2352

Increased Childhood Recurrent Wheezing and Asthma After Respiratory Syncytial Virus (RSV) Infection in Full-term Infants

Asuncion Mejias, MD, PhD, MSCS[1,*]; Bingcao Wu, MS[2]; Neeta Tandon, MA[2]; Wing Chow, PharmD, MPH[2]; Nancy Connolly, MPH[2]; Sahil Lakhotia, BS[3]; Eduardo Franco, MD[2]; Octavio Ramilo, MD[1]

*Presenting author.

[1]Center for Vaccines and Immunity, The Research Institute at Nationwide Children's Hospital, Columbus, OH, USA; [2]Janssen Scientific Affairs, LLC, Titusville, NJ, USA; [3]Mu Sigma Business Solutions Pvt. Ltd., Bengaluru, India.

BACKGROUND

- The majority of current studies detailing the association between respiratory syncytial virus (RSV) and asthma/recurrent wheezing (RW) have been focused on premature infants who are at a particularly high risk for the development of long-term lung morbidity.
- Large population-based studies in full-term, previously healthy infants can fill an existing knowledge gap.
- Identification of risk factors in infancy may help in identifying and capturing the onset of asthma/RW later in life, thereby potentially minimizing disease burden.

OBJECTIVE

- To identify the incidence and risk factors associated with the development of post-RSV asthma/RW during the first 5 years of life among full-term, healthy infants in a commercially insured US population.

METHODS

- **Data source:** The study used Truven MarketScan Commercial Claims and Encounters Database (CCAE) from 1/1/2000 to 12/31/2016 (study period), which includes de-identified claims of inpatient admissions, outpatient services, pharmacy prescription fills, and enrollment status for individuals enrolled in commercial health insurance plans. The identification period was 1/1/2000 through 12/31/2015.
- **Study design:** Retrospective matched cohort study evaluating the risk of asthma/RW among full-term infants with documented RSV infections during their first year of life versus those without (see **Figure 1**).
 - **Derived birth date:** Day on which the infant's insurance coverage began, as assigned by the infant's insurance provider. Note that full, actual birth dates are not available in claims databases.
 - **Index date:** 1 year after the birth date.
 - **Attribute matching:** RSV infants were paired 1:1 with non-RSV infants based on exact matching of gender, birth month and year, health plan type, and residence region. If there was >1 potential match, the matched counterpart was randomly selected from among the potential matches.
 - **Identification of study population:**
 - RSV infection during the first year of life was defined by the presence of one of the following International Classification of Diseases, Ninth/Tenth Revision (ICD-9/10) diagnosis codes:
 - ◊ RSV (ICD-9: 079.6%; ICD-10: B97.4%).
 - ◊ Acute bronchiolitis due to RSV (ICD-9: 466.11; ICD-10: J21.0%).
 - ◊ Pneumonia due to RSV (ICD-9: 480.1%; ICD-10: J12.1%).
 - Non-RSV controls were infants not meeting the RSV definition and without any of the following ICD-9/10 codes during their first year of life:
 - ◊ Acute bronchitis due to RSV (ICD-9: 466.0%; ICD-10: J20.5%).
 - ◊ Acute bronchiolitis due to other infectious organisms (ICD-9: 466.19; ICD-10: J21.1%, J21.8%, J21.9%).
 - ◊ Unspecified viral pneumonia (ICD-9: 480.9%; ICD-10: J12.9%).
 - ◊ Bronchopneumonia, organism unspecified (ICD-9: 485.%; ICD-10: J18.0%).
 - ◊ Pneumonia, organism unspecified (ICD-9: 486.%; ICD-10: J18.8%, J18.9%).
 - **Full-term infants:** During their first year, defined as having no diagnosis code for short gestation/prematurity (<37 weeks), low birth weight (<2,500 grams), slow fetal growth and malnutrition, congenital heart disease, congenital lung disease, and asthma or wheezing. Non-RSV infants were excluded if they were hospitalized for any reason after their birth discharge during the first year.
 - **Continuous enrollment:** ≥2 years' continuous enrollment following the birth month was required to ensure ≥1 year of post-index follow-up.
- **Inclusion/exclusion criteria:** see **Figure 2**.

