Therapeutic Effects of Baloxavir Marboxil against Influenza A Virus Infection in Ferrets

Mitsutaka Kitano, Takanobu Matsuzaki, Ryoko Oka, Kaoru Baba, Takahiro Noda, Yuki Yoshida, Kenji Sato, Ryu Yoshida, Akihiko Sato, Hiroshi Kamimori, Takao Shishido, and Akira Naito ; SHIONOGI & CO., LTD., Osaka, Japan

SHIONOGI & CO., LTD.

Contact Information:
mitsutaka.kitano@shionogi.co.jp
No conflicts of interest to declare

Introduction

Seasonal influenza remains major public health concerns worldwide despite the availability of antiviral drugs and vaccines, therefore more effective novel anti-influenza drugs are needed. The baloxavir acid (BXA), derived from the prodrug baloxavir marboxil (BXM), potently and selectively inhibits the cap-dependent endonuclease within the polymerase PA subunit of influenza A and B viruses, including oseltamivir-resistant strain and avian strain (H7N9, H5N1) [1-3]. BXM was approved in February 2018 for the treatment of influenza A and B viruses in adult and pediatric patients and marketed under the brand name Xofluza™ in Japan. In this study, pharmacokinetic profiles of BXM and BXA were firstly examined in ferrets, and then the therapeutic effects of BXM against influenza A virus infection were compared with that of oseltamivir phosphate in ferrets.

Study Design

Pharmacokinetic study: Ferrets (n=4 per group) were given BXM (10 or 30 mg/kg) suspension orally using intragastric tube. To obtain the plasma exposure of BXA in ferrets, water containing 5% SDS and 10% Tween 80 were used as a vehicle. Blood samples were collected from 0.5, 1, 2, 4, 6, 8, and 24 hours after dosing and centrifuged to obtain plasma. The concentrations of BXA and BXM in plasma were determined by liquid chromatography-tandem mass spectrometry (LC/MS/MS). The pharmacokinetic parameters of BXA and BXM were calculated using WinNonlin software based on a non-compartment model analysis with uniform weighting.

Efficacy study-1: On day 1 post-infection with A/Kadoma/2006 (H1N1) (1000 $TCID_{50}$), ferrets (n=4 per group) were given orally BXM (10 or 30 mg/kg twice daily for 1 day), oseltamivir phosphate (5 mg/kg twice daily for 2 days), or vehicle (water containing 5% SDS and 10% Tween 80) twice daily (12-hour interval between each dosing). Nasal wash fluids were obtained from each ferret on days 1, 2, and 3 post-infection. The virus titer of nasal wash fluids were determined by standard $TCID_{50}$ method. Body temperature was monitored by a data logger (DS1921H-5F; Maxim Integrated Products, Inc.) implanted under the skin of the back of ferret and analyzed using One Wire Viewer version 0.3.15.50 . The body temperature change were expressed by calculating the average temperatures for a 8-hour period of time.

Efficacy study-2: On day 2 post-infection with A/Kadoma/2006 (H1N1) (5000 $TCID_{50}$), ferrets (n=6 per group) were given orally BXM (10 mg/kg twice daily for 1 day), oseltamivir phosphate (5 mg/kg twice daily for 2 days), or vehicle twice daily. The virus titer in nasal wash fluids and change of body temperature of ferrets were evaluated in the same manner as described efficacy study-1.

Figure 1. Plasma concentration -time profiles of BXA and BXM in ferrets after a single oral administration of BXM (Pharmacokinetic study)

