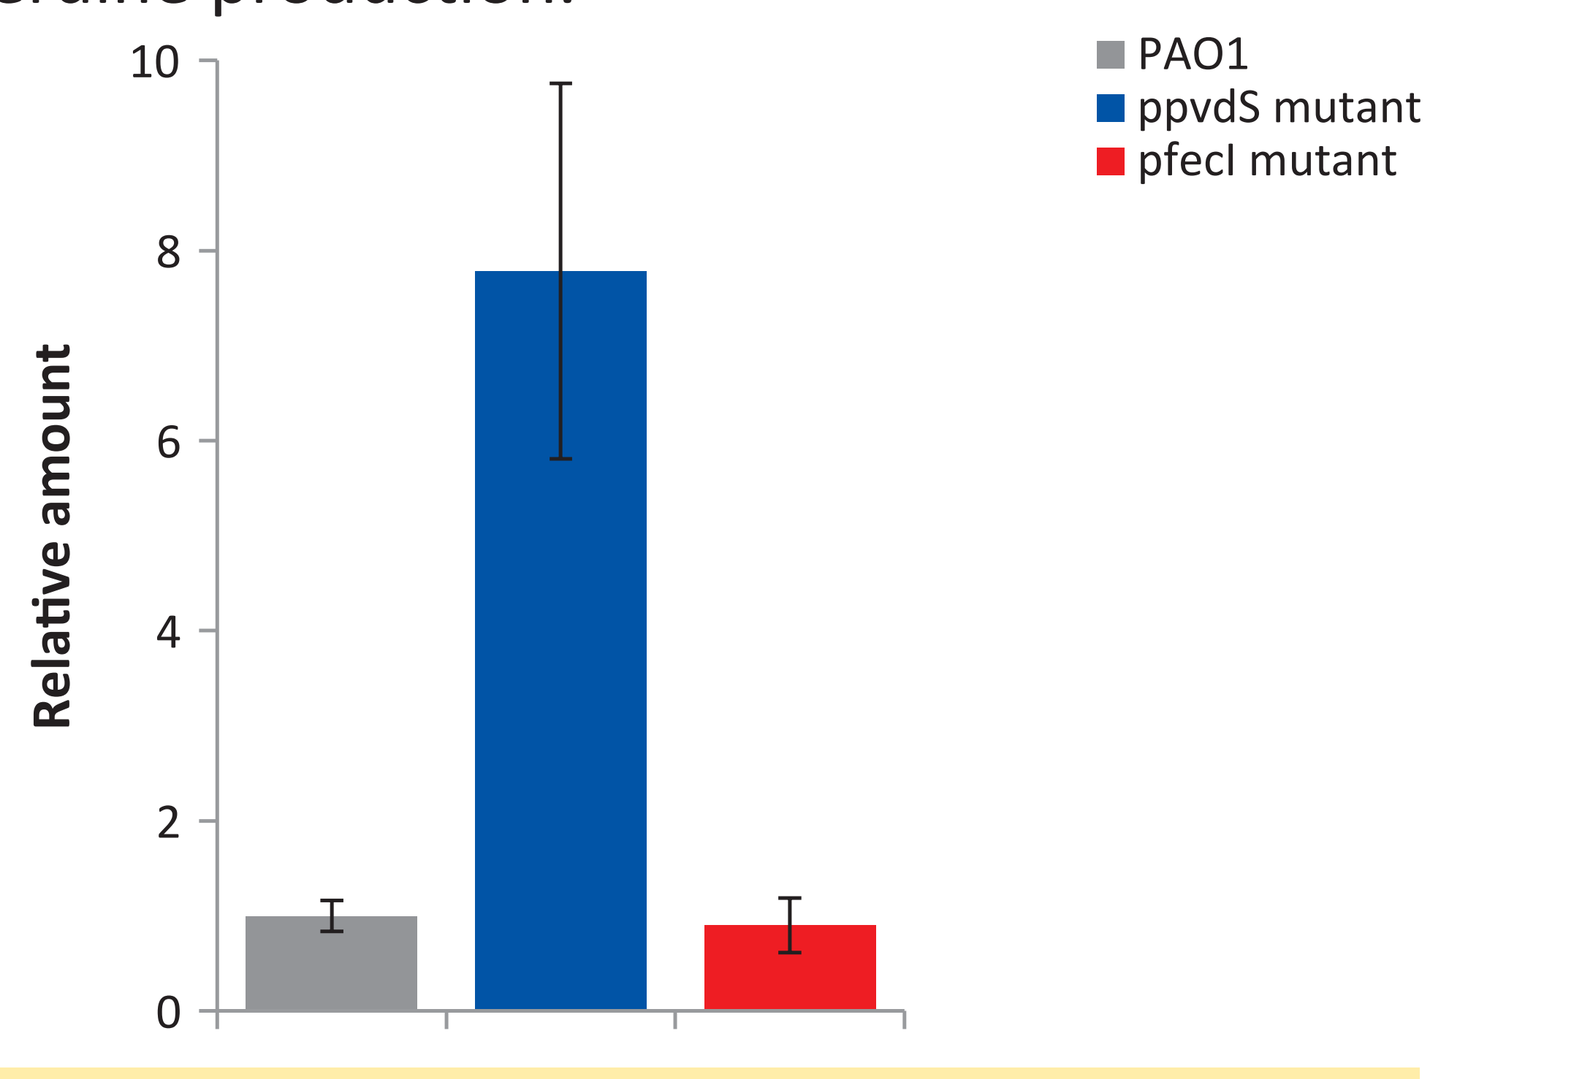
Relative amount
PAO1
ppvdS mutant
pfecI mutant