Figure 1. Study Design, Retrospective Matched Cohort

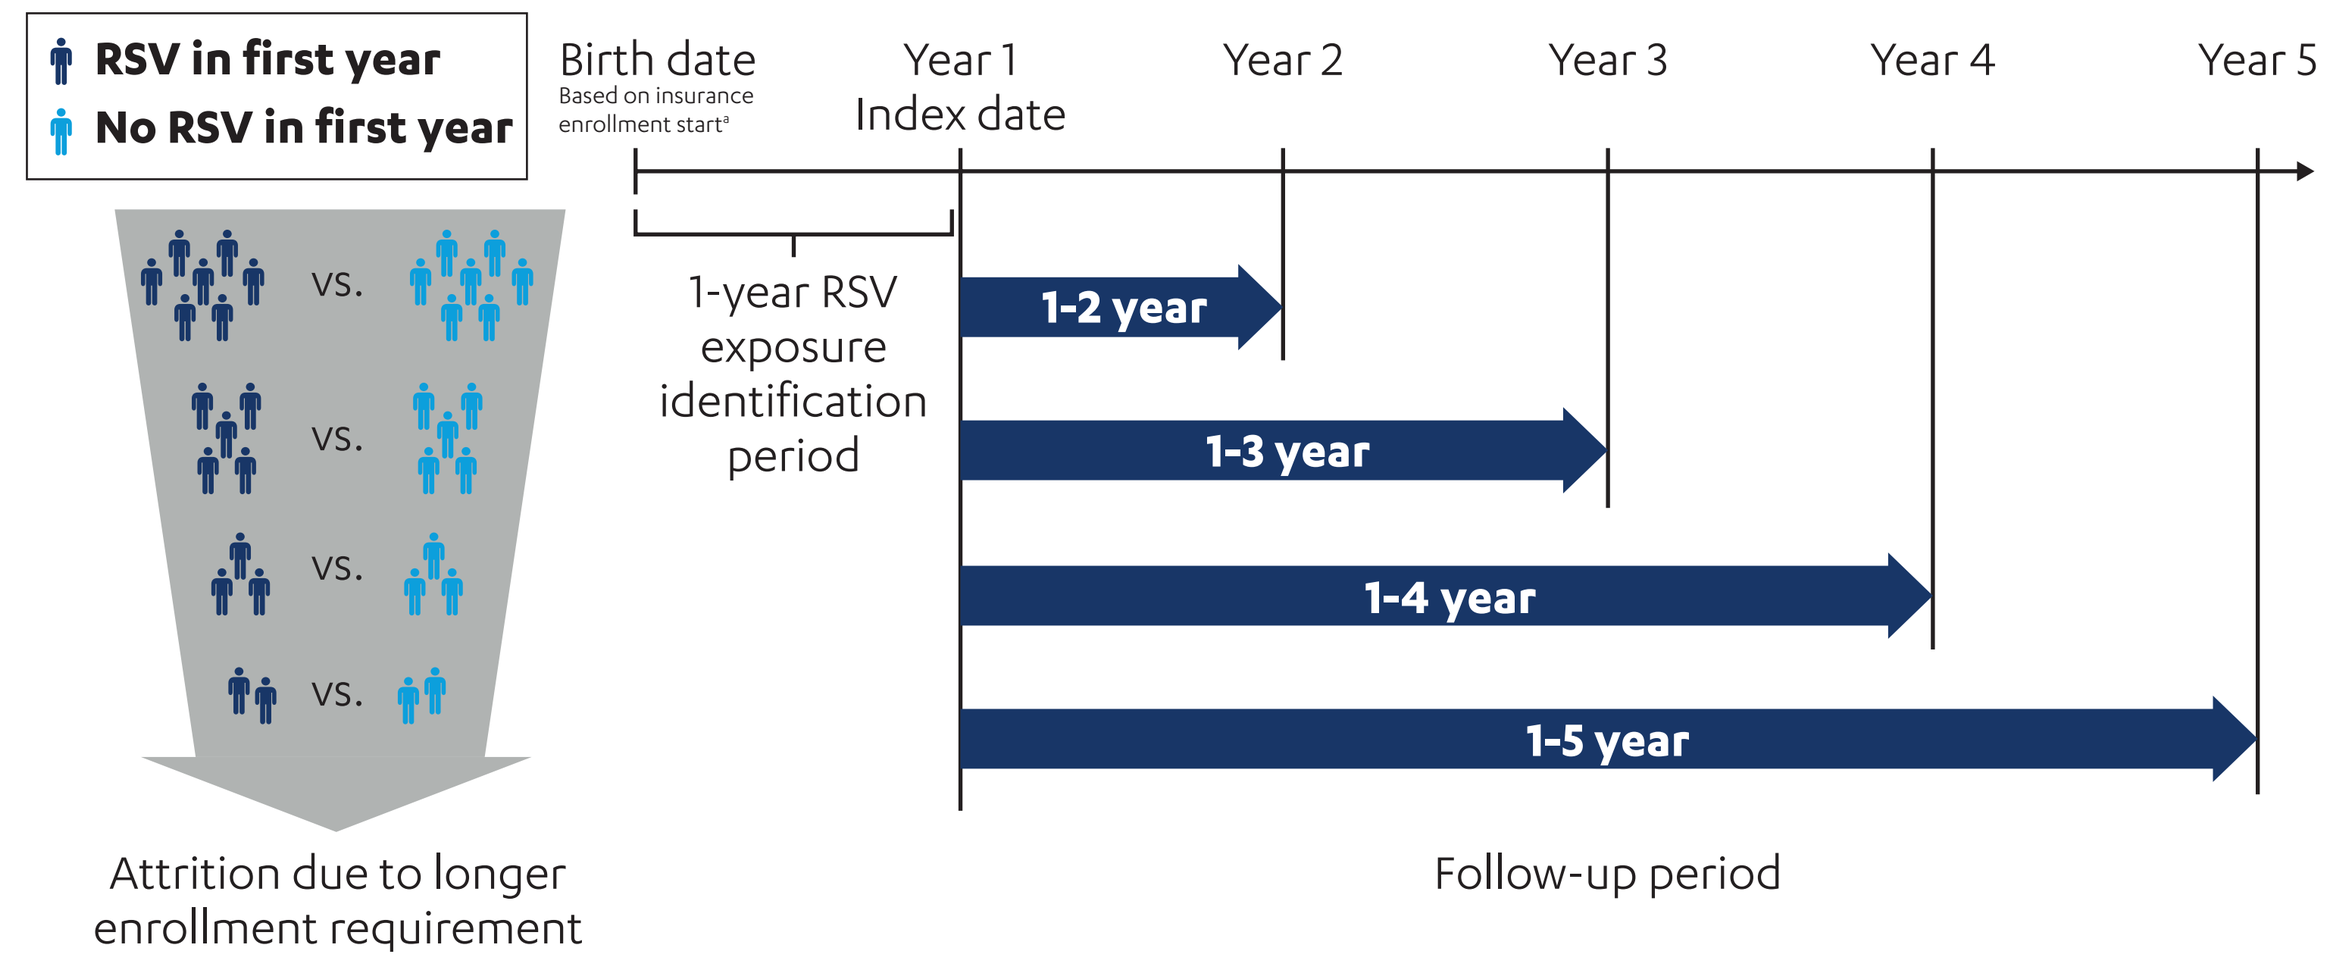
RSV in first year
No RSV in first year
Birth date
Based on insurance enrollment start[a]
Year 1
Index date
Year 2
Year 3
Year 4
Year 5
vs.
1-year RSV exposure identification period
1-2 year
1-3 year
1-4 year
1-5 year
Attrition due to longer enrollment requirement
Follow-up period

RSV: respiratory syncytial virus.
[a]Insurance companies generally assign the first day of the month as the newborn's birth date; therefore, claims databases do not include the infant's actual date of birth. Thus, the difference between the assigned and actual dates can be up to 30 days, with an average difference of approximately 15 days.