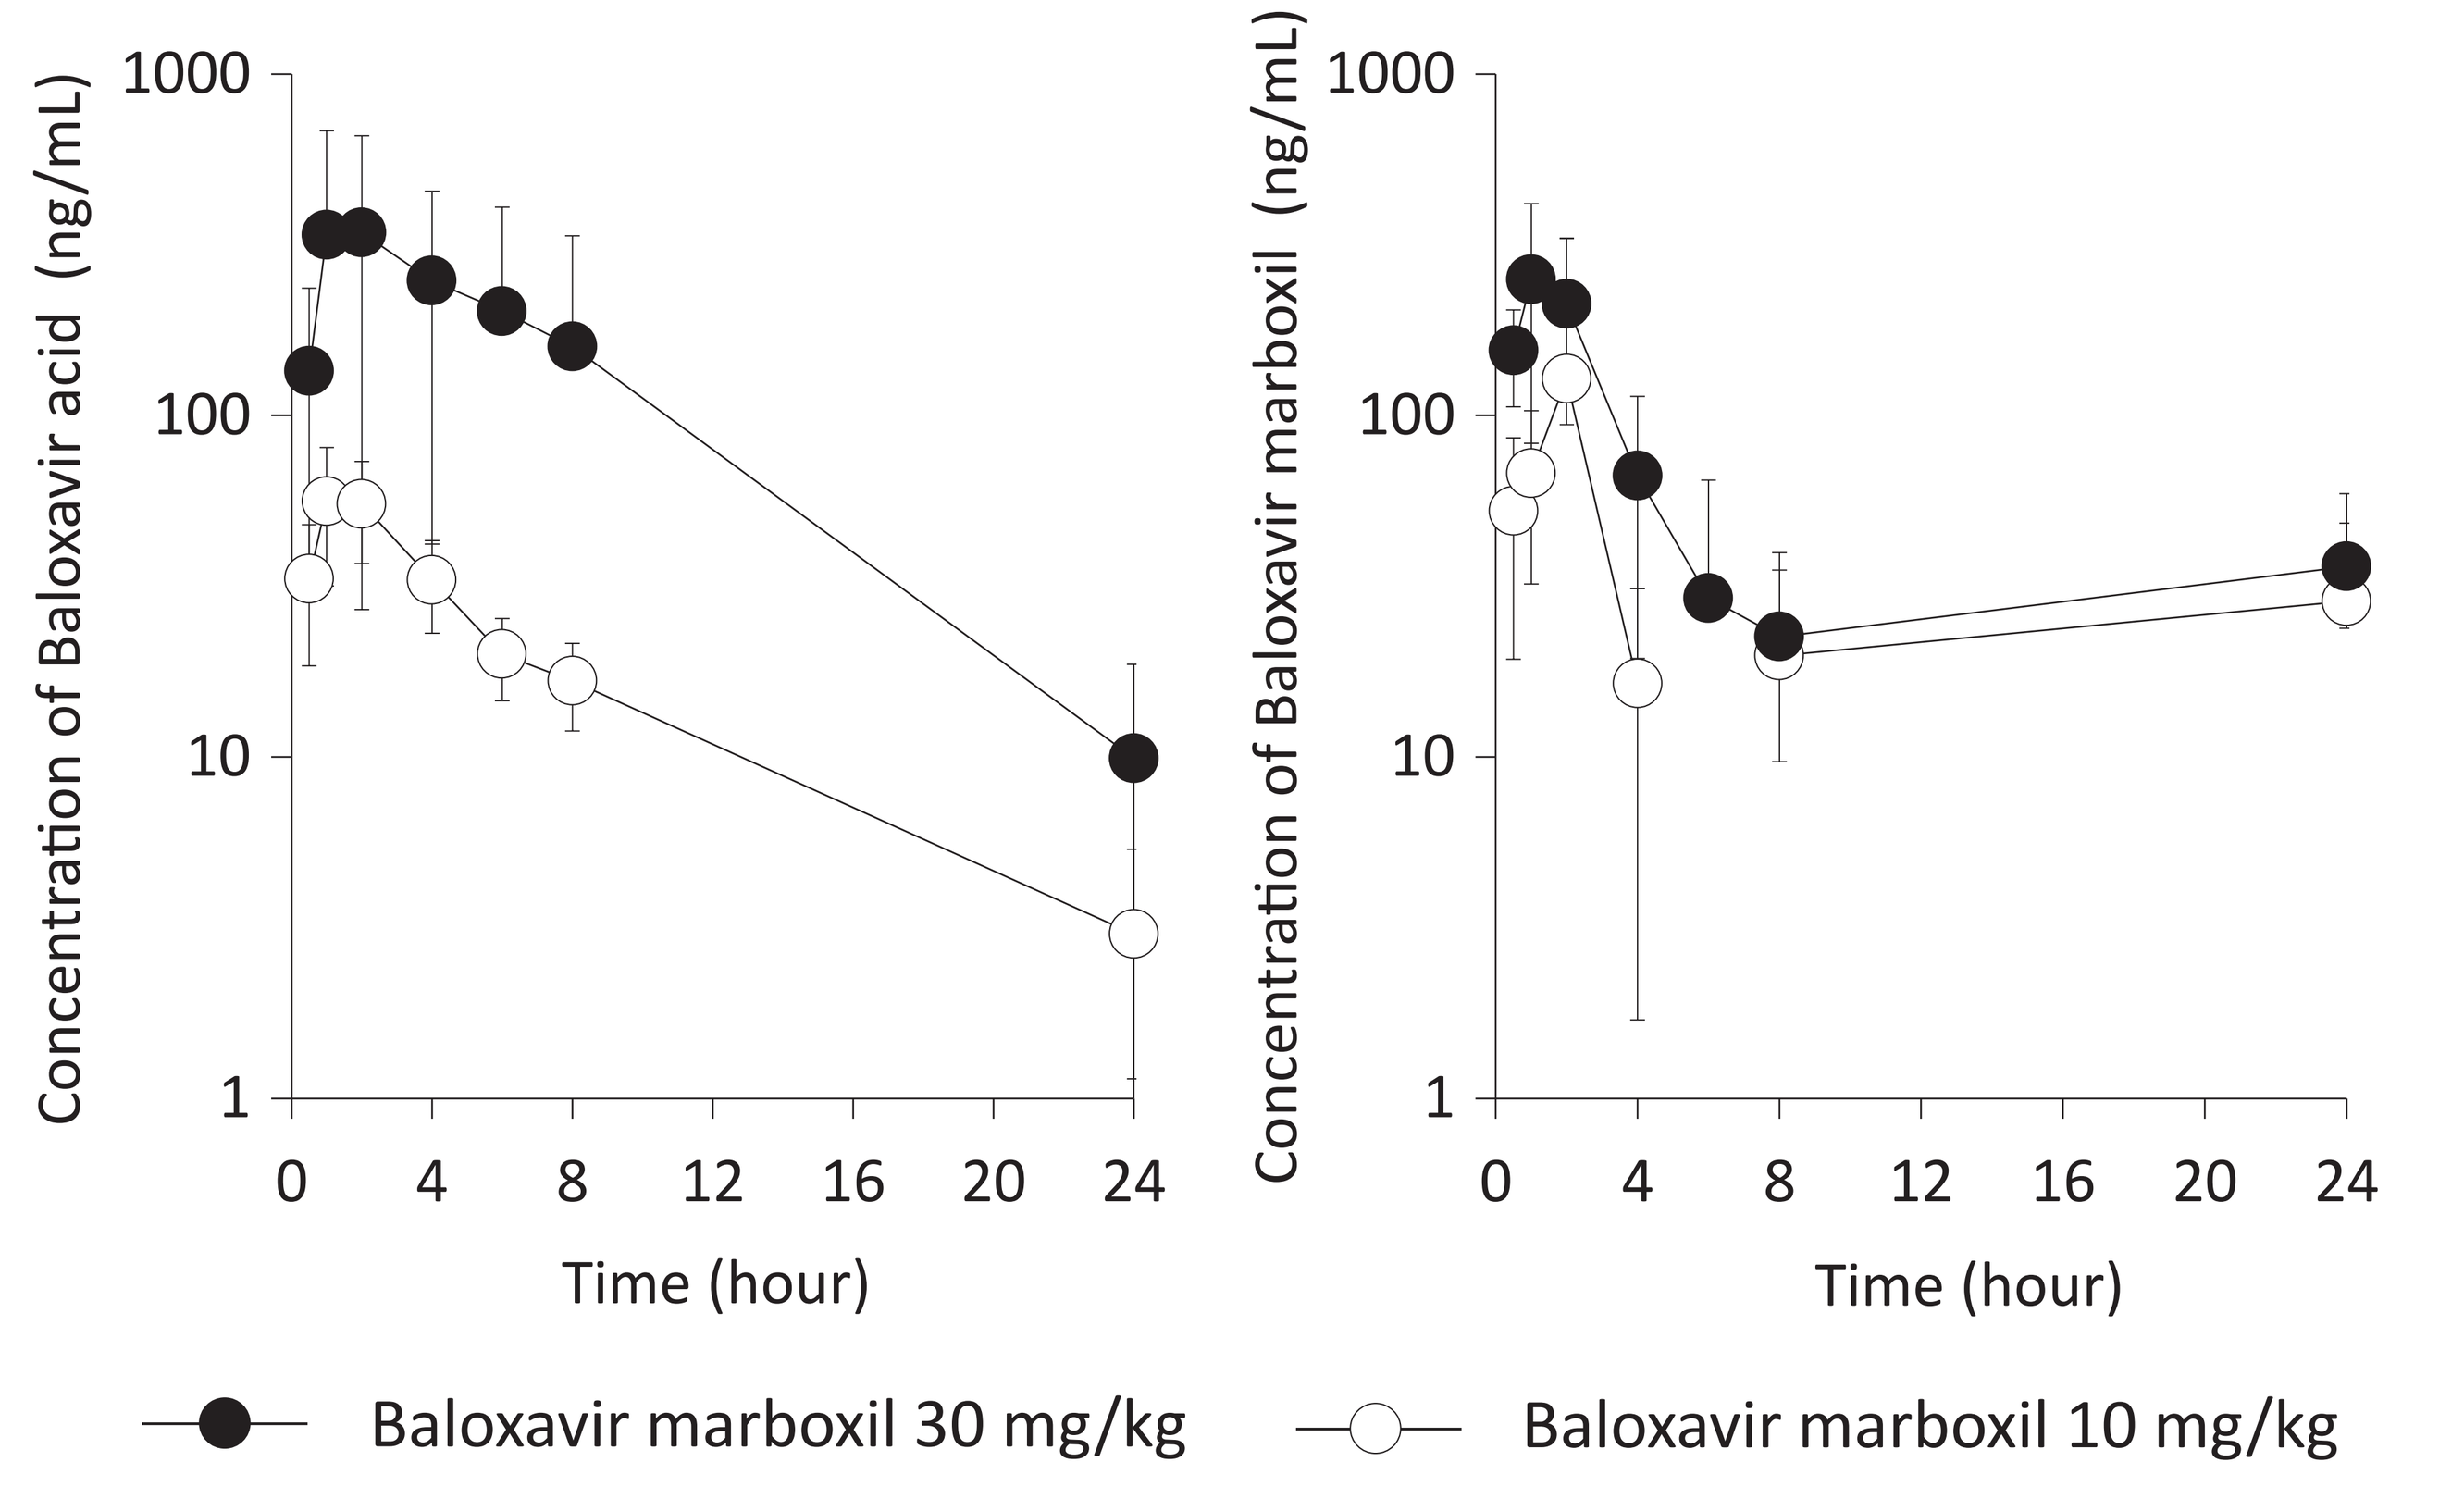
Plasma conc. of baloxavir acid
Plasma conc. of baloxavir marboxil
Concentration of Baloxavir acid (ng/mL)
Concentration of Baloxavir marboxil (ng/mL)
Time (hour)
Baloxavir marboxil 30 mg/kg
Baloxavir marboxil 10 mg/kg