- Amount of pyoverdine increased in the p*pvdS* mutants.

Figure 3. Proteomic analysis of outer membrane proteins (OMPs).

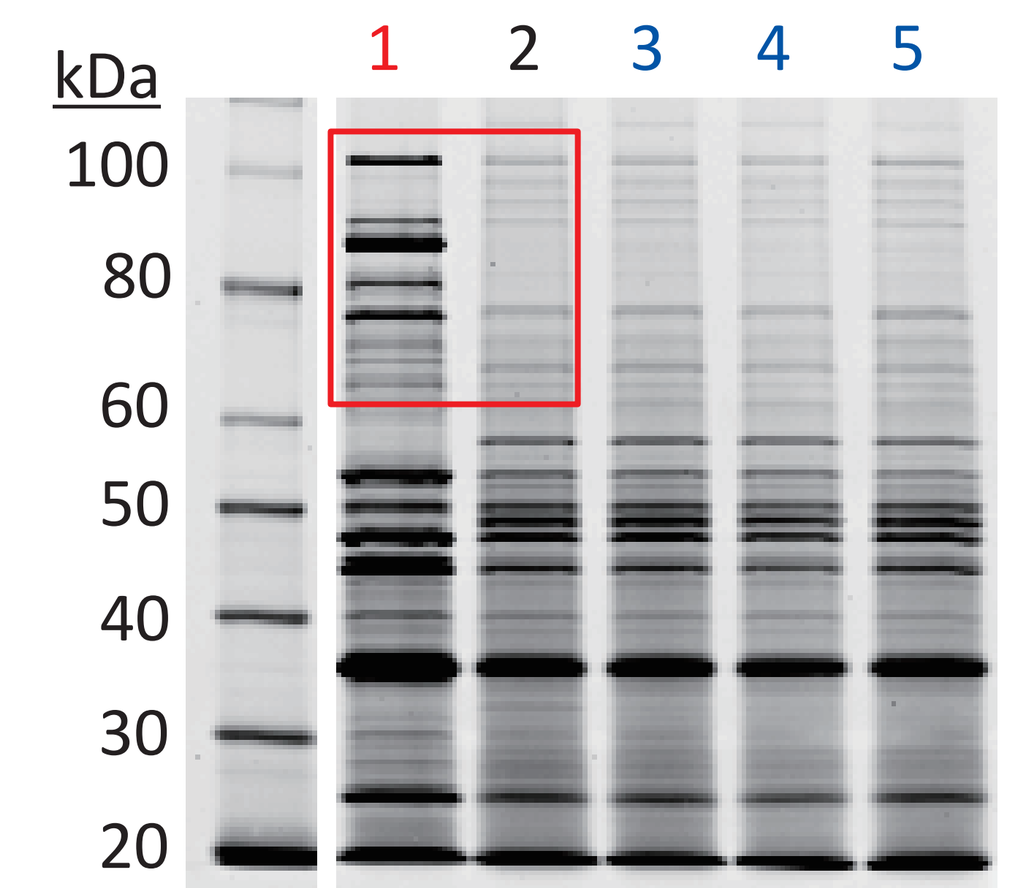
kDa
1
2
3
4
5
100
80
60
50
40
30
20

- 1, p*fecI* mutant
- 2, PAO1
- 3 – 5, p*pvdS* mutant

- Iron regulated OMP (70 –100 kDa) were highly expressed in p*fecI* mutant even in the iron-sufficient condition, and FecA was detected by proteomic analysis.