Figure 2. Attrition

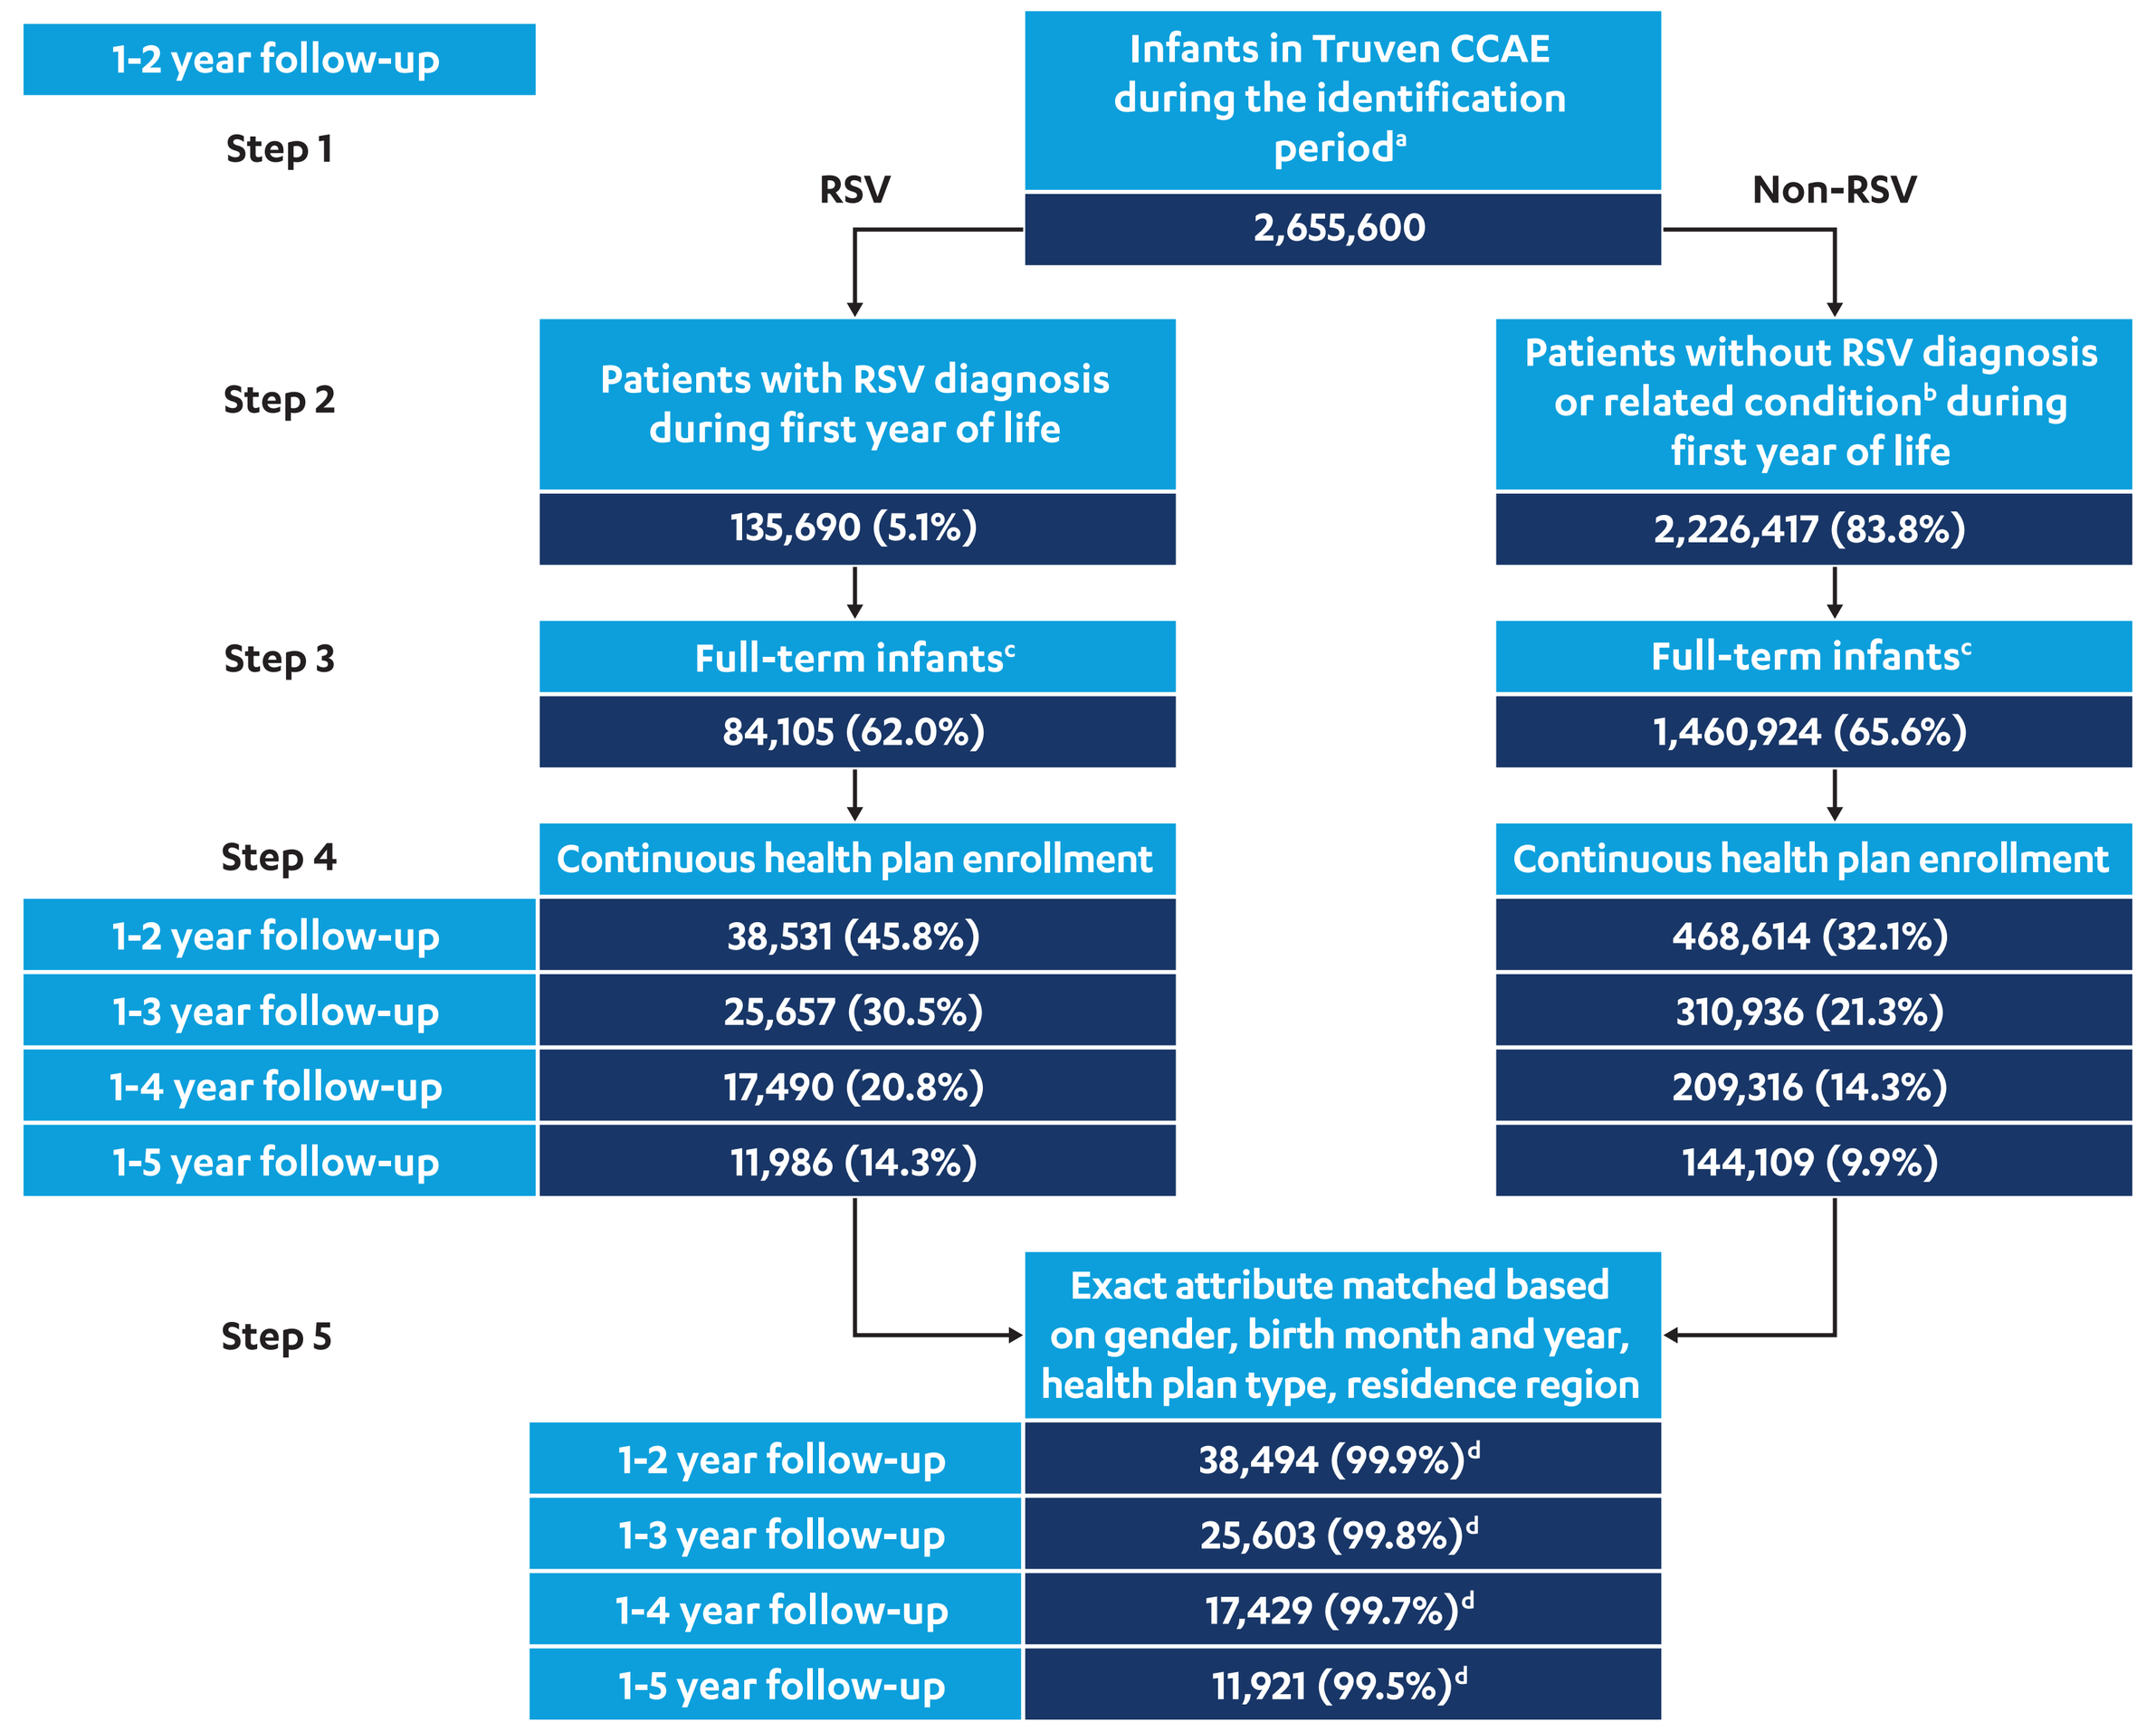
1-2 year follow-up
Step 1
Infants in Truven CCAE during the identification period[a]
2,655,600
RSV
Non-RSV
Step 2
Patients with RSV diagnosis during first year of life
135,690 (5.1%)
Patients without RSV diagnosis or related condition[b] during first year of life
2,226,417 (83.8%)
Step 3
Full-term infants[c]
84,105 (62.0%)
Full-term infants[c]
1,460,924 (65.6%)
Step 4
Continuous health plan enrollment
1-2 year follow-up
38,531 (45.8%)
468,614 (32.1%)
1-3 year follow-up
25,657 (30.5%)
310,936 (21.3%)
1-4 year follow-up
17,490 (20.8%)
209,316 (14.3%)
1-5 year follow-up
11,986 (14.3%)
144,109 (9.9%)
Step 5
Exact attribute matched based on gender, birth month and year, health plan type, residence region
1-2 year follow-up
38,494 (99.9%)[d]
1-3 year follow-up
25,603 (99.8%)[d]
1-4 year follow-up
17,429 (99.7%)[d]
1-5 year follow-up
11,921 (99.5%)[d]