Table 1. Pharmacokinetic parameters of BXA and BXM in ferrets after a single oral administration of BXM

Pharmacokinetic parameters	Baloxavir acid		Baloxavir marboxil	
	10 mg/kg	30 mg/kg	10 mg/kg	30 mg/kg
Cmax (ng/mL)	66.6 ± 14.6	365 ± 316	165 ± 179	298 ± 88
Tmax (hr)	1.50 ± 0.58	2.00 ± 1.41	1.13 ± 0.63	0.875 ± 0.250
$T_{1/2}$ (hr)	6.91 ± 3.79	4.44 ± 0.67	N.C.	N.C.
$AUC_{0\text{-}24hr}$ (ng·hr/mL)	421 ± 119	3240 ± 3240	715 ± 758	1260 ± 150

Data are expressed as the mean ± standard deviation of four ferrets.
N.C., not calculated.

Statistical Analysis

The uniformity of the mean body weight among groups was confirmed by one-way analysis of variance. For the comparison of the virus titers in nasal washes and change from baseline in body temperature over time from day 2 to 3 or day 2 to 4 post-infection among the BXM, oseltamivir phosphate, and the vehicle-treated groups, two-way analysis of variance model was applied. The comparison of change from baseline in body temperature was performed in a similar fashion to the comparison of the virus titers. P values of less than 0.05 were considered to be statistically significant. Statistical analysis was performed by using the statistical analysis software, SAS Version 9.2.

Figure 2. Virus titers and change of body temperature of infected ferrets treated with BXM, starting at 1 day post-infection (Efficacy study-1)