Figure 4. Mechanism of *fecA* regulation.

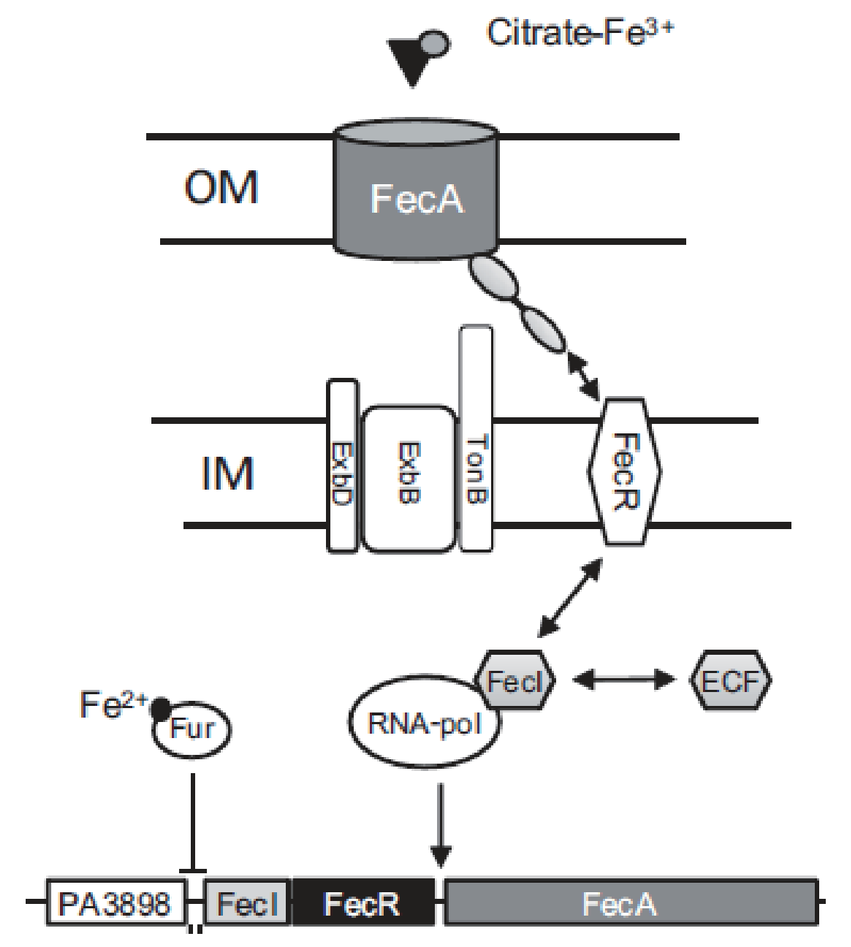
Citrate-Fe3+
OM
FecA
IM
ExbD
ExbB
TonB
FecR
Fe2+
Fur
RNA-pol
FecI
ECF
PA3898
FecI
FecR
FecA

- *fecR*: Inner membrane sensor for Fe^{2+}-dicitrate
- *fecA*: Outer membrane receptor for Fe^{3+}-dicitrate

Delden et al., 2013, AAC 57:2095 -2102

Figure 5. *In vivo* efficacy of CFDC against the mutants.