CCAE: Commercial Claims and Encounters Database; RSV: respiratory syncytial virus.
[a]CCAE: 1/1/2000 to 12/31/2015.
[b]RSV-related conditions include: acute bronchitis due to RSV; acute bronchiolitis due to other infectious organisms; unspecified viral pneumonia; bronchopneumonia, organism unspecified; and pneumonia, organism unspecified.
[c]Defined as infants without any diagnosis for short gestation (<37 weeks), low birth weight (<2,500 grams), slow fetal growth and malnutrition, congenital heart disease, congenital lung disease, and asthma or wheezing during the first year of life. Non-RSV infants were excluded if hospitalized for any reason after their birth discharge during the first year.
[d]Reflects percentage of patients in the corresponding follow-up cohort in Step 4. Ns represent matched pairs.

- **Primary outcomes and statistical methods:**
 - Risk of asthma/RW was measured by cumulative incidence and incidence rate.
 - Asthma was identified if there was ≥1 claim with the ICD-9/10 diagnosis (ICD-9: 493.%, 506.3%; ICD-10: J45.%, J68.3%) during the follow-up period.
 - RW was identified on the date of first wheezing diagnosis (ICD-9: 786.07; ICD-10: R06.2%) during the follow-up period, if there were ≥2 additional diagnoses for wheezing during the follow-up period.
 - Cumulative incidence of asthma/RW was estimated through 1, 2, 3, and 4 years' post-index follow-up (Year 1 - Year 4) among the matched RSV and non-RSV cohorts. Adjusted odds ratio with 95% confidence interval (CI) and *p*-value were calculated via multivariable conditional logistic regression model for each follow-up period.
 - Incidence rate per 1,000 patient-years was calculated among the matched RSV and non-RSV cohorts of infants with ≥1 year of follow-up. Adjusted incidence rate ratio with 95% CI and *p*-value were calculated via multivariable conditional general linear model, assuming Poisson distribution and logarithm link function and offsetting by at-risk time.
 - Dichotomous covariates for the multivariable models included presence or absence of the following:
 - "Comorbid infections specific to the perinatal period," such as congenital or neonatal infection with rubella, cytomegalovirus, or Candida, among others (ICD-9: 771.%; ICD-10: P35.% - P39.%), or "other respiratory conditions of fetus and newborn," such as congenital pneumonia, fluid aspiration, apnea, or cyanosis, among others (ICD-9: 770.%; ICD-10: P23.% - P28.%).
 - Influenza vaccination.
 - Pneumococcal vaccination.
- **Other statistical analyses:**
 - Demographic variables were descriptively reported.
 - Baseline clinical characteristics were presented and compared between the RSV and non-RSV cohorts via bivariate analyses.
 - *p*<0.05 was considered statistically significant. All analyses used SAS Enterprise Guide, version 7 (SAS Institute, Cary, NC).

RESULTS

- Sample size: 38,494 (Year 1), 25,603 (Year 2), 17,429 (Year 3), and 11,921 (Year 4) matched pairs.
- Demographics: see **Table 1**.
 - More prevalent were: 1. Birth months late in the year, reflective of RSV seasonality; 2. Southern geographic region, likely driven by insurance plans represented and the high prevalence of RSV infection in the state of Florida; 3. Preferred provider organization (PPO) plan type.