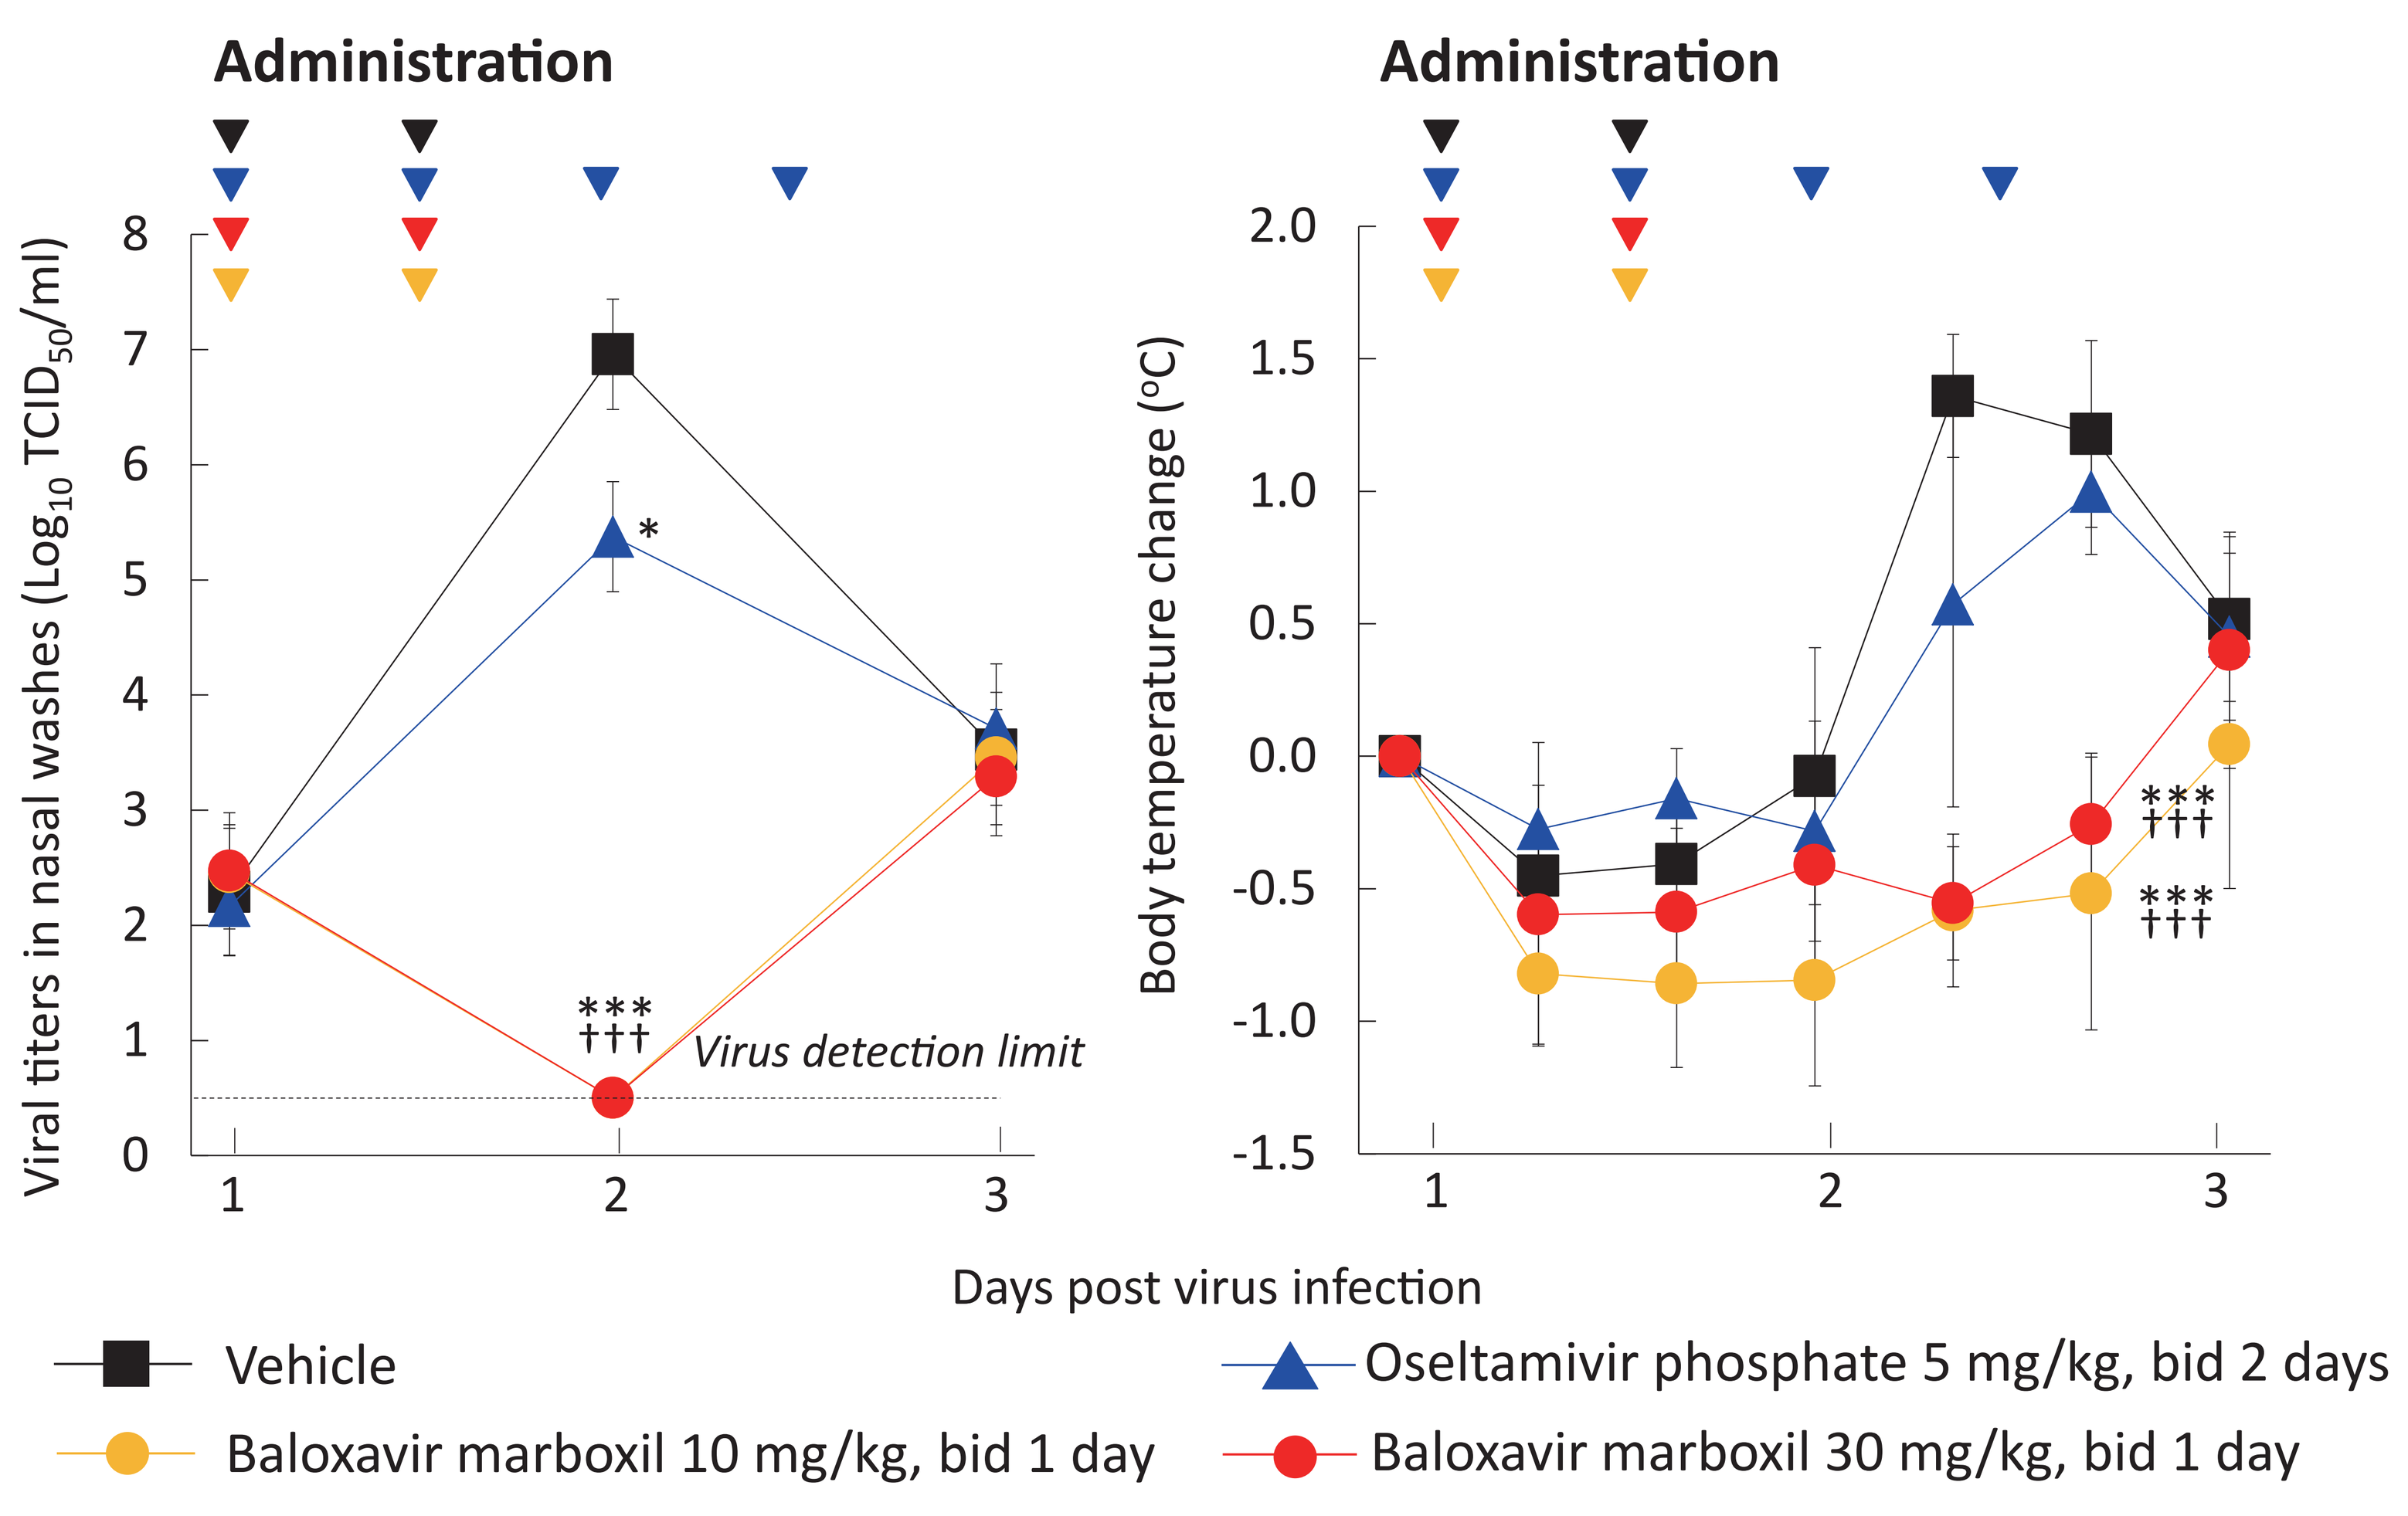
Virus titer in nasal washes
Change of body temperature
Administration
Viral titers in nasal washes (Log_{10} $TCID_{50}$/ml)
Body temperature change (°C)
Virus detection limit
Days post virus infection
Vehicle
Oseltamivir phosphate 5 mg/kg, bid 2 days
Baloxavir marboxil 10 mg/kg, bid 1 day
Baloxavir marboxil 30 mg/kg, bid 1 day