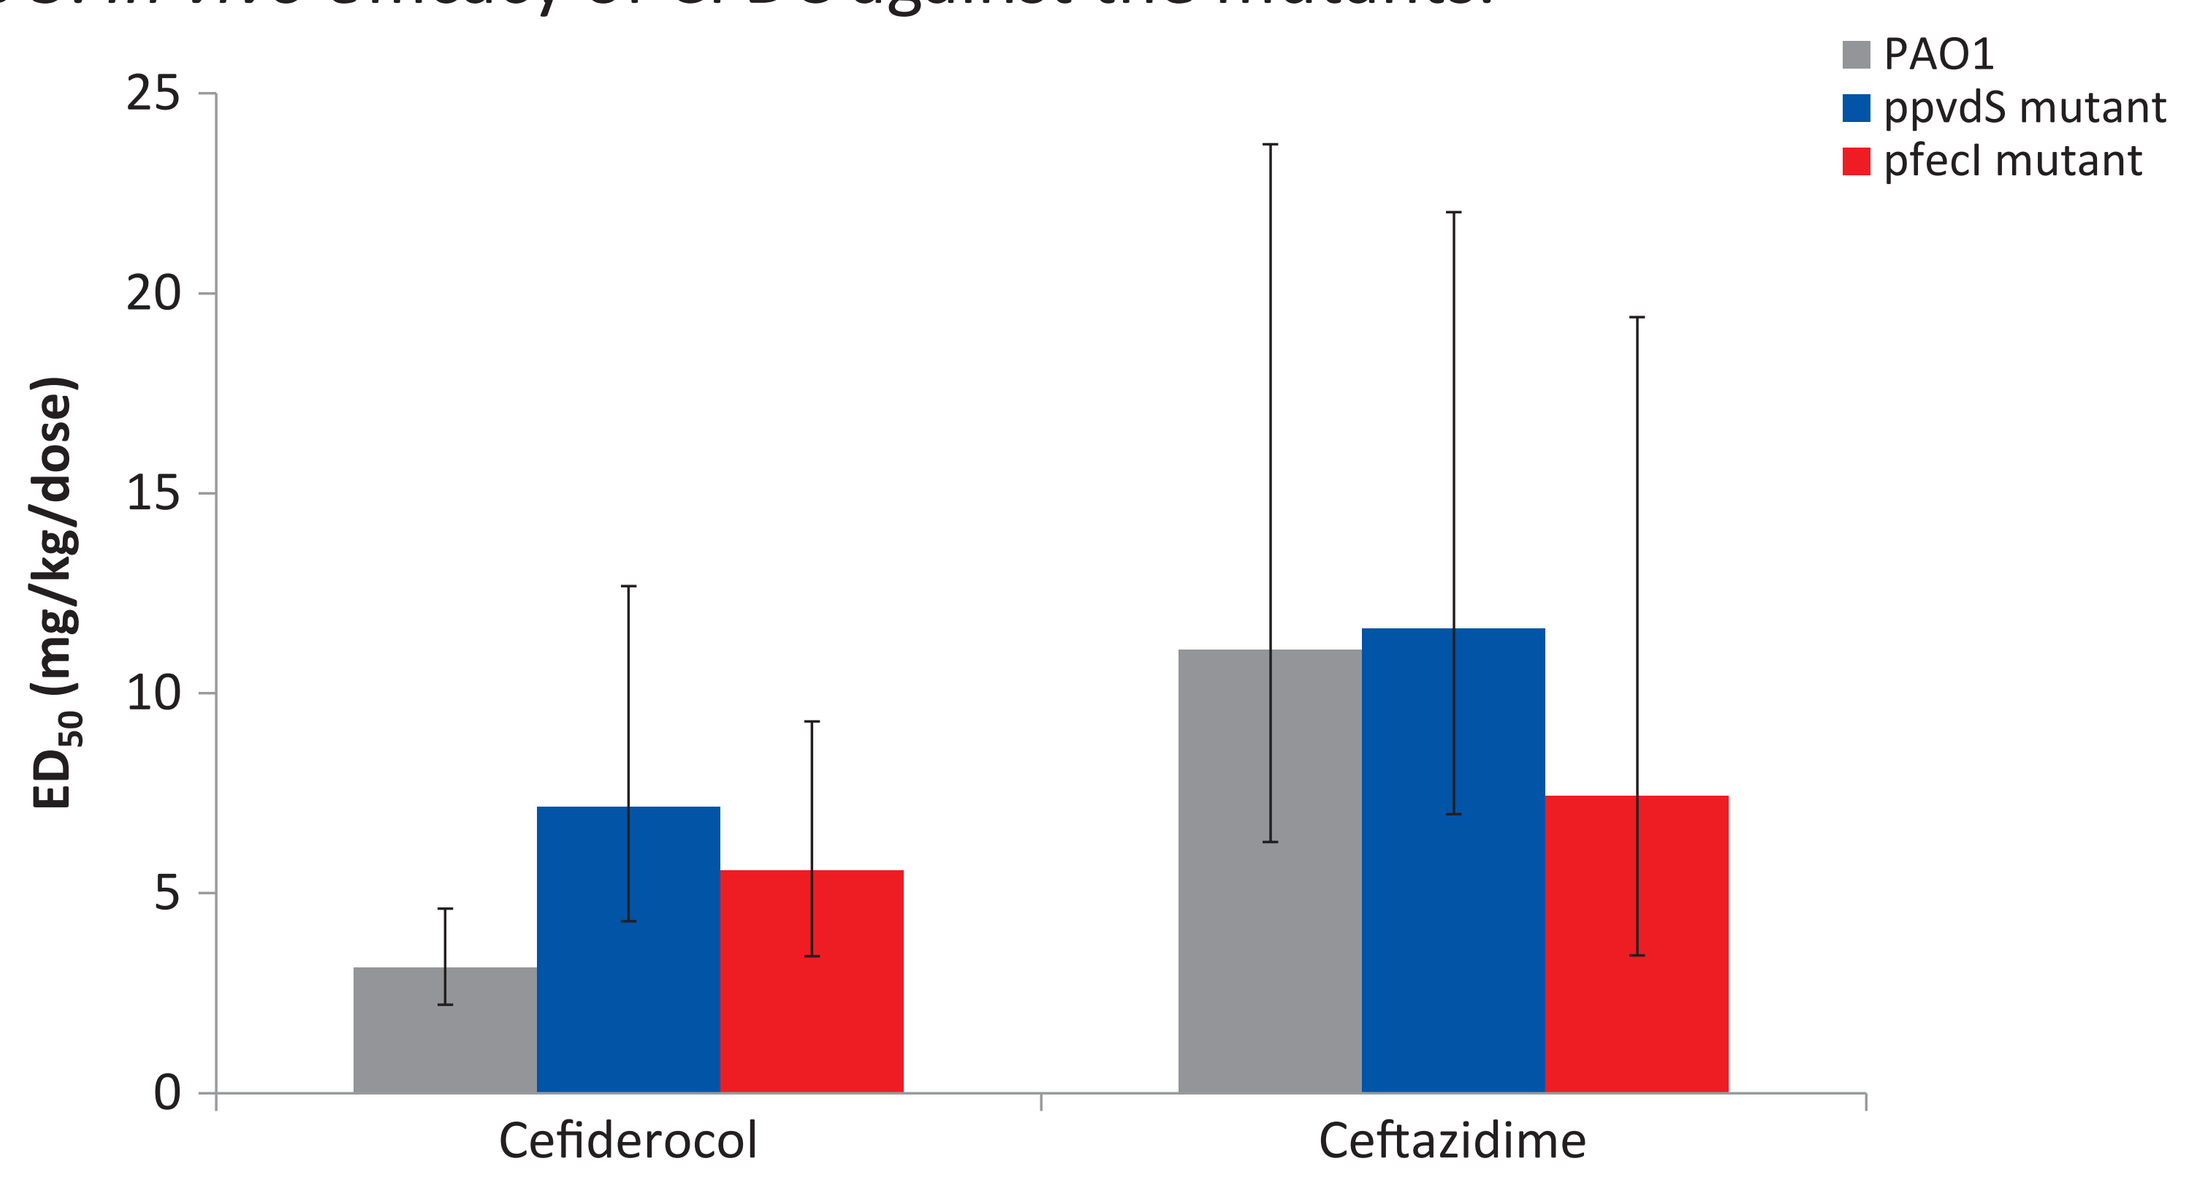
ED50 (mg/kg/dose)
Cefiderocol
Ceftazidime
PAO1
ppvdS mutant
pfecI mutant