Table 1. Demographics of the Matched RSV/Non-RSV Cohorts

	Truven Commercial RSV/non-RSV 1-2 year follow-up	
Sample size per cohort, n	38,494	
Female, n, %	17,606	45.7%
Month of birth, n, %		
January	2,494	6.5%
February	2,263	5.9%
March	2,552	6.6%
April	2,882	7.5%
May	2,945	7.7%
June	3,119	8.1%
July	3,851	10.0%
August	3,844	10.0%
September	3,904	10.1%
October	4,094	10.6%
November	3,478	9.0%
December	3,068	8.0%
Region, n, %		
Northeast	4,527	11.8%
Midwest	8,194	21.3%
South	20,981	54.5%
West	4,338	11.3%
Other	454	1.2%
Plan type, n, %		
HMO	6,412	16.7%
PPO	23,590	61.3%
POS	3,268	8.5%
Other	4,782	12.4%

HMO: health maintenance organization; POS: point of service; PPO: preferred provider organization; RSV: respiratory syncytial virus.

- Baseline characteristics: see **Table 2**.
 - RSV infants had a significantly longer hospital stay than non-RSV infants due to RSV-related admissions. For non-RSV infants, the results represent the hospital stay due to birth.
 - Most of the other medical conditions examined during the first year of life were rare.
 - Expectedly, continuous use of corticosteroids for ≥14 days and use of antibiotics were substantially more prevalent in RSV infants than non-RSV infants.

Table 2. Baseline Characteristics of the Matched RSV/Non-RSV Cohorts

	Truven Commercial RSV/non-RSV 1-2 year follow-up		
	RSV	Non-RSV	*p*-value
Sample size, n	38,494	38,494	
Length of hospitalization, days, mean (SD)	3.9 (3.8)	2.0 (1.8)	<0.0001
Comorbid conditions, %			
Bronchopulmonary dysplasia	0.03%	0.00%	0.0707
Congestive heart failure	0.08%	0.01%	<0.0001
Cystic fibrosis	0.07%	0.03%	0.0076
Down's syndrome	0.15%	0.04%	<0.0001
Infant respiratory distress syndrome	1.44%	0.43%	<0.0001
Infections specific to the perinatal period	5.43%	3.24%	<0.0001
Immunodeficiency	0.13%	0.02%	<0.0001
Neuromuscular disease	0.04%	0.01%	0.2206
Other respiratory conditions of fetus and newborn	5.82%	2.57%	<0.0001
Primary pulmonary hypertension	0.03%	0.00%	0.0013
Medication/vaccination use in first year, %			
Influenza vaccination	38.2%	36.1%	<0.0001
Pneumococcal vaccination	89.4%	84.6%	<0.0001
Antibiotics	76.7%	44.7%	<0.0001
Chronic systemic corticosteroid therapy[a]	24.8%	5.4%	<0.0001
Inhaled corticosteroid therapy	8.7%	0.3%	<0.0001
Other asthma/RW risk factors, %			
Presence of RSV test	45.2%	3.0%	<0.0001

RSV: respiratory syncytial virus; RW: recurrent wheezing; SD: standard deviation.
[a]Chronic systemic corticosteroid therapy refers to continuous corticosteroid use for ≥14 days.

- Cumulative incidence of asthma/RW was significantly higher in RSV infants than non-RSV infants. The risk was particularly high in the early years (see **Figure 3**).

Figure 3. Cumulative Incidence and Adjusted Odds Ratio[a] of Asthma/RW Across Follow-up Periods