N = 4,*, P < 0.01; **, P < 0.001; ***, P < 0.0001 v.s. vehicle; †††, P < 0.0001 v.s. oseltamivir phosphate 5 mg/kg (two-way analysis of variance model)

Figure 3. Virus titers and change of body temperature of infected ferrets treated with BXM, starting at 2 days post-infection (Efficacy study-2)

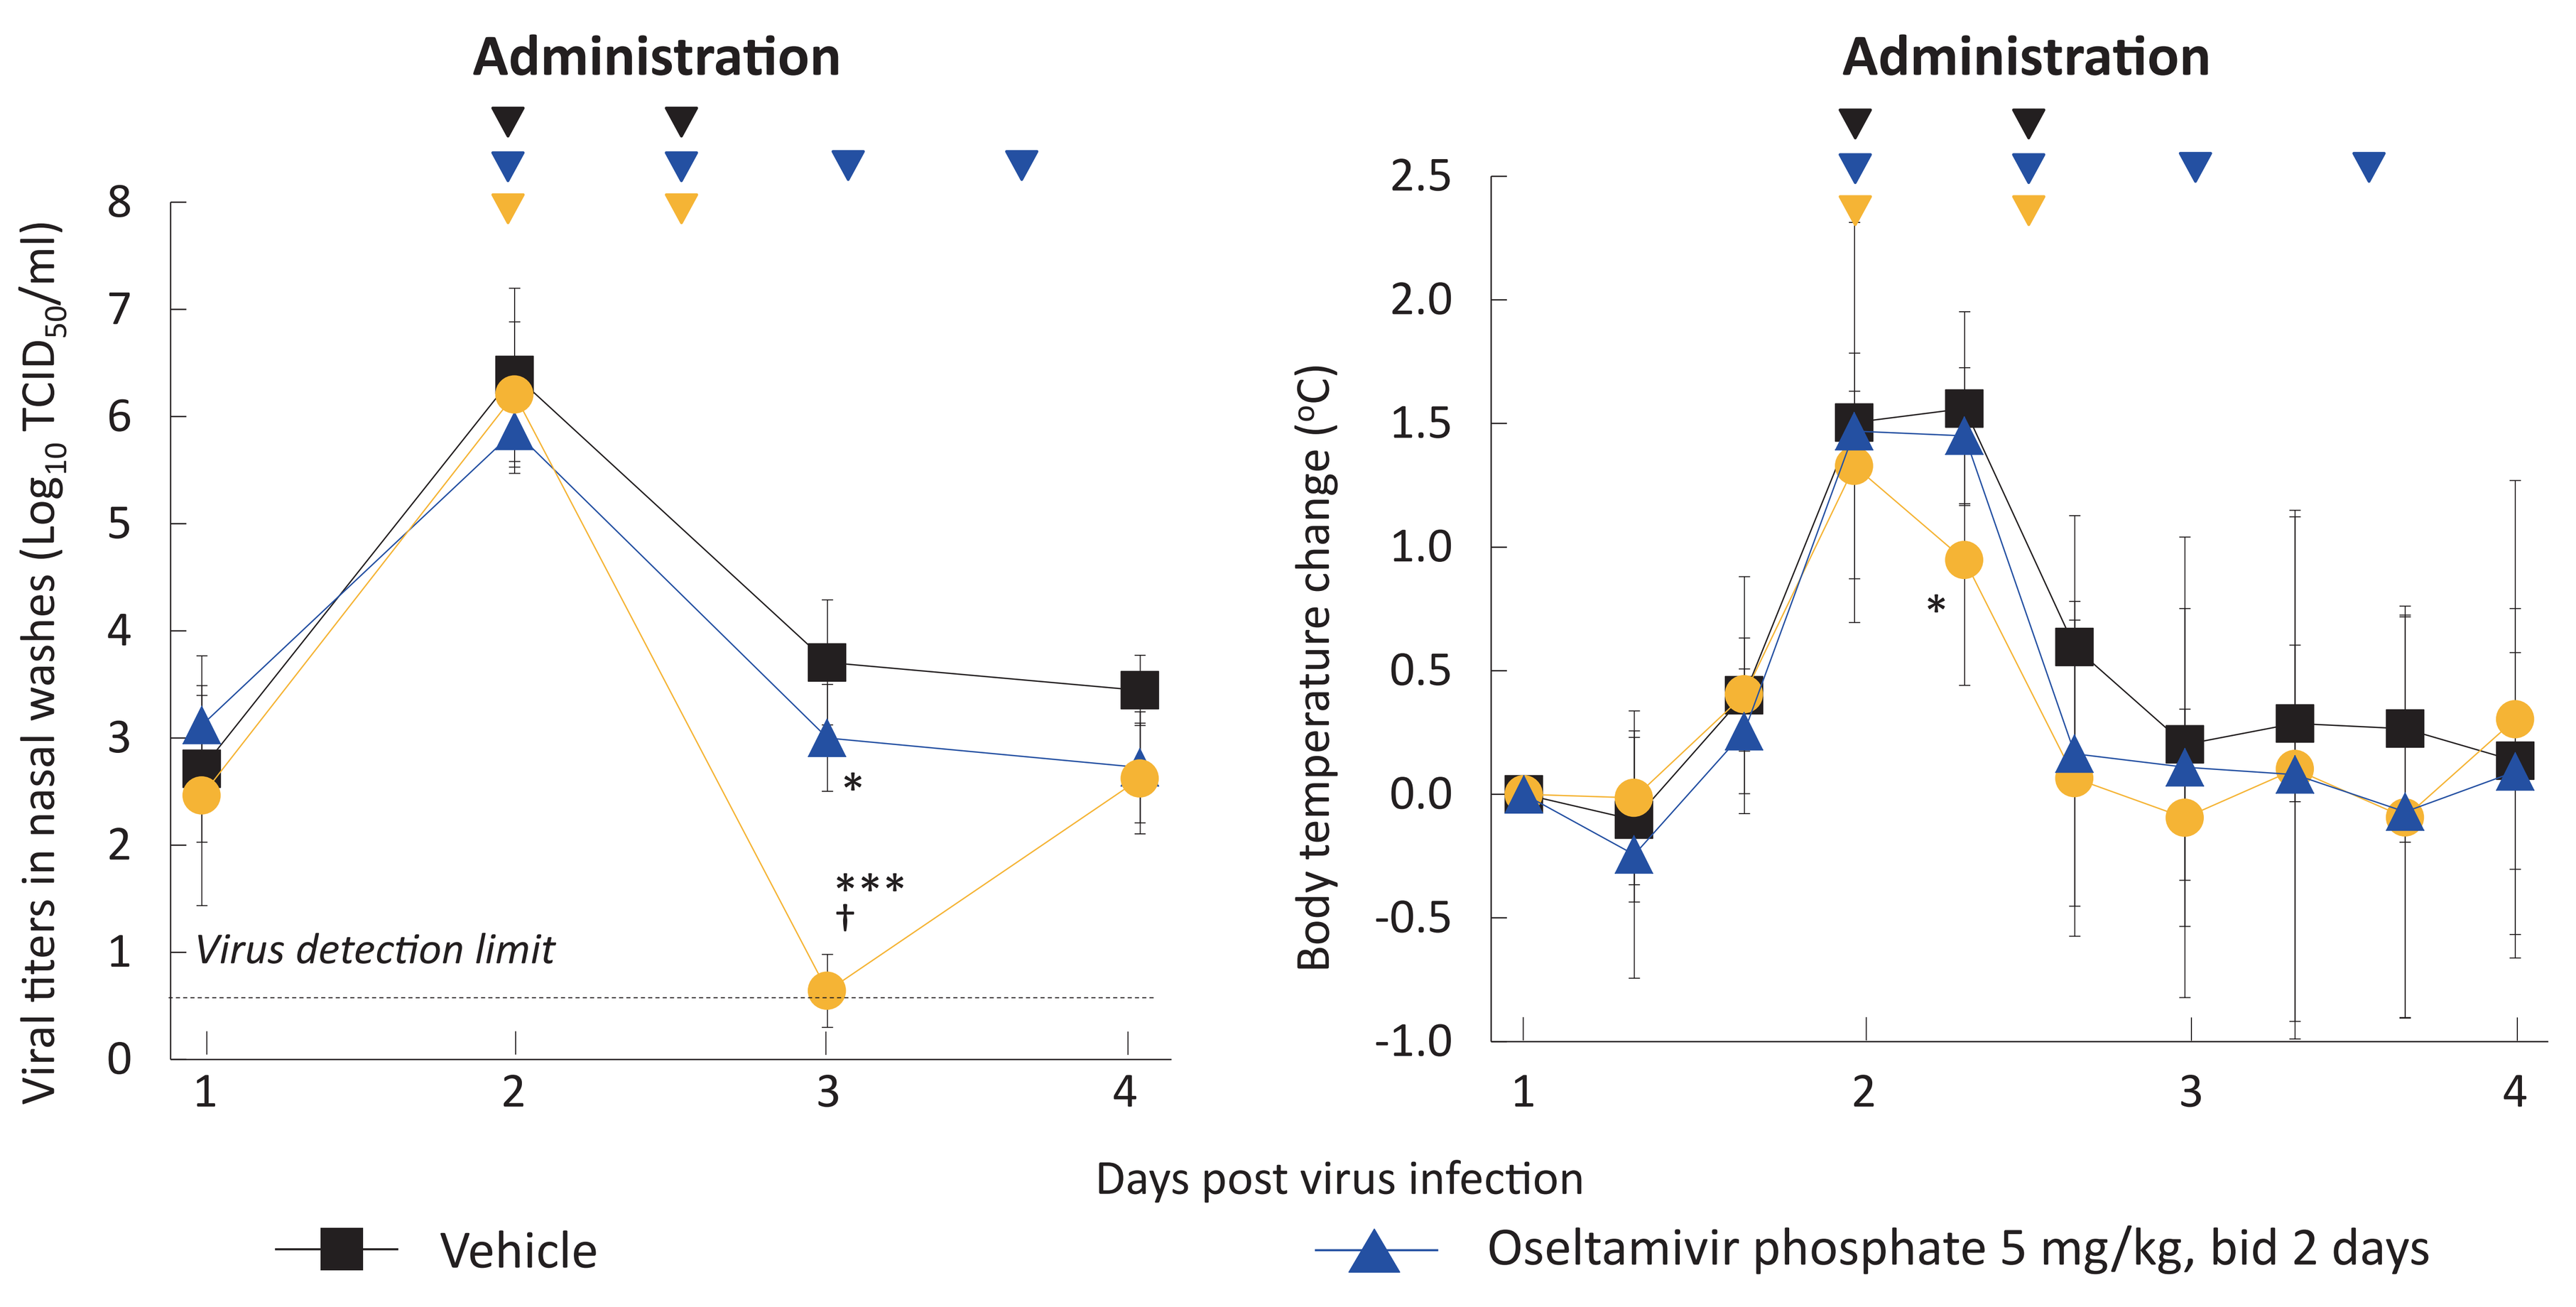
Virus titer in nasal washes
Change of body temperature
Administration
Viral titers in nasal washes (Log_{10} $TCID_{50}$/ml)
Body temperature change (°C)
Virus detection limit
Days post virus infection
Vehicle
Oseltamivir phosphate 5 mg/kg, bid 2 days
Baloxavir marboxil 10 mg/kg, bid 1 day