- ED_{50} of CFDC against mutant strains were higher than that against PAO1, though fold change were less than 3-fold.
- Two-fold decrease of *in vivo* efficacy was observed due to the mutation in p*pvdS* and p*fecI*, which caused 4-fold increase of MIC.

Figure 6. Bactericidal activity of cefiderocol against PAO1 under human PK exposure recreated by *in vitro* PD model.

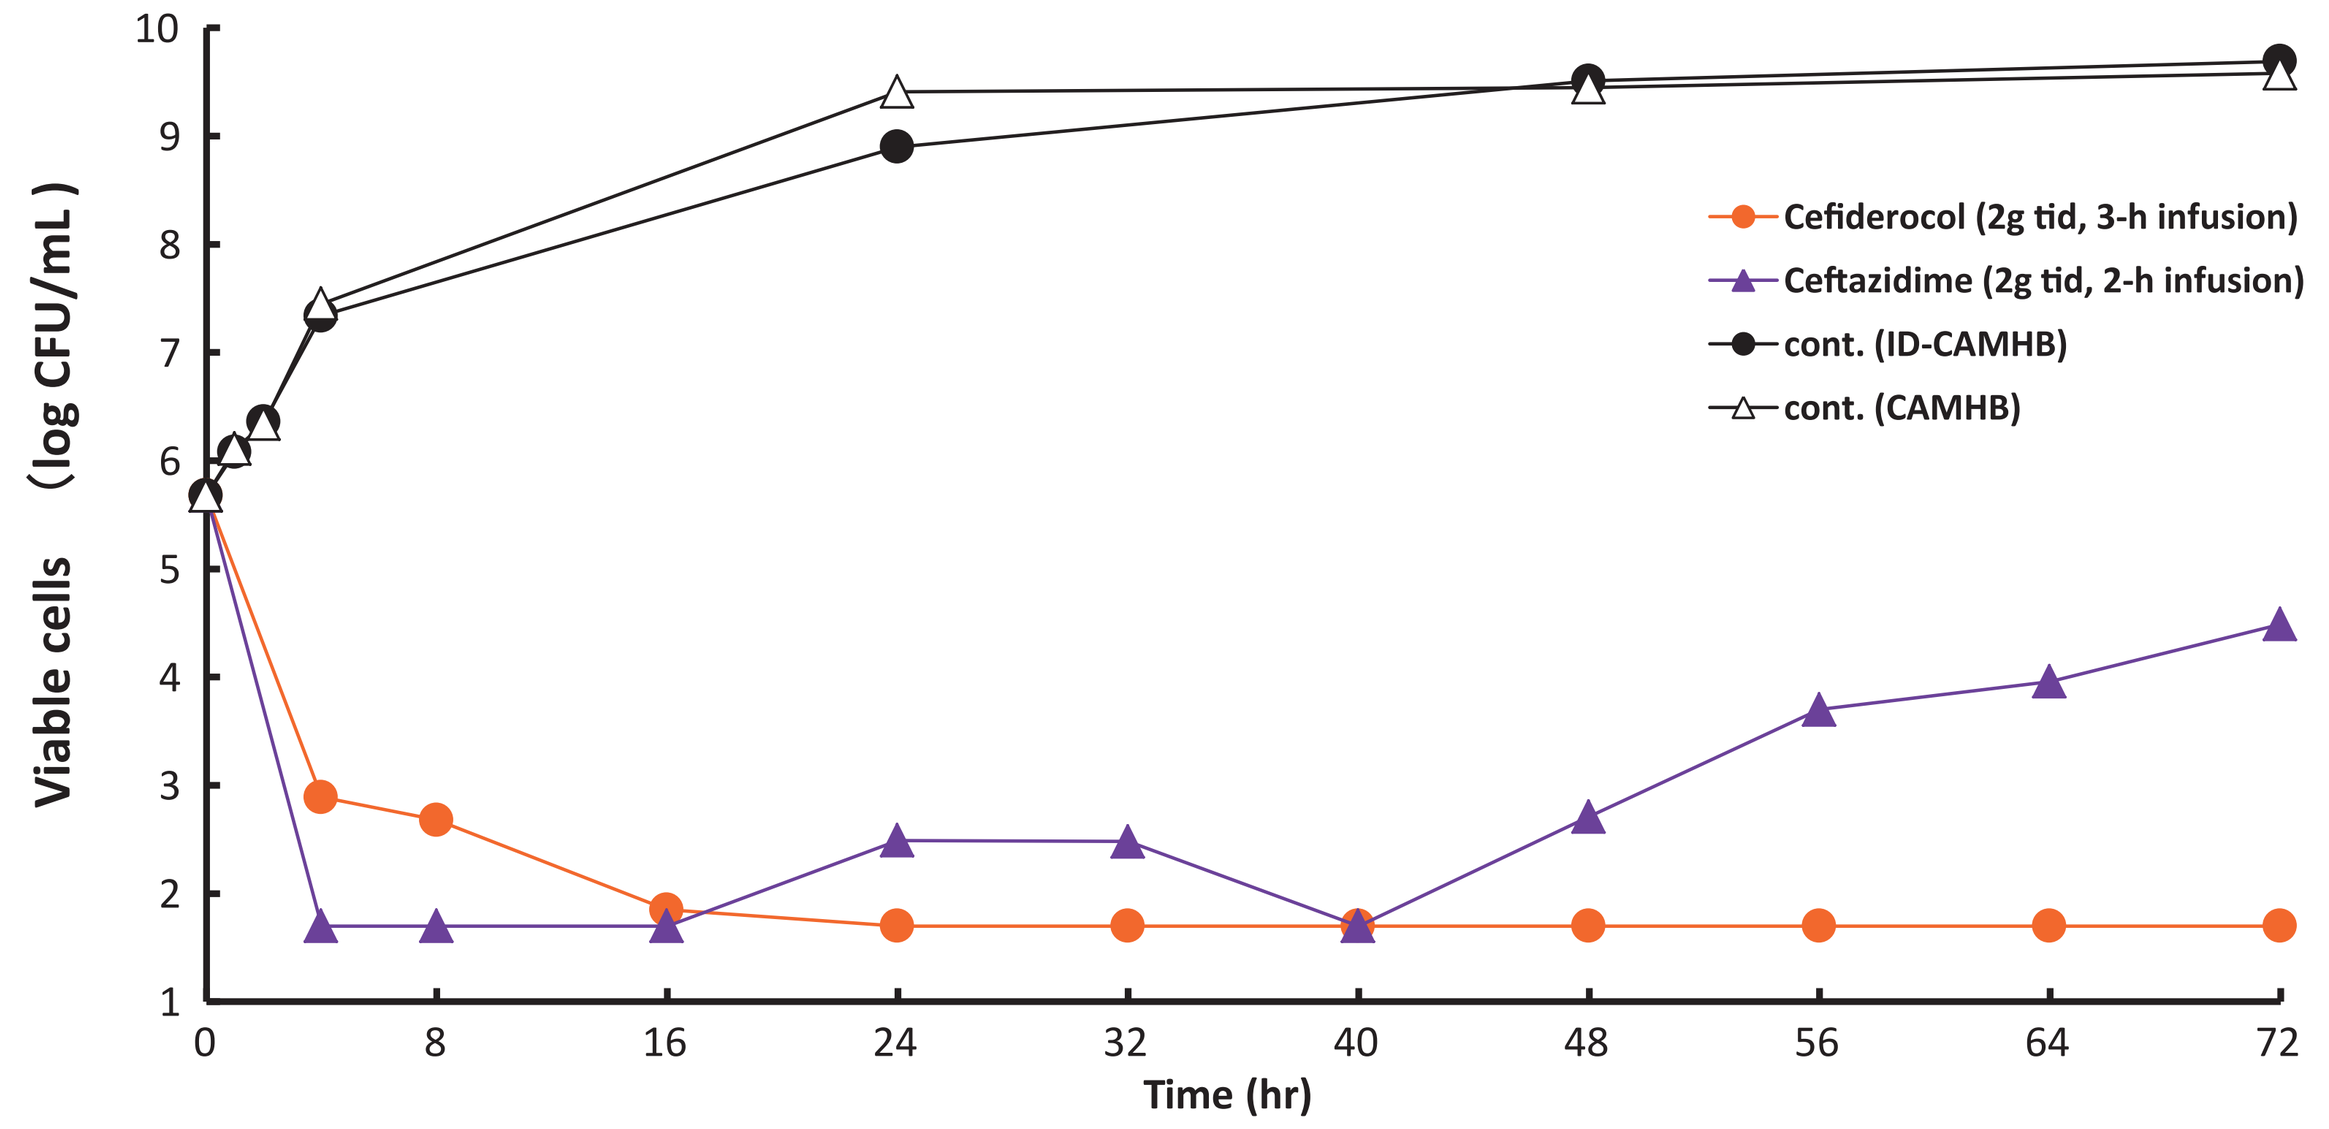
Viable cells (log CFU/mL)
Time (hr)
Cefiderocol (2g tid, 3-h infusion)
Ceftazidime (2g tid, 2-h infusion)
cont. (ID-CAMHB)
cont. (CAMHB)