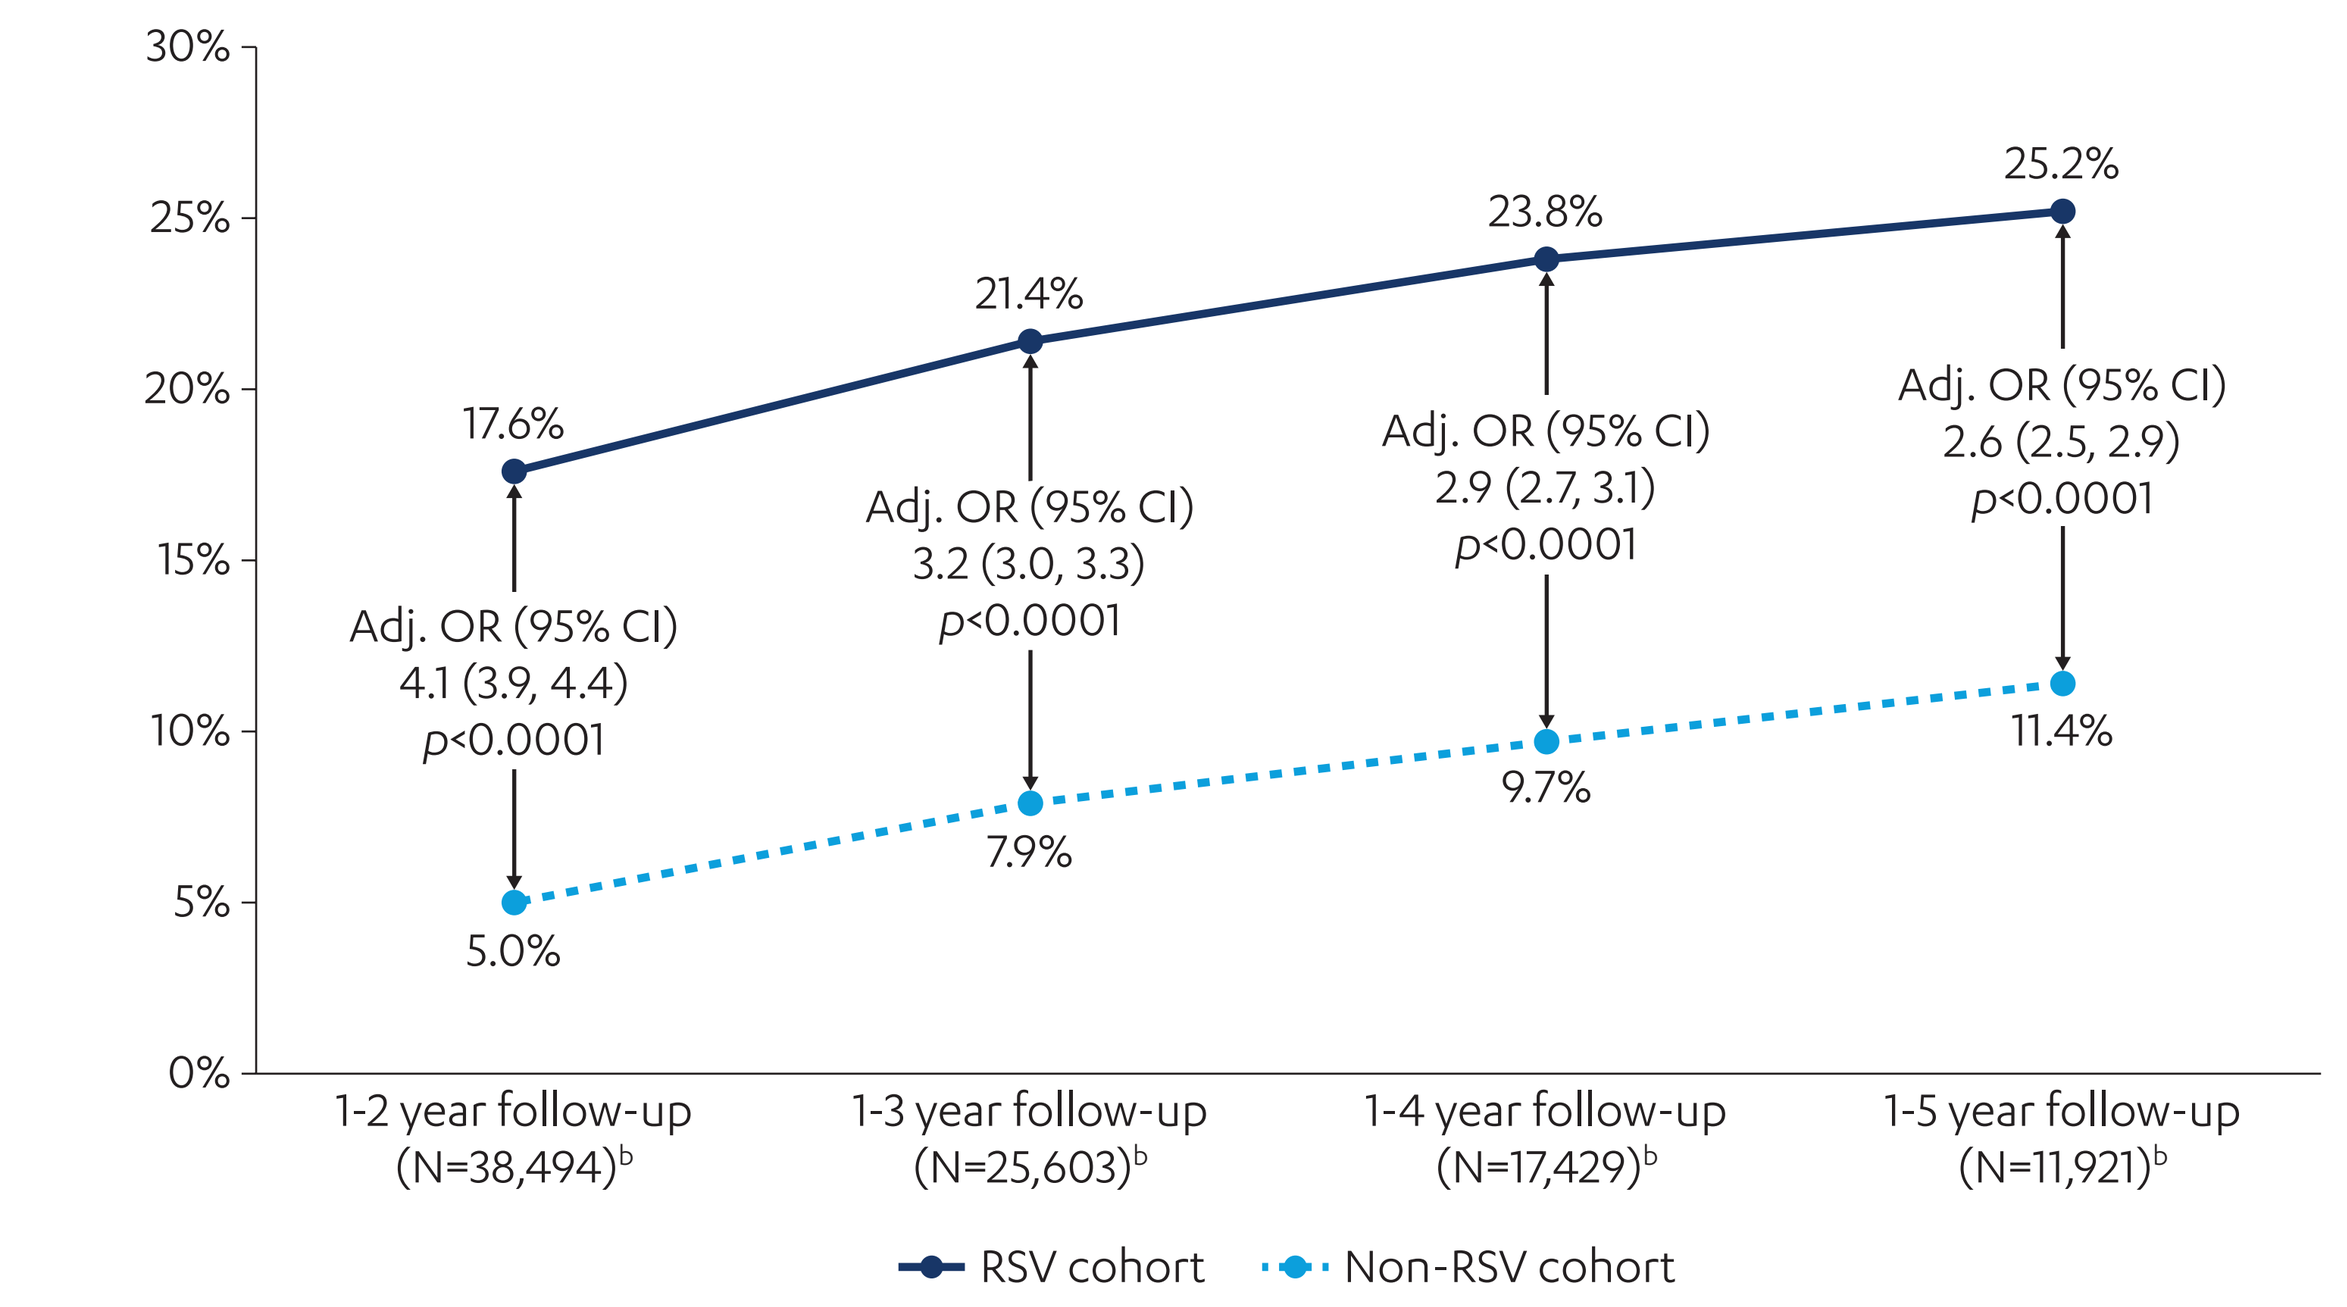
30%
25%
20%
15%
10%
5%
0%
17.6%
21.4%
23.8%
25.2%
Adj. OR (95% CI)
4.1 (3.9, 4.4)
p<0.0001
Adj. OR (95% CI)
3.2 (3.0, 3.3)
p<0.0001
Adj. OR (95% CI)
2.9 (2.7, 3.1)
p<0.0001
Adj. OR (95% CI)
2.6 (2.5, 2.9)
p<0.0001
5.0%
7.9%
9.7%
11.4%
1-2 year follow-up (N=38,494)[b]
1-3 year follow-up (N=25,603)[b]
1-4 year follow-up (N=17,429)[b]
1-5 year follow-up (N=11,921)[b]
RSV cohort
Non-RSV cohort

Adj: adjusted; CI: confidence interval; OR: odds ratio; RSV: respiratory syncytial virus.
[a]Adj. ORs were calculated via multivariable conditional logistic regression controlling for presence or absence of: 1. infections specific to the perinatal period or other respiratory conditions of fetus and newborn, 2. influenza vaccination, and 3. pneumococcal vaccination.
[b]Ns represent matched pairs.

- Incidence rate of asthma/RW was 2.4 (95% CI: 2.3, 2.5) times higher in RSV infants (69.7 per 1,000 patient-years) compared with non-RSV infants (28.7 per 1,000 patient-years; see **Table 3**).

Table 3. Incidence Rate per 1,000 Patient-years and Adjusted Incidence Rate Ratio

			RSV vs. non-RSV (reference)	
	RSV	Non-RSV	Adj. IRR (95% CI)	*p*-value
Truven Commercial				
Total patients at-risk, n	38,494	38,494		
Incident cases, n	8,005	3,612		
Total patient-years at-risk	114,833	126,005		
Incidence rate per 1,000 patient-years	69.7	28.7	2.4 (2.3, 2.5)	<0.0001

Adj: adjusted; CI: confidence interval; IRR: incidence rate ratio; RSV: respiratory syncytial virus.

LIMITATIONS

- This retrospective, observational study had limitations (eg, identification of RSV, asthma, and RW depended on ICD-9/10 diagnosis codes). Also, potential coding errors and inconsistencies in diagnostic methodologies used across providers may have contributed to potential misclassification biases.
- Mothers' characteristics and medical histories were not adjusted in this study due to data limitations on mother-child linkage, but potential bias is considered minor in this full-term infant population.
- Date of birth is precise only to the month and year of birth, introducing a slight potential for information bias, but the proxy approach used in this study is reasonable.

CONCLUSIONS

- **Full-term, commercially insured children infected with RSV during infancy had a significantly increased risk of developing asthma or RW during the first 5 years of life.**
- **Future studies to explore the mechanisms of this association are needed, but this study confirms the increased risk of developing asthma/RW after RSV infection.**
- **These results also confirm an important medical need for safe and effective interventions targeting RSV infection in infants to reduce its associated long-term respiratory morbidity.**

DISCLOSURES

A. Mejias has received consulting fees and research grants from Janssen, and speaker honoraria from AbbVie. O. Ramilo has received consulting fees from Janssen. This study was funded by Janssen Scientific Affairs, LLC. Technical editorial assistance was provided by Meghan O'Leary, of MedErgy, and was supported by Janssen Scientific Affairs, LLC.

janssen Infectious Diseases & Vaccines
PHARMACEUTICAL COMPANIES OF Johnson & Johnson

An electronic version of the poster can be viewed by scanning the QR code. The QR code is intended to provide scientific information for individual reference. The PDF should not be altered or reproduced in any way.

POSTER PRESENTED AT IDWeek™ 2018; OCTOBER 3-7, 2018; SAN FRANCISCO, CALIFORNIA.