N = 6, *, P < 0.05; **, P < 0.01; ***, P < 0.001; ****, P < 0.0001 v.s. vehicle; †, P < 0.01 v.s. oseltamivir phosphate 5 mg/kg (two-way analysis of variance model)

Results

Pharmacokinetic study: The plasma concentrations of BXA after the dosing at 10 and 30 mg/kg reached the maximum at 1.50 and 2.00 hours, and then declined with the values of elimination half-life ($t_{1/2}$) of 6.91 and 4.44 hours, respectively. In addition, the plasma concentrations of BXA were 3.03 ± 2.33 and 9.91 ± 8.77 ng/mL at 24 h ours after administration of 10 and 30 mg/kg, respectively. BXM was also detected in ferret plasma after a single oral administration of BXM.

Efficacy study-1: BXM (10 and 30 mg/kg twice daily for 1 day) given to ferrets on 1 day post-infection showed reduction of virus titer to an undetectable level on day 2 post-infection and a statistically significant reduction in virus titer over time from day 2 to 3 post-infection compared with vehicle and clinically-equivalent dose of oseltamivir phosphate (5 mg/kg twice daily for 2 days [4]). In addition, the change of body temperature over time from the first administration to 3 days post-infection was significantly lower in BXM than vehicle and oseltamivir phosphate.

Efficacy study-2 : Even when treatment of BXM (10 mg/kg twice daily for 1 day) was initiated at 2 day post-infection, a significant reduction was observed in virus titer over time from day 2 to 4 post-infection compared with vehicle and oseltamivir phosphate (5 mg/kg twice daily for 2 days). Furthermore, the change of body temperature over time from the first administration to 3 days post-infection was significantly lower in BXM than vehicle. The viral titers in ferrets treated with BXM increased after the end of treatment and reached the levels similar to those in ferrets treated with vehicle and oseltamivir phosphate probably because antibodies against the virus were not induced in a short period after viral infection. There was no mutation in sequence of the virus isolated in ferrets treated with BXM when compared PA gene sequence of virus obtained from nasal wash fluid on day 2 and 4 post-infection.

Conclusions

- Single day oral administration of BXM had beneficial effects on viral titer and symptoms in ferrets infected with influenza A virus, which were superior to those observed with oseltamivir phosphate even when treatment is initiated after onset of fever in ferrets.
- BXM could have potential to drastically suppress virus replication, which was critical to the significant suppression of a rise in body temperature in ferrets
- These results suggest that BXM could be a promising anti-influenza drug for treatment of influenza virus infection

References

1. T. Noshi et al. Poster presentation at OPTIONS IX, August 2016
2. K.Taniguchi et al. Poster presentation at ESWI, September 2017.
3. K.Taniguchi et al. Poster presentation at ECCMID, April 2017.
4. Ward P, Smith J, Suter P, Dutkowski R. Oseltamivir (Tamiflu) and its potential for use in the event of an influenza pandemic. J Antimicrob Chemother 2005; 55: Suppl 1: i5-i21.