- CFDC showed more than 3 log reduction of viable cells and did not cause re-growth.
- CFDC showed >3-log reduction of viable cells, and no resistant mutants were detected under human PK exposure, presumably due to the low MIC of the mutants.

CONCLUSIONS

Frequencies of resistance to CFDC were lower than those to CAZ. The colonies appeared by exposure to CFDC possessed mutations in the promoter regions of *pvdS* or *fecI*, and MICs of CFDC against these mutants increased 4-fold. The p*pvdS* mutants showed increased production of pyoverdine, and p*fecI* mutant showed increased expression of FecA OMP. These genes may contribute to the resistance of *P. aeruginosa* to CFDC, though *in vitro* PD model demonstrated that the risk of resistance emergence during the treatment with CFDC was low.

REFERENCES

1. Clinical and Laboratory Standards Institute (CLSI), 2015. M07-A10.
2. Clinical and Laboratory Standards Institute (CLSI), 2016. M100-S26.
3. Ito A et al. Antimicrob Agents Chemother 2016; 60:7396-7401.
4. Huband MD et al. Diagn Microbiol Infect Dis 2017; 88:198-200.
5. Morita A et al. Proteomics. 2006; 6: 5880-5890.
6. Delden et al., Antimicrob Agents Chemother 2013; 57:2095-2